Poster: 183 ePoster: 252

Post Hoc Analyses of Serum Phosphate Changes With Tenapanor in Black/African American and White Patients on Maintenance Dialysis

Stuart M. Sprague,[1] Yang Yang,[2] Suling Zhao,[2] Susan Edelstein,[2] and David M. Spiegel[2]

[1]NorthShore University HealthSystem-University of Chicago Pritzker School of Medicine, Chicago, IL, USA;
[2]Ardelyx, Inc., Waltham, MA, USA

Background

- According to the Dialysis Outcomes and Practice Patterns Study (DOPPS) practice monitor, around 70% of patients with chronic kidney disease (CKD) on dialysis have serum phosphate (sP) >4.5 mg/dL in any given month despite receiving treatment for hyperphosphatemia, suggesting that current treatment strategies are insufficient.[1,2]
- Tenapanor is a novel, investigational phosphate absorption inhibitor (PAI) that targets the primary, paracellular pathway of phosphate absorption in the gastrointestinal tract through inhibition of sodium/hydrogen exchanger isoform 3 (NHE3).[3-5]
- In the US, Black/African Americans are at greater risk of developing CKD than White Americans and make up a disproportionate number of patients on maintenance dialysis relative to the general population.[6]
- We assessed sP changes in Black/African American and White patients in post hoc analyses from 3 clinical trials of tenapanor (NCT02675998, PHREEDOM [NCT03427125], AMPLIFY [NCT03824587]).

Methods

- Methods for NCT02675998,[7] PHREEDOM,[8] and AMPLIFY[9] have been previously described.
- Efficacy data are presented for the randomized withdrawal period (RWP) of NCT02675998 and PHREEDOM, where the sP is expected to increase in the placebo group, and for the 4-week randomized treatment period (RTP) of AMPLIFY. Safety data are presented for the RTP of each study.
- For each study, we evaluated least squares (LS) mean change of sP from baseline in patients from the following analysis set who identified as Black/African American or White:
 - NCT02675998 and PHREEDOM: the efficacy analysis set was a subset of the intent-to-treat (ITT) analysis set for the RWP. It included all ITT patients who met the study inclusion/exclusion criteria, received ≥1 dose of tenapanor during the RTP, completed the RTP, and achieved a sP reduction of ≥1.2 mg/dL from baseline to the end of the RTP.
 - AMPLIFY: the full analysis set, which included all ITT patients with ≥1 postbaseline sP measurement, was used for the evaluation of efficacy endpoints.

Results

Patients

- Demographics and baseline characteristics of Black/African American and White patients in the safety analysis set are presented for placebo and tenapanor groups across each trial in **Table 1**.

Table 1. Demographics and Baseline Characteristics by Race (Safety Analysis Set)

	NCT02675998[a]		PHREEDOM				AMPLIFY			
	Tenapanor (N=210)		Sevelamer (N=130)		Tenapanor (N=384)		Placebo+PB (N=119)		Tenapanor+PB (N=117)	
Characteristic category/statistic	White (n=85)	Black (n=125)	White (n=70)	Black (n=60)	White (n=189)	Black (n=195)	White (n=60)	Black (n=50)	White (n=57)	Black (n=52)
Age at screening, years	59.07 (11.05)	53.82 (10.19)	58.91 (13.07)	58.09 (11.91)	58.20 (12.94)	57.05 (12.62)	55.09 (12.32)	51.96 (12.15)	56.42 (10.61)	54.62 (14.27)
Male, n (%)	60 (70.6)	62 (49.6)	45 (64.3)	41 (68.3)	123 (65.1)	121 (62.1)	41 (68.3)	29 (58.0)	35 (63.2)	23 (44.2)
Ethnicity, n (%)										
Hispanic or Latino	35 (41.2)	2 (1.6)	39 (55.7)	2 (3.3)	110 (58.2)	3 (1.5)	28 (46.7)	1 (2.0)	36 (63.2)	1 (1.9)
Not Hispanic or Latino	50 (58.8)	123 (98.4)	31 (44.3)	58 (96.7)	79 (41.8)	191 (98.0)	31 (51.7)	49 (98.0)	20 (35.1)	51 (98.1)
NR/unknown	0	0	0	0	0	1 (0.5)	1 (1.7)	0	1 (1.7)	0
Duration since first dialysis at baseline, months	60.11 (70.15)	59.13 (48.85)	54.12 (62.44)	65.53 (53.44)	46.75 (38.59)	58.61 (55.20)	40.08 (38.62)	53.04 (44.95)	57.06 (45.02)	58.96 (54.34)
Baseline sP, mg/dL	7.51 (1.50)	7.47 (1.60)	7.20 (1.51)	7.21 (1.40)	7.67 (1.57)	7.23 (1.31)	6.89 (1.32)	7.02 (1.52)	6.89 (1.43)	6.45 (1.17)

Unless otherwise indicated, data are mean (SD). Baseline was defined as the last measurement collected before the first dose of tenapanor in the study. Demographic and baseline characteristic data presented are from the safety analysis set of each study.
[a]Safety data were collected during the RTP. There was no placebo arm during the treatment period of this study.
NR, not reported; PB, phosphate binder; RTP, randomized treatment period; sP, serum phosphate.

Results

NCT02675998

- In NCT02675998, the LS mean difference (95% CI) between tenapanor and placebo in sP change from period-level baseline to the end of the RWP was −1.08 (−1.87, −0.29) mg/dL among Black/African American patients and −0.66 (−1.89, 0.57) mg/dL among White patients in the efficacy analysis set (**Figure 1A**).
- No significant differences in mean sP were observed between Black/African American and White patients (**Figure 1B**).

