TH-PO162

Reduction of Serum Phosphate With Tenapanor in Patients With Chronic Kidney Disease on Maintenance Dialysis With Severe Hyperphosphatemia

Stuart M. Sprague,[1] Yang Yang,[2] David M. Spiegel,[2] David P. Rosenbaum[2]

[1]NorthShore University HealthSystem-University of Chicago Pritzker School of Medicine, Chicago, IL, USA; [2]Ardelyx, Inc., Waltham, MA, USA

Background

- Tenapanor is a first-in-class, investigational phosphate absorption inhibitor (PAI). Preclinical studies have shown that tenapanor blocks paracellular phosphate absorption in the gastrointestinal tract by local inhibition of intestinal sodium/hydrogen exchanger isoform 3 (NHE3), which increases transepithelial electrical resistance and decreases intestinal permeability to phosphate.[1] Inhibition of NH3 by tenapanor also leads to increased stool sodium and water content in animals and humans.[1,2]
- In patients with chronic kidney disease, increasing serum phosphate (sP) concentrations are associated with increasing risk of mortality, hospitalization, and cardiovascular morbidity and mortality.[3-8]
- Tenapanor met the primary sP-lowering endpoint in 2 phase 3 monotherapy trials (NCT02675998 and PHREEDOM [NCT03427125]) and demonstrated an acceptable safety and tolerability profile in patients with hyperphosphatemia undergoing maintenance dialysis.[9,10]
- Tenapanor also improved control of hyperphosphatemia in patients on maintenance dialysis when used in combination with phosphate binders compared with placebo plus phosphate binders in the phase 3 AMPLIFY trial (NCT03824587).[11]
- Using data from the NCT02675998 and PHREEDOM trials, we evaluated the effect of tenapanor monotherapy in patients on maintenance dialysis with severe hyperphosphatemia, which we defined as having a baseline sP ≥7.5 mg/dL.

Methods

- The study designs and patient populations of the NCT02675998 and PHREEDOM trials have been reported previously.[9,10]
 - Both trials enrolled patients on maintenance dialysis with sP ≥6.0 and ≤10.0 mg/dL and a ≥1.5 mg/dL increase in sP after phosphate binder washout.
 - In the NCT02675998 trial, patients were randomized (1:1:1) to receive tenapanor 3, 10, or 30 mg twice a day (bid) for an 8-week open-label randomized treatment period (RTP); the 30 mg dose could be down-titrated stepwise during the first 4 weeks of the RTP based on gastrointestinal tolerability. After the RTP, patients were rerandomized (1:1) to continue their tenapanor dose or switch to placebo in a 4-week randomized withdrawal period (RWP).
 - In PHREEDOM, patients were randomized (3:1) to tenapanor starting at a dose of 30 mg bid for 26 weeks during the RTP or sevelamer carbonate (hereafter referred to as sevelamer) per the label for 52 weeks. Those who completed 26 weeks of tenapanor treatment were rerandomized (1:1) to continue their tenapanor dose or switch to placebo for a 12-week RWP, followed by a 14-week open-label safety extension.
 - Titration of the tenapanor dose in 10 mg increments was allowed during the RTP and safety extension, down to a minimum of 10 mg bid or up to a maximum of 30 mg bid. Sevelamer dose could be titrated up or down, as needed, based on the community standard of care.
- In this nonprespecified, post hoc analysis we evaluated the sP change and safety during the RTP of each study in patients with severe hyperphosphatemia at baseline (sP ≥7.5 mg/dL).
- For this analysis, we assessed sP change in evaluable tenapanor- or sevelamer-treated patients during the first 26 weeks (ie, RTP) of the study.
 - For the tenapanor-treated populations in NCT02675998 and PHREEDOM, this included all patients who were randomized to tenapanor, received ≥1 dose of tenapanor, and had ≥1 postbaseline sP assessment during the RTP of each respective study.
 - For the sevelamer-treated population in PHREEDOM, this included all patients who were randomized to sevelamer, received ≥1 dose of sevelamer, and had ≥1 postbaseline sP assessment during the first 26 weeks of treatment.
 - The sevelamer group was a safety control in the PHREEDOM trial, and the study was not intended or powered to make efficacy comparisons between the tenapanor and sevelamer treatment groups.
- In an additional nonprespecified, post hoc analysis of patients from PHREEDOM who completed the 26-week RTP or the 52-week study, we evaluated the percentage of patients who achieved sP reductions ≥1.2, ≥1.5, or ≥2.0 mg/dL or who achieved sP <5.5 mg/dL.
- Safety was evaluated in the safety analysis set, which included all patients who received ≥1 dose of study treatment in the RTP.

