Poster: 182 ePoster: 251

An In-Depth Analysis of Serum Phosphate Changes in Patients With CKD on Dialysis: Results From the PHREEDOM Study

Stuart M. Sprague,[1] Yang Yang,[2] David P. Rosenbaum,[2] Susan Edelstein,[2] and David M. Spiegel[2]

[1]NorthShore University HealthSystem-University of Chicago Pritzker School of Medicine, Chicago, IL, USA; [2]Ardelyx, Inc., Waltham, MA, USA

Background

- According to the Dialysis Outcomes and Practice Patterns Study (DOPPS) practice monitor, around 70% of patients with chronic kidney disease (CKD) on dialysis have serum phosphate (sP) >4.5 mg/dL in any given month despite receiving treatment for hyperphosphatemia, suggesting that current treatment strategies are insufficient.[1,2]
- Tenapanor is a novel, investigational phosphate absorption inhibitor (PAI) that targets the primary, paracellular pathway of phosphate absorption in the gastrointestinal tract through selective inhibition of sodium/hydrogen exchanger isoform 3 (NHE3).[3-5]
- We previously reported results of the phase 3 PHREEDOM (NCT03427125) study that evaluated tenapanor for the treatment of hyperphosphatemia in patients with CKD on dialysis.[6]
 - In PHREEDOM, patients treated with sevelamer served as an active safety control.[6]
- Here we present post hoc analyses of sP concentrations in patients with CKD on dialysis in response to treatment with tenapanor or sevelamer from PHREEDOM.

Methods

- Methods for PHREEDOM have been previously described[6]; briefly:
 - PHREEDOM enrolled patients on maintenance dialysis with sP ≥6.0 and ≤10.0 mg/dL and a ≥1.5 mg/dL increase in sP after phosphate binder washout.
 - Patients were randomized 3:1 to receive tenapanor 30 mg twice a day (bid) or sevelamer per standard of care during a 26-week randomized treatment period (RTP).
 - Upon completion of the 26-week RTP, tenapanor-treated patients were then re-randomized 1:1 to tenapanor or placebo in a 12-week withdrawal period, followed by open-label tenapanor treatment in a 14-week safety extension.
 - Sevelamer-treated patients received sevelamer for up to 52 weeks.
- In the current analysis, sP concentrations were analyzed in tenapanor-treated and sevelamer-treated patients who had a 1.2 mg/dL reduction from baseline at week 26 (defined as sP responders) and for all patients who completed treatment for 52 weeks.

Results

Patients

- Demographics and baseline characteristics were previously published for the tenapanor (N=419) and sevelamer (N=137) groups and were similar between the 2 groups.[6]
 - Patients treated with tenapanor had a baseline mean (SD) sP of 7.4 (1.4) mg/dL and patients treated with sevelamer had a baseline mean (SD) sP of 7.2 (1.5) mg/dL.

Treatment Response

- The least squares (LS) mean change (SE) ranged from −1.06 (0.065) to −1.37 (0.088) mg/dL in the tenapanor group and −1.12 (0.115) to −1.80 (0.130) mg/dL in the sevelamer group for all patients across the RTP (**Figure 1A**).
- In sP responders, the LS mean (SE) sP reductions at the end of the RTP was −2.57 (0.08) mg/dL in the tenapanor group and −2.68 (0.11) mg/dL in the sevelamer group (**Figure 1B**).