Figure 1. sP Change From Period-Level Baseline to End of RWP by Race in NCT02675998 (Efficacy Analysis Set)

A. LS mean difference in sP in the RWP

Subgroup	N	Placebo	Tenapanor	LSM change difference	*P* value
All subjects	79	1.38	0.56	−0.82 (−1.44, −0.21)	0.010
Race					
White	117	1.51	0.86	−0.66 (−1.89, 0.57)	0.280
Black or African American	101	1.55	0.47	−1.08 (−1.87, −0.29)	0.008

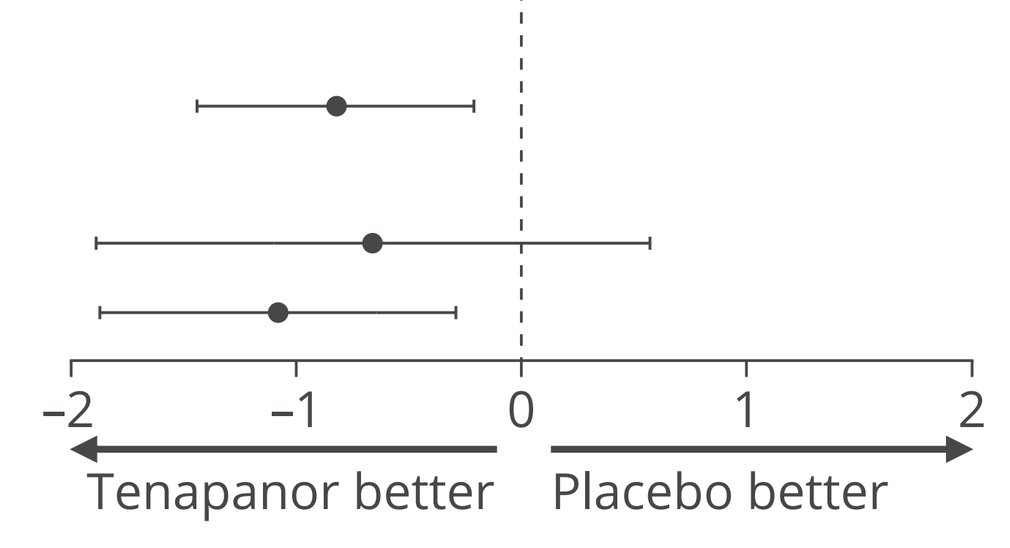
LS mean change difference and 95% CI
−2
−1
0
1
2
Tenapanor better
Placebo better

B. Mean sP in the RWP

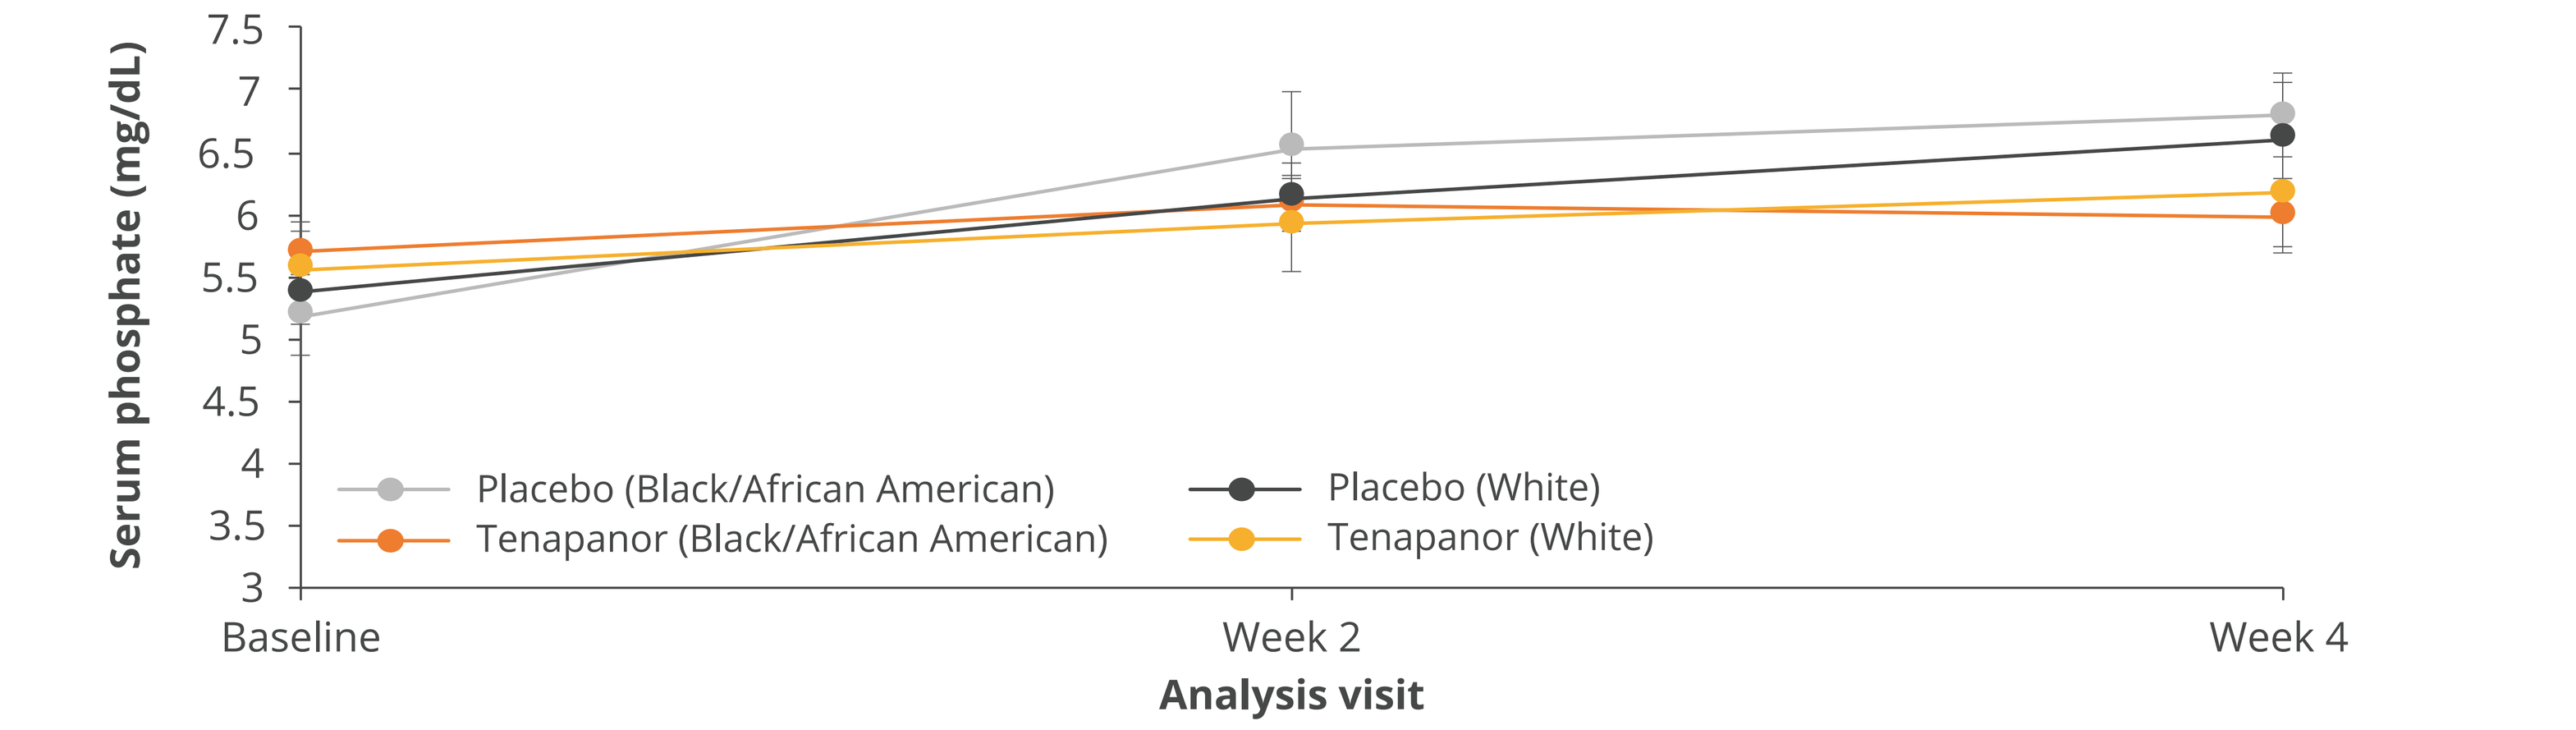
Serum phosphate (mg/dL)
7.5
7
6.5
6
5.5
5
4.5
4
3.5
3
Placebo (Black/African American)
Tenapanor (Black/African American)
Placebo (White)
Tenapanor (White)
Baseline
Week 2
Week 4
Analysis visit