Results

Patients

- In the NCT02675998 trial, 107 of 215 patients had severe hyperphosphatemia at baseline across the 3 dose groups. In the 30 mg dose group, 37 of 72 patients had severe hyperphosphatemia.
- In PHREEDOM, 204 of 407 tenapanor-treated patients had severe hyperphosphatemia at baseline, and 52 of 136 sevelamer-treated patients had severe hyperphosphatemia at baseline.
- Demographic and baseline disease characteristics are reported in **Table 1**.

Table 1. Patient Demographics and Baseline Characteristics for Patients With Severe Hyperphosphatemia (sP ≥7.5 mg/dL)

	NCT02675998 trial		PHREEDOM	
	All doses[a] (n=107)	30 mg dose (n=37)	Tenapanor[b] (n=204)	Sevelamer (n=52)
Age, mean years (SD)	54.2 (12.1)	54.0 (12.7)	55.5 (12.8)	56.6 (12.6)
Sex, n (%)				
Male	62 (57.9)	24 (64.9)	130 (63.7)	32 (61.5)
Female	45 (42.1)	13 (35.1)	74 (36.3)	20 (38.5)
Race, n (%)				
Black or African American	59 (55.1)	21 (56.8)	80 (39.2)	23 (44.2)
White	44 (41.1)	16 (43.2)	107 (52.5)	27 (51.9)
Asian	1 (0.9)	0	10 (4.9)	2 (3.8)
American Indian or Alaska Native	2 (1.9)	0	4 (2.0)	0
Other[c]	1 (0.9)	0	3 (1.5)	0
Ethnicity, n (%)				
Not Hispanic or Latino	85 (79.4)	26 (70.3)	136 (66.7)	40 (76.9)
Hispanic or Latino	22 (20.6)	11 (29.7)	66 (32.4)	12 (23.1)
Unknown	0	0	2 (1.0)	0
BMI, mean kg/m² (SD)	32.5 (8.4)	34.0 (8.5)	32.1 (7.6)	32.0 (13.6)
Type of maintenance dialysis, n (%)				
Hemodialysis	107 (100)	37 (100)	179 (87.8)	44 (84.6)
Peritoneal dialysis	0	0	25 (12.3)	8 (15.4)
Baseline sP, mean mg/dL (SD)	8.8 (1.1)	8.7 (1.1)	8.5 (1.0)	8.7 (1.0)
Baseline iFGF23, mean pg/mL (SD)	13,400.4 (14,139.2)	14,397.8 (12,951.1)	17,976.3 (17,700.1)	15,921.7 (16,325.8)
Baseline PTH, mean pg/mL (SD)	418.1 (243.6)	373.7 (197.1)	454.0 (260.3)	465.1 (296.0)
Most recent Kt/V, mean (SD)	1.6 (0.3)	1.6 (0.3)	1.6 (0.4)	1.7 (0.5)

Data are reported for patients with severe hyperphosphatemia from the safety analysis set.
[a]Patients in the NCT02675998 trial received tenapanor at a dose of 3 mg (fixed), 10 mg (fixed), or 30 mg (with dose titration) bid. [b]Tenapanor dose in PHREEDOM started at 30 mg bid and could be down-titrated. [c]Native Hawaiian or other Pacific Islander or other.
bid, twice a day; BMI, body mass index; iFGF23, intact fibroblast growth factor 23; Kt/V, a marker of dialysis adequacy; PTH, parathyroid hormone; sP, serum phosphate.

Reduction in sP in patients with severe hyperphosphatemia

- Mean sP decreased over time in patients with severe hyperphosphatemia in both studies (**Figures 1 and 2**).
- In PHREEDOM, the least-squares (LS) mean reductions in sP among patients with severe hyperphosphatemia during the RTP were similar between the tenapanor and sevelamer groups (**Figure 3**).
- In PHREEDOM, among patients with severe hyperphosphatemia who completed the 26-week RTP, a similar trend was observed in the proportions of tenapanor- and sevelamer-treated patients with sP reductions of ≥1.2, ≥1.5, or ≥2.0 mg/dL or who achieved sP <5.5 mg/dL (**Figure 4**). A similar pattern was observed for patients with severe hyperphosphatemia who completed all 52 weeks of the study.