Figure 1. Change From Baseline in sP

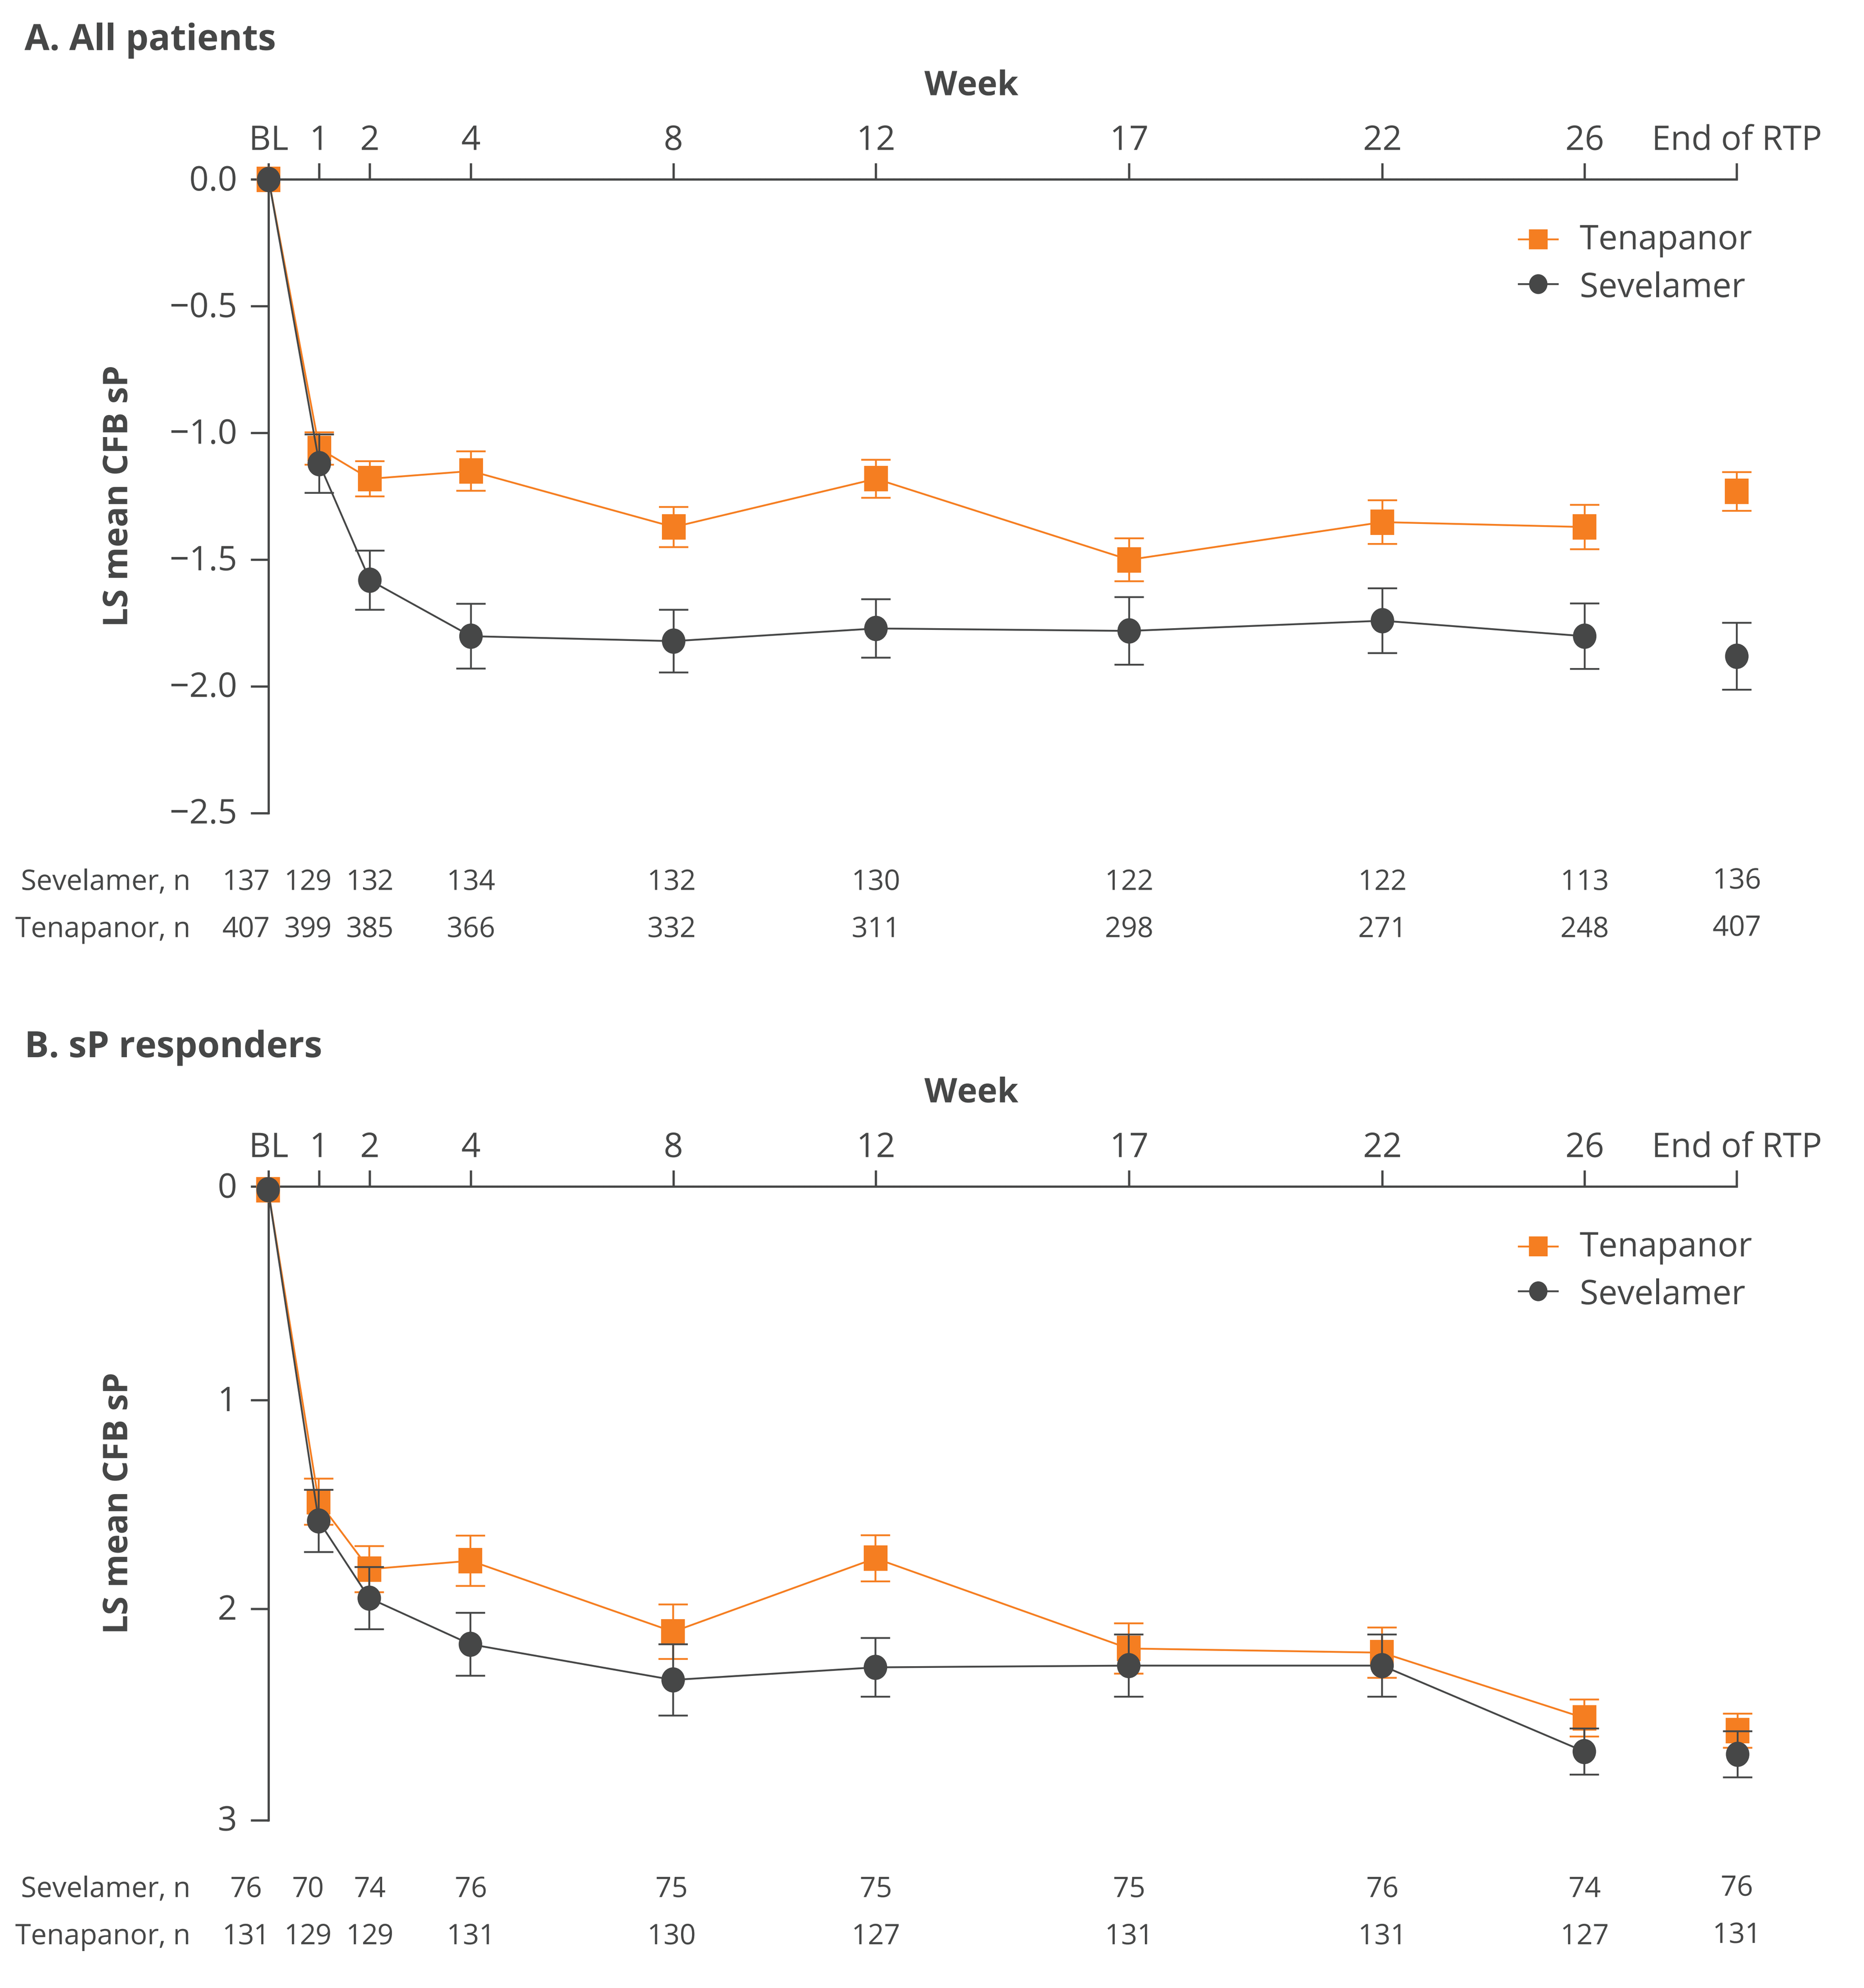
A. All patients
Week
BL 1 2 4 8 12 17 22 26 End of RTP
0.0
−0.5
−1.0
−1.5
−2.0
−2.5
LS mean CFB sP
Tenapanor
Sevelamer
Sevelamer, n 137 129 132 134 132 130 122 122 113 136
Tenapanor, n 407 399 385 366 332 311 298 271 248 407
B. sP responders
Week
BL 1 2 4 8 12 17 22 26 End of RTP
0
1
2
3
LS mean CFB sP
Tenapanor
Sevelamer
Sevelamer, n 76 70 74 76 75 75 75 76 74 76
Tenapanor, n 131 129 129 131 130 127 131 131 127 131