The LS mean estimates presented in Figure 1A are obtained from the pre-specified ANCOVA for the primary efficacy endpoint. The mean ± standard error presented in Figure 1B are obtained from the descriptive summary of the sP level by analysis visit during the RWP.
ANCOVA, analysis of covariance; LS, least squares; LSM, least squares mean; RWP, randomized withdrawal period; sP, serum phosphate.

PHREEDOM

- During the PHREEDOM RWP, the LS mean difference (95% CI) between tenapanor and placebo in sP change was −1.36 (−2.15, −0.57) mg/dL among Black/African American patients and −1.45 (−2.32, −0.59) mg/dL among White patients in the efficacy analysis set (**Figure 2A**).
- No significant differences in mean sP were observed between Black/African American and White patients in the efficacy analysis set of the RTP (**Figure 2B**) or the RWP (**Figure 2C**).

Figure 2. sP Change From Period-Level Baseline to End of RWP by Race in PHREEDOM (Efficacy Analysis Set)

A. LS mean difference in sP in the RWP

Subgroup	N	Placebo	Tenapanor	LSM change difference	*P* value
All subjects	131	1.80	0.43	−1.37 (−1.92, −0.82)	<0.0001
Race					
White	59	1.88	0.43	−1.45 (−2.32, −0.59)	0.0014
Black or African American	66	2.03	0.67	−1.36 (−2.15, −0.57)	0.0010

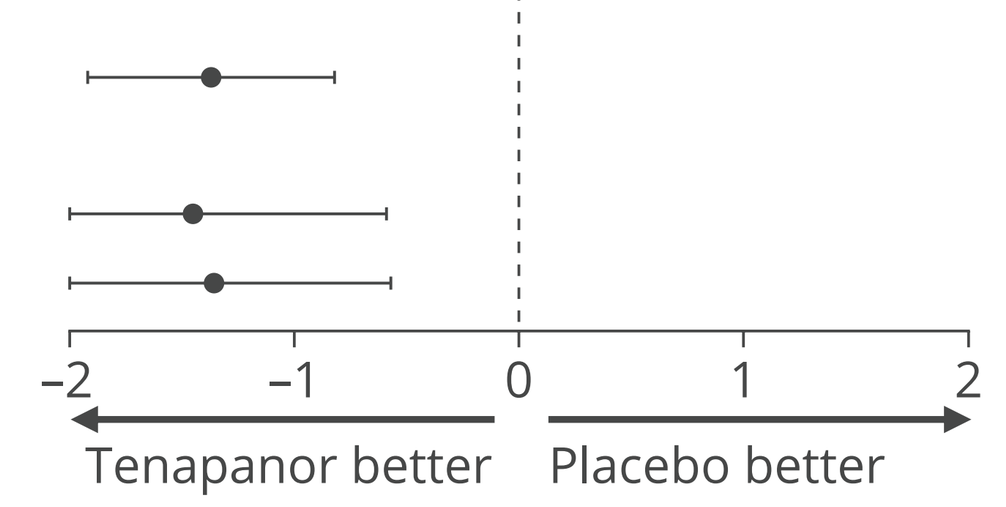
LS mean change difference and 95% CI
−2
−1
0
1
2
Tenapanor better
Placebo better

B. Mean sP in the RTP[a]

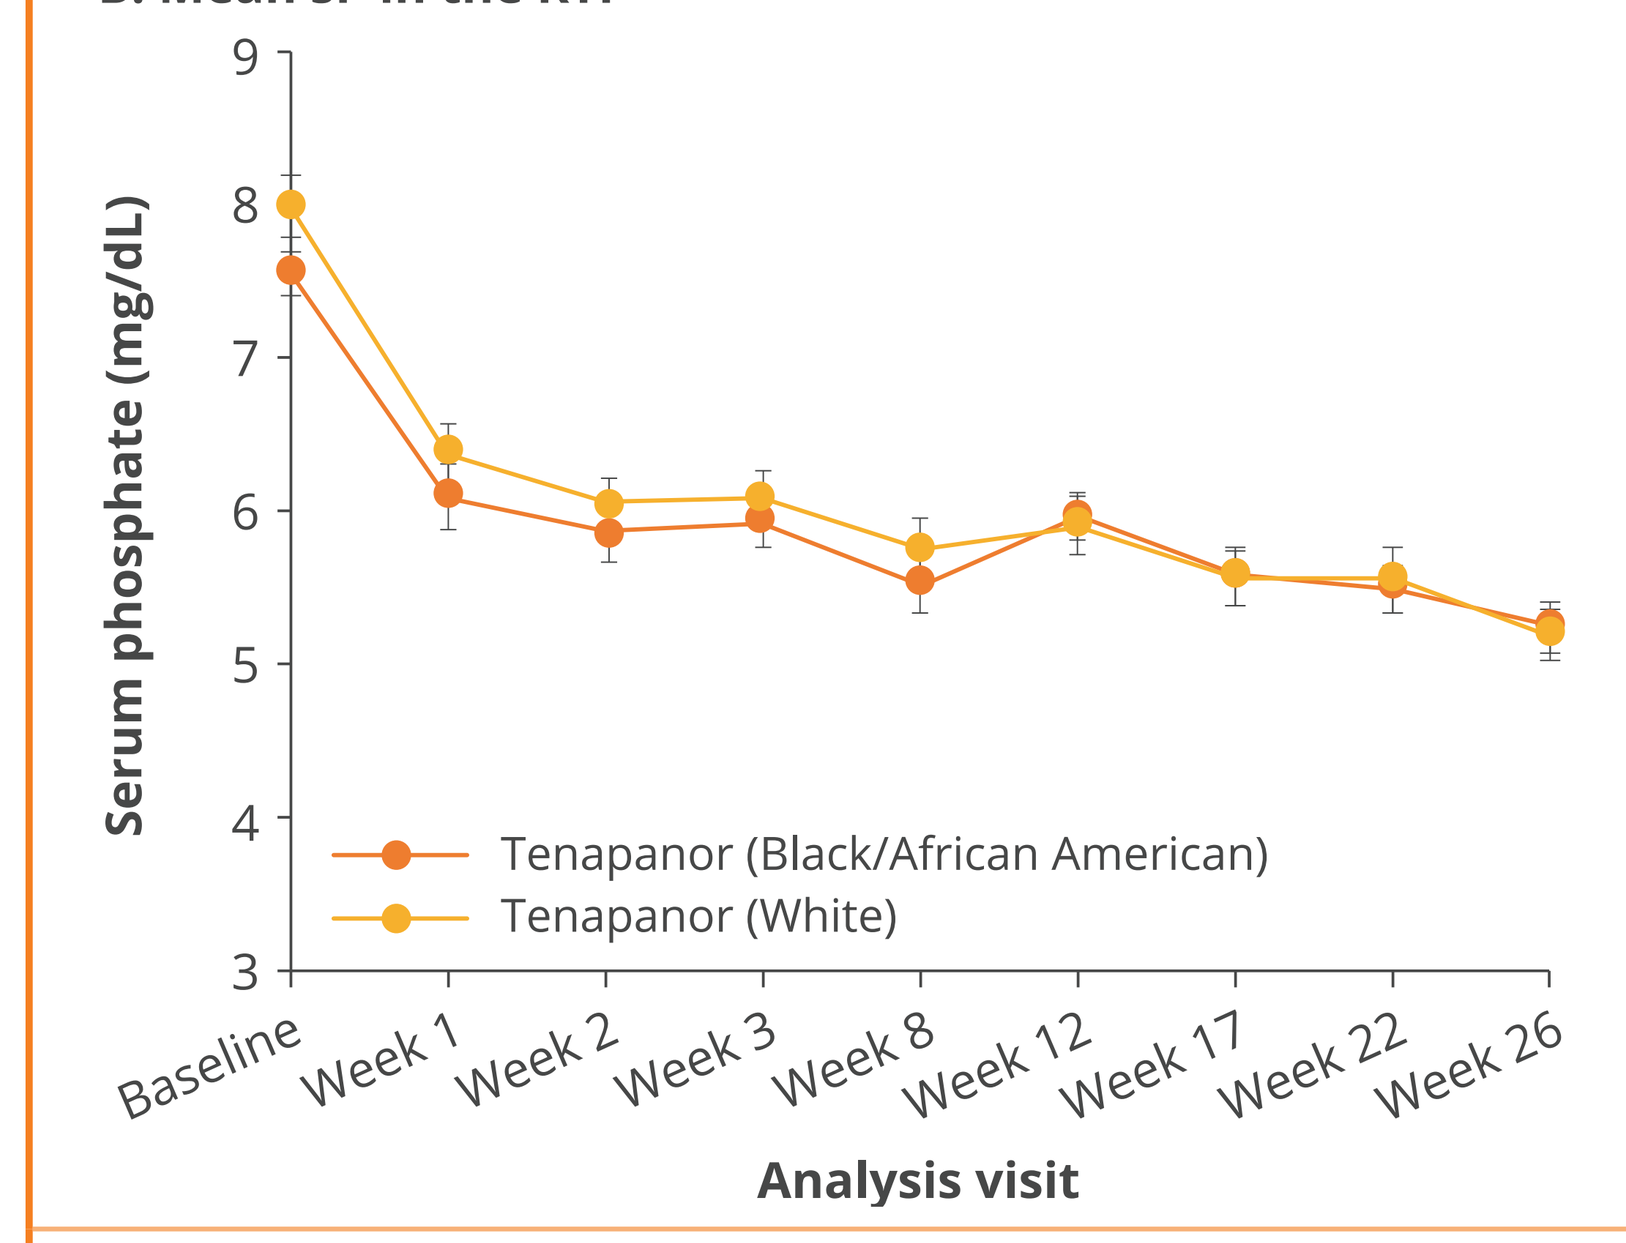
Serum phosphate (mg/dL)
9
8
7
6
5
4
3
Tenapanor (Black/African American)
Tenapanor (White)
Baseline
Week 1
Week 2
Week 3
Week 8
Week 12
Week 17
Week 22
Week 26
Analysis visit