BL, baseline; CFB, change from baseline; LS, least squares; RTP, randomized treatment period; sP, serum phosphate.

- Among patients who continued on treatment for 52 weeks with tenapanor (n=88) or sevelamer (n=108), similar percentages of patients achieved sP reductions of ≥1.5 (50% vs 49%, respectively) or ≥2.0 mg/dL (38% vs 39%, respectively) and achieved target sP <5.5 mg/dL (47% vs 44%, respectively) at week 52 (**Figure 2**).

Figure 2. Percentage of 52-Week Completers Achieving sP Reduction Threshold or Reaching sP <5.5 mg/dL

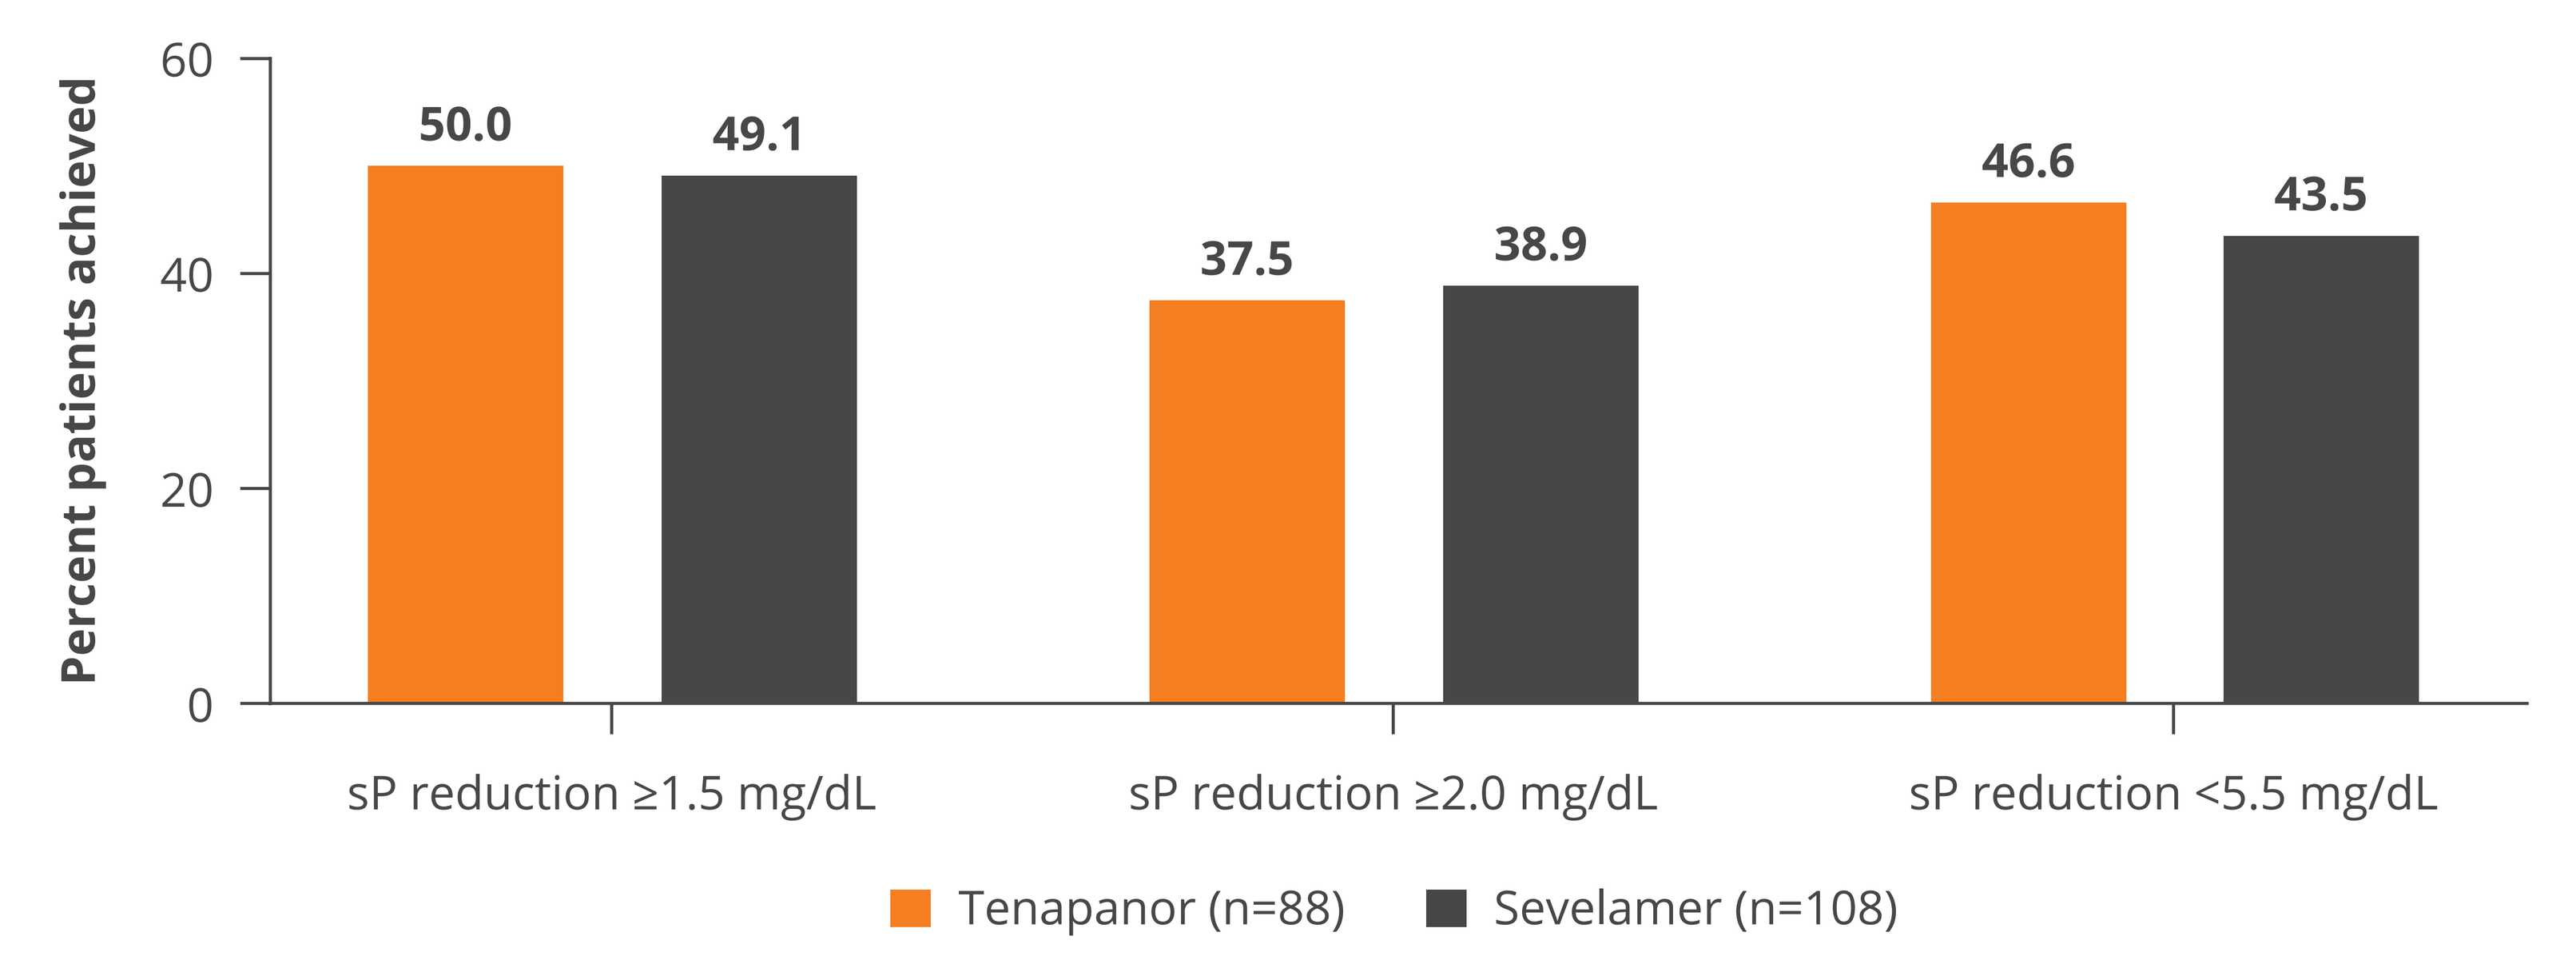
Percent patients achieved
60
40
20
0
50.0
49.1
37.5
38.9
46.6
43.5
sP reduction ≥1.5 mg/dL
sP reduction ≥2.0 mg/dL
sP reduction <5.5 mg/dL
Tenapanor (n=88)
Sevelamer (n=108)