C. Mean sP in the RWP

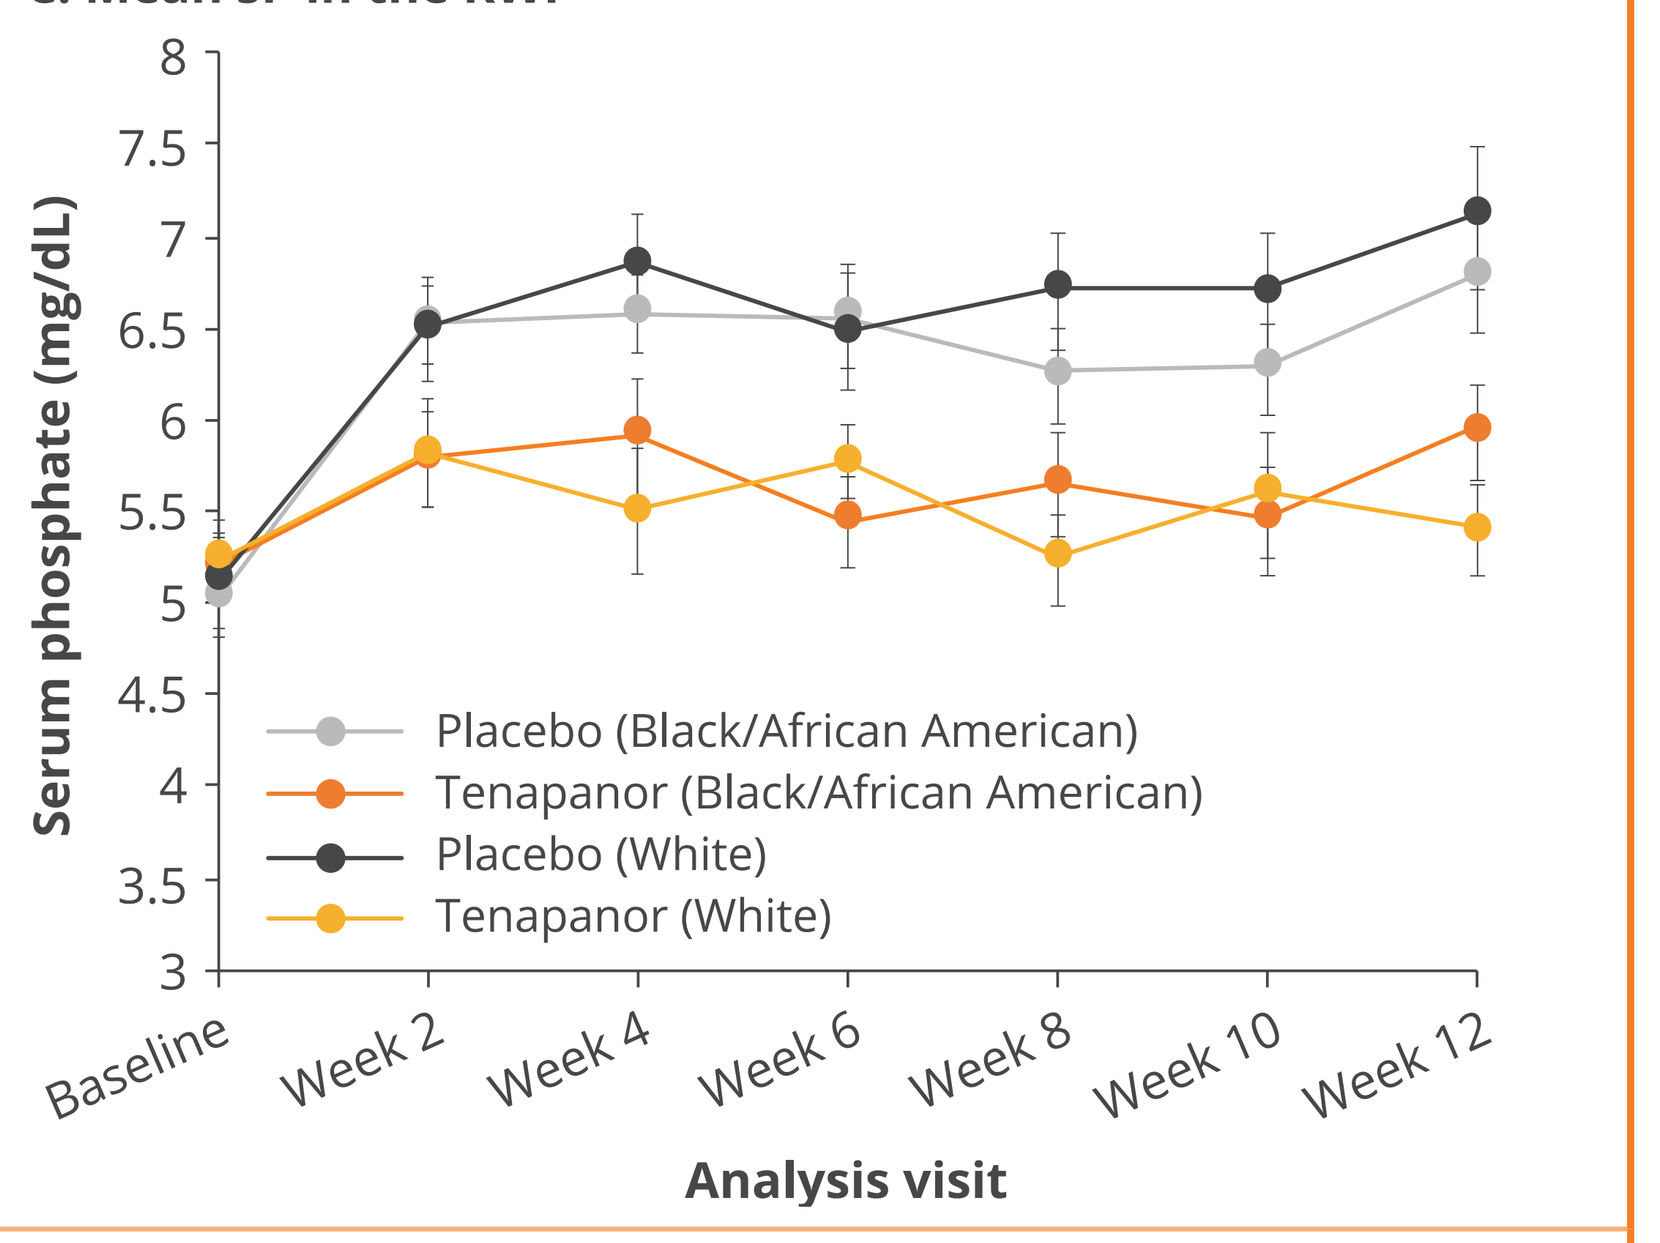
Serum phosphate (mg/dL)
8
7.5
7
6.5
6
5.5
5
4.5
4
3.5
3
Placebo (Black/African American)
Tenapanor (Black/African American)
Placebo (White)
Tenapanor (White)
Baseline
Week 2
Week 4
Week 6
Week 8
Week 10
Week 12
Analysis visit

The LS mean estimates presented in Figure 2A are obtained from the pre-specified ANCOVA for the primary efficacy endpoint. The mean ± standard error presented in Figure 2C are obtained from the descriptive summary of the sP level by analysis visit during the RWP.
[a]Responder population defined as patients who achieved an sP level <5.5 mg/dL.
ANCOVA, analysis of covariance; LS, least squares; LSM, least squares mean; RTP, randomized treatment period; RWP, randomized withdrawal period; sP, serum phosphate.

AMPLIFY

- In AMPLIFY, LS mean difference (95% CI) between tenapanor + phosphate binder and placebo + phosphate binder in sP change at week 4 was −0.60 (−1.17, −0.03) mg/dL among Black/African American patients and −0.64 (−1.16, −0.12) mg/dL among White patients (**Figure 3A**).
- No significant differences in mean sP were observed between Black/African American and White patients (**Figure 3B**).

Figure 3. sP Change From Baseline to Week 4 by Race in AMPLIFY (Full Analysis Set)

A. LS mean difference in sP

Subgroup	N	Placebo	Tenapanor	LSM change difference	*P* value
All subjects	235	−0.19	−0.84	−0.65 (−1.01, −0.29)	0.0004
Race					
White	117	−0.22	−0.86	−0.64 (−1.16, −0.12)	0.0170
Black or African American	101	−0.22	−0.82	−0.60 (−1.17, −0.03)	0.0395

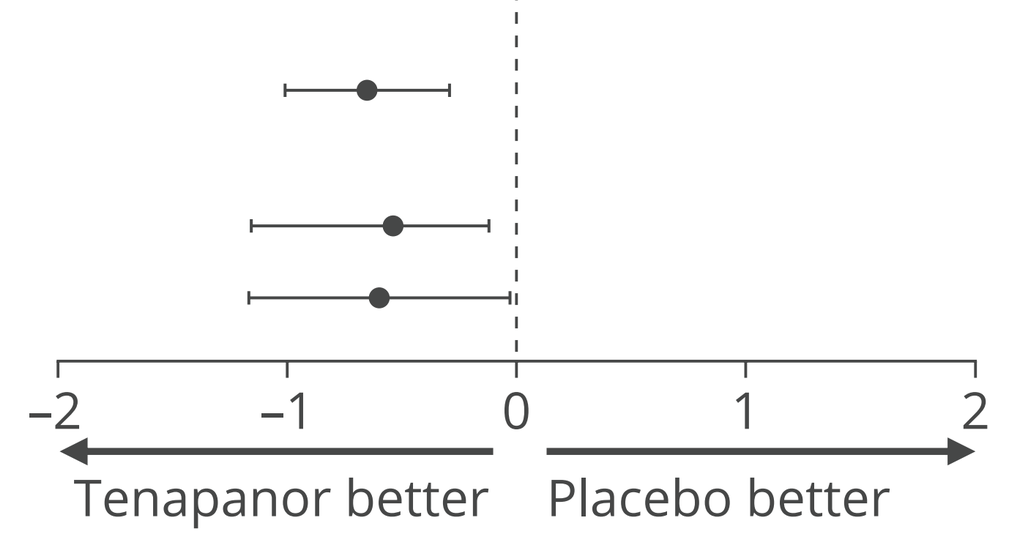
LS mean change difference and 95% CI
−2
−1
0
1
2
Tenapanor better
Placebo better