52-week completers who were re-randomized to placebo during the withdrawal period were excluded. sP, serum phosphate.

Medication Burden

- The comparable percentages of patients achieving the sP reductions depicted in Figure 2 between groups were achieved with 2 tenapanor tablets vs a median of 9 sevelamer tablets per day at study end.

Safety

- During the 26-week treatment period, 80.4% of patients in the tenapanor arm and 64.2% of patients in the sevelamer arm reported a treatment-emergent adverse event (TEAE).
 - 63.5% of patients in the sevelamer group had been treated with sevelamer prior to entering the RTP.
- Serious AEs were reported at a higher overall incidence rate in patients receiving sevelamer compared with tenapanor (**Table 1**).
- Diarrhea was the most commonly reported TEAE in both the tenapanor and sevelamer groups (**Table 1**).

Table 1. Treatment-Emergent Adverse Events During the 26-Week Treatment Period (Safety Analysis Set)

	Tenapanor (N=419)	Sevelamer (N=137)
All TEAEs	337 (80.4)	88 (64.2)
Serious AEs	73 (17.4)	32 (23.4)
TEAEs leading to death	1 (0.2)	0
TEAEs leading to study drug discontinuation	102 (24.3)	2 (1.5)
TEAEs by preferred term reported by >3% of patients		
Diarrhea	222 (53.0)	10 (7.3)
Fall	11 (2.6)	10 (7.3)
Cough	9 (2.1)	9 (6.6)
Arteriovenous fistula thrombosis	6 (1.4)	7 (5.1)
Fluid overload	8 (1.9)	7 (5.1)
Hypertension	15 (3.6)	7 (5.1)
Pneumonia	4 (1.0)	7 (5.1)
Constipation	2 (0.5)	6 (4.4)
Hyperkalemia	19 (4.5)	6 (4.4)
Hypotension	5 (1.2)	6 (4.4)
Arteriovenous fistula site complication	7 (1.7)	5 (3.6)
Pain in extremity	3 (0.7)	5 (3.6)
Pulmonary mass	0	5 (3.6)
Upper respiratory tract infection	5 (1.2)	5 (3.6)
Vomiting	13 (3.1)	5 (3.6)
Nausea	15 (3.6)	4 (2.9)
Hyperphosphatemia	27 (6.4)	3 (2.2)

Data are n (%). AE, adverse event; TEAE, treatment-emergent adverse event.

Conclusions

This analysis demonstrates that patients with CKD on dialysis who respond to treatment with tenapanor or sevelamer for hyperphosphatemia have comparable sP reductions.

Tenapanor treatment had the added benefit of bid dosing and a much lower pill burden.

Diarrhea was the most common TEAE reported by patients treated with tenapanor, with a higher incidence rate observed in patients treated with tenapanor. Prior sevelamer use by over 60% of the sevelamer-treated patients may in part explain the lower overall rate of TEAEs than reported in the sevelamer prescribing information.

Overall, this post hoc analysis of the PHREEDOM study demonstrates the value of tenapanor as a treatment option for hyperphosphatemia in patients with CKD receiving maintenance dialysis.

Disclosures

Stuart M. Sprague receives research grants from and is a consultant for Ardelyx, Inc. Yang Yang, David Rosenbaum, Susan Edelstein, and David M. Spiegel are employees of Ardelyx, Inc.

References

1. Clegg DJ et al. *Nephrol Renal Dis*. 2022;7:1-7.
2. Guedes M et al. *Kidney Med*. 2022;5:100584.
3. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
4. Spencer AG et al. *Sci Transl Med*. 2014;6:227ra36.
5. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
6. Block GA et al. *Kidney 360*. 2021;2:1600-10.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr. Sprague can be contacted for further information on this study at SSprague@northshore.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

National Kidney Foundation Spring Clinical Meetings (SCM23); April 11-15, 2023; Austin, TX, USA

MED-US--2400027

Electronic Certificate

Version: 2 . 0

Document Number: MED-US--2400027

Document Name: 1175748_NKF2023_PHREEDOM_Poster_FINAL

Country: United States

Product: Tenapanor-HP

Type: External Communication

Sub Type: MSL / Field Material

Classification: Poster

Certification Statement
We certify that the final electronic form of this material is in accordance with the regulations set forth by the health authority for the country of this document, and is a fair and truthful presentation of the facts about the product.

Role	Signature
Role: Approver Workflow: Medical Coordinator: Assess Outcome Outcome: Approved for Use	Name: May Luong (mluong@ardelyx.com) Date: 26-Feb-2024 20:33:47 GMT+0000 Statement: Assess the outcome of review, then click the complete button for this task to select the next workflow.