B. Mean sP

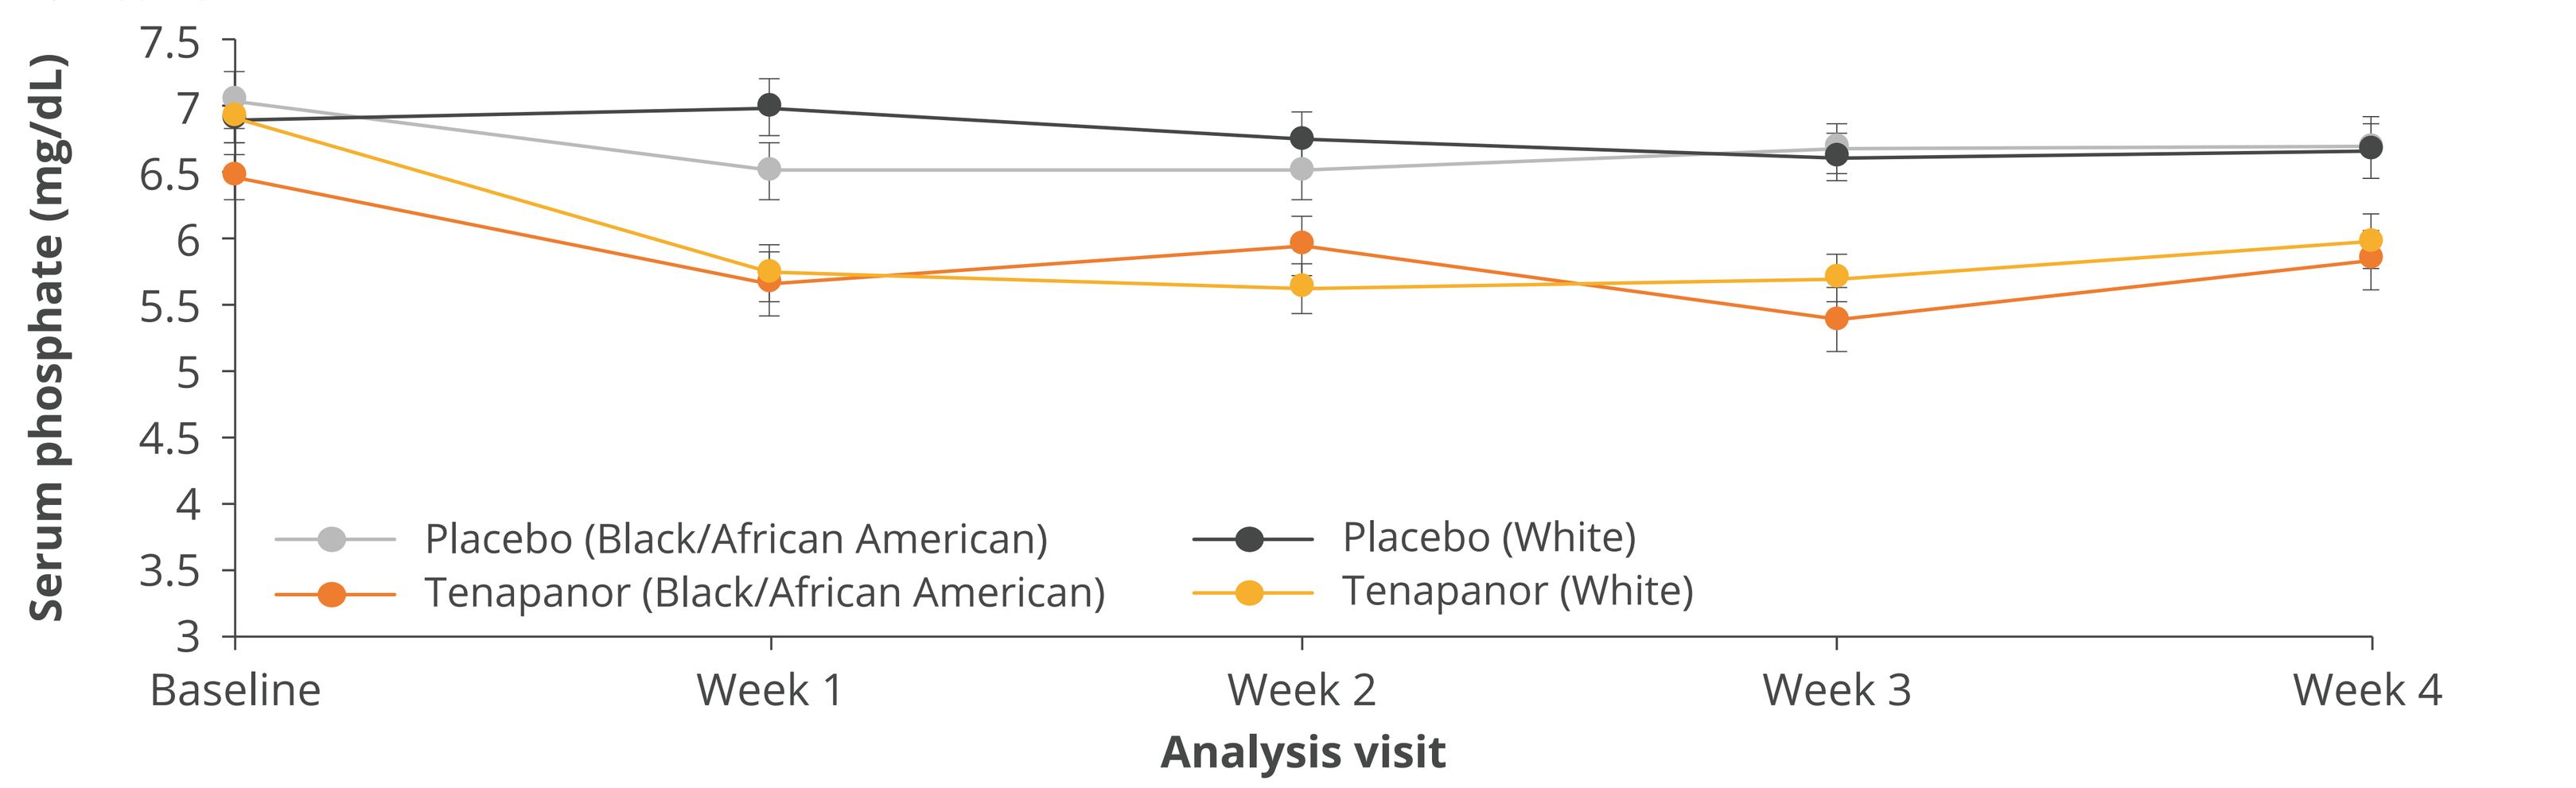
Serum phosphate (mg/dL)
7.5
7
6.5
6
5.5
5
4.5
4
3.5
3
Placebo (Black/African American)
Tenapanor (Black/African American)
Placebo (White)
Tenapanor (White)
Baseline
Week 1
Week 2
Week 3
Week 4
Analysis visit

The LS mean estimates presented in Figure 3A are obtained from the pre-specified mixed-effects model for repeated measures. The mean ± standard error presented in Figure 3B are obtained from the descriptive summary of the sP level by analysis visit during the RTP.
LS, least squares; LSM, least squares mean; RTP, randomized treatment period; sP, serum phosphate.

Safety

- Safety findings during the RTP for the safety analysis sets in NCT02675998,[7] PHREEDOM,[8] and AMPLIFY[9] were previously reported.
- Diarrhea was the most commonly reported adverse event in all 3 studies. It generally occurred early in treatment and was mild to moderate in severity.
- During the NCT02675998 RTP, Black/African American patients had a lower incidence of diarrhea compared with White patients (**Table 2**). However, the incidence of diarrhea was similar between Black/African American and White patients during PHREEDOM RTP and AMPLIFY (**Table 2**).

Table 2. Summary of TEAEs During RTP by Race (Safety Analysis Set)

	NCT02675998[a]		PHREEDOM				AMPLIFY			
	Tenapanor (N=210)		Sevelamer (N=130)		Tenapanor (N=384)		Placebo+PB (N=110)		Tenapanor+PB (N=109)	
Adverse event	White (n=85)	Black (n=125)	White (n=70)	Black (n=60)	White (n=189)	Black (n=195)	White (n=60)	Black (n=50)	White (n=57)	Black (n=52)
Any AE (%)	66 (78)	65 (52)	47 (67)	38 (63)	159 (84)	148 (76)	19 (32)	13 (26)	28 (49)	27 (52)
Any SAE (%)	7 (8)	12 (10)	17 (24)	15 (25)	38 (20)	30 (15)	2 (3)	2 (4)	1 (2)	2 (4)
Any AE leading to death (%)	1 (1)	0	0	0	1 (0.5)	0	0	0	0	0
Any AE leading to study drug discontinuation (%)	18 (21)	16 (13)	0	1 (2)	52 (28)	38 (19)	1 (2)	1 (2)	1 (2)	4 (8)
TEAEs reported by ≥5% of participants										
Diarrhea	54 (63.5)	29 (23.2)	6 (8.6)	4 (6.7)	101 (53.4)	100 (51.3)	4 (6.7)	4 (8.0)	24 (42.1)	22 (42.3)
Vomiting	2 (2.4)	7 (5.6)	2 (2.9)	4 (6.7)	9 (4.8)	4 (2.1)	3 (5.0)	1 (2.0)	1 (1.8)	2 (3.8)
Nausea	1 (1.2)	4 (3.2)	4 (5.7)	2 (3.3)	8 (4.2)	6 (3.1)	1 (1.7)	2 (4.0)	2 (3.5)	4 (7.7)
Constipation	2 (2.4)	1 (0.8)	3 (4.3)	3 (5.0)	2 (1.1)	0	2 (3.3)	1 (2.0)	0	0
Hyperphosphatemia	5 (5.9)	7 (5.6)	1 (1.4)	1 (1.7)	12 (6.3)	12 (6.2)	1 (1.7)	0	0	1 (1.9)
Hyperkalemia	1 (1.2)	2 (1.6)	4 (5.7)	3 (5.0)	12 (6.3)	6 (3.1)	3 (5.0)	0	0	0
Fluid overload	2 (2.4)	2 (1.6)	4 (5.7)	4 (6.7)	3 (1.6)	5 (2.6)	0	1 (2.0)	0	0
Cellulitis	5 (5.9)	1 (0.8)	2 (2.9)	0	4 (2.1)	1 (0.5)	0	0	0	0
Bronchitis	1 (1.2)	0	4 (5.7)	0	6 (3.2)	0	0	0	0	0
Pneumonia	1 (1.2)	2 (1.6)	5 (7.1)	2 (3.3)	3 (1.6)	2 (1.0)	1 (1.7)	0	1 (1.8)	0

Data are n (%). [a]There was no placebo arm during the RTP of this study.
AE, adverse event; PB, phosphate binder; RTP, randomized treatment period; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

Conclusions

Results across 3 pivotal studies of tenapanor for hyperphosphatemia demonstrate that tenapanor treatment consistently lowers sP levels in patients with CKD on dialysis regardless of Black/African American or White race.

Tenapanor has a consistent and acceptable safety profile for patients with CKD on dialysis regardless of race, with diarrhea being the most common adverse event.

Disclosures

Stuart M. Sprague receives research grants from and is a consultant for Ardelyx, Inc. Yang Yang, Suling Zhao, Susan Edelstein, and David M. Spiegel are employees of Ardelyx, Inc.

References

1. Clegg DJ et al. *Nephrol Renal Dis*. 2022;7:1-7.
2. Guedes M et al. *Kidney Med*. 2022;5:100584.
3. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
4. Spencer AG et al. *Sci Transl Med*. 2014;6:227ra36.
5. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
6. Chu CD et al. *JAMA Netw Open*. 2021;4:e2127014.
7. Block GA et al. *J Am Soc Nephrol*. 2019;30:641-52.
8. Block GA et al. *Kidney 360*. 2021;2:1600-10.
9. Pergola P et al. *J Am Soc Nephrol*. 2021;32:1465-73.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr. Sprague can be contacted for further information on this study at SSprague@northshore.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

National Kidney Foundation Spring Clinical Meetings (SCM23); April 11-15, 2023; Austin, TX, USA

MED-US--2400028

Electronic Certificate

Version: 2 . 0

Document Number: MED-US--2400028

Document Name: 1175750_NKF2023_RaceSubgroups_Poster_FINAL

Country: United States

Product: Tenapanor-HP

Type: External Communication

Sub Type: MSL / Field Material

Classification: Poster

Certification Statement
We certify that the final electronic form of this material is in accordance with the regulations set forth by the health authority for the country of this document, and is a fair and truthful presentation of the facts about the product.

Role	Signature
Role: Approver Workflow: Medical Coordinator: Assess Outcome Outcome: Approved for Use	Name: May Luong (mluong@ardelyx.com) Date: 26-Feb-2024 20:34:39 GMT+0000 Statement: Assess the outcome of review, then click the complete button for this task to select the next workflow.