Figure 1. Mean sP Over Time During the RTP of the NCT02675998 Trial in Patients with Severe Hyperphosphatemia (sP ≥7.5 mg/dL)

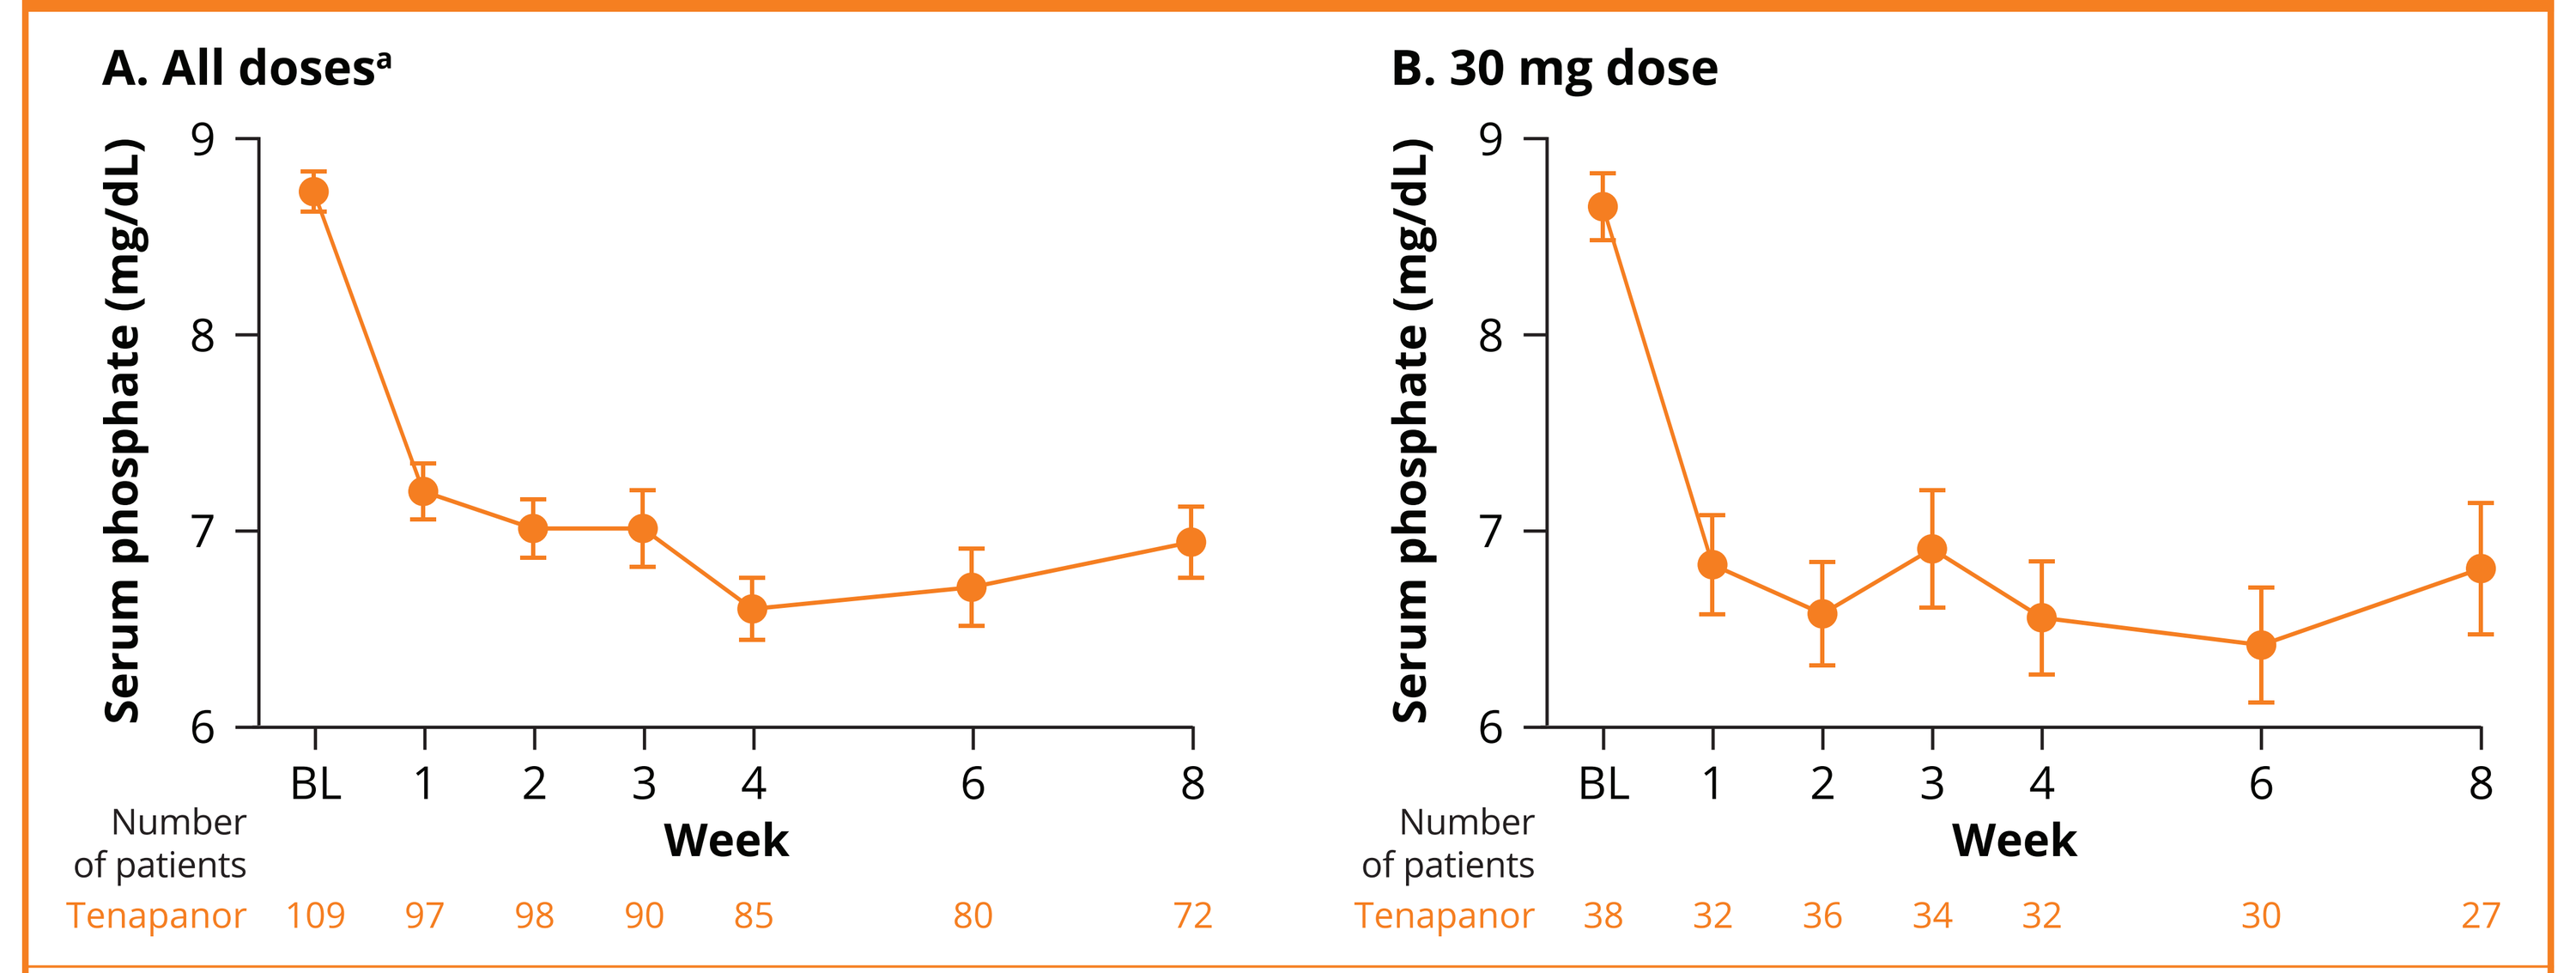
A. All doses[a]
B. 30 mg dose
Serum phosphate (mg/dL)
9
8
7
6
BL 1 2 3 4 6 8
Week
Number of patients
Tenapanor 109 97 98 90 85 80 72
Tenapanor 38 32 36 34 32 30 27

Error bars show SE.
[a]Patients in the NCT02675998 trial received tenapanor at a dose of 3 mg (fixed), 10 mg (fixed), or 30 mg (with dose titration) bid.
bid, twice a day; BL, baseline; RTP, randomized treatment period; sP, serum phosphate.

Figure 2. Mean sP Over Time During the RTP of the PHREEDOM Trial in Patients with Severe Hyperphosphatemia (sP ≥7.5 mg/dL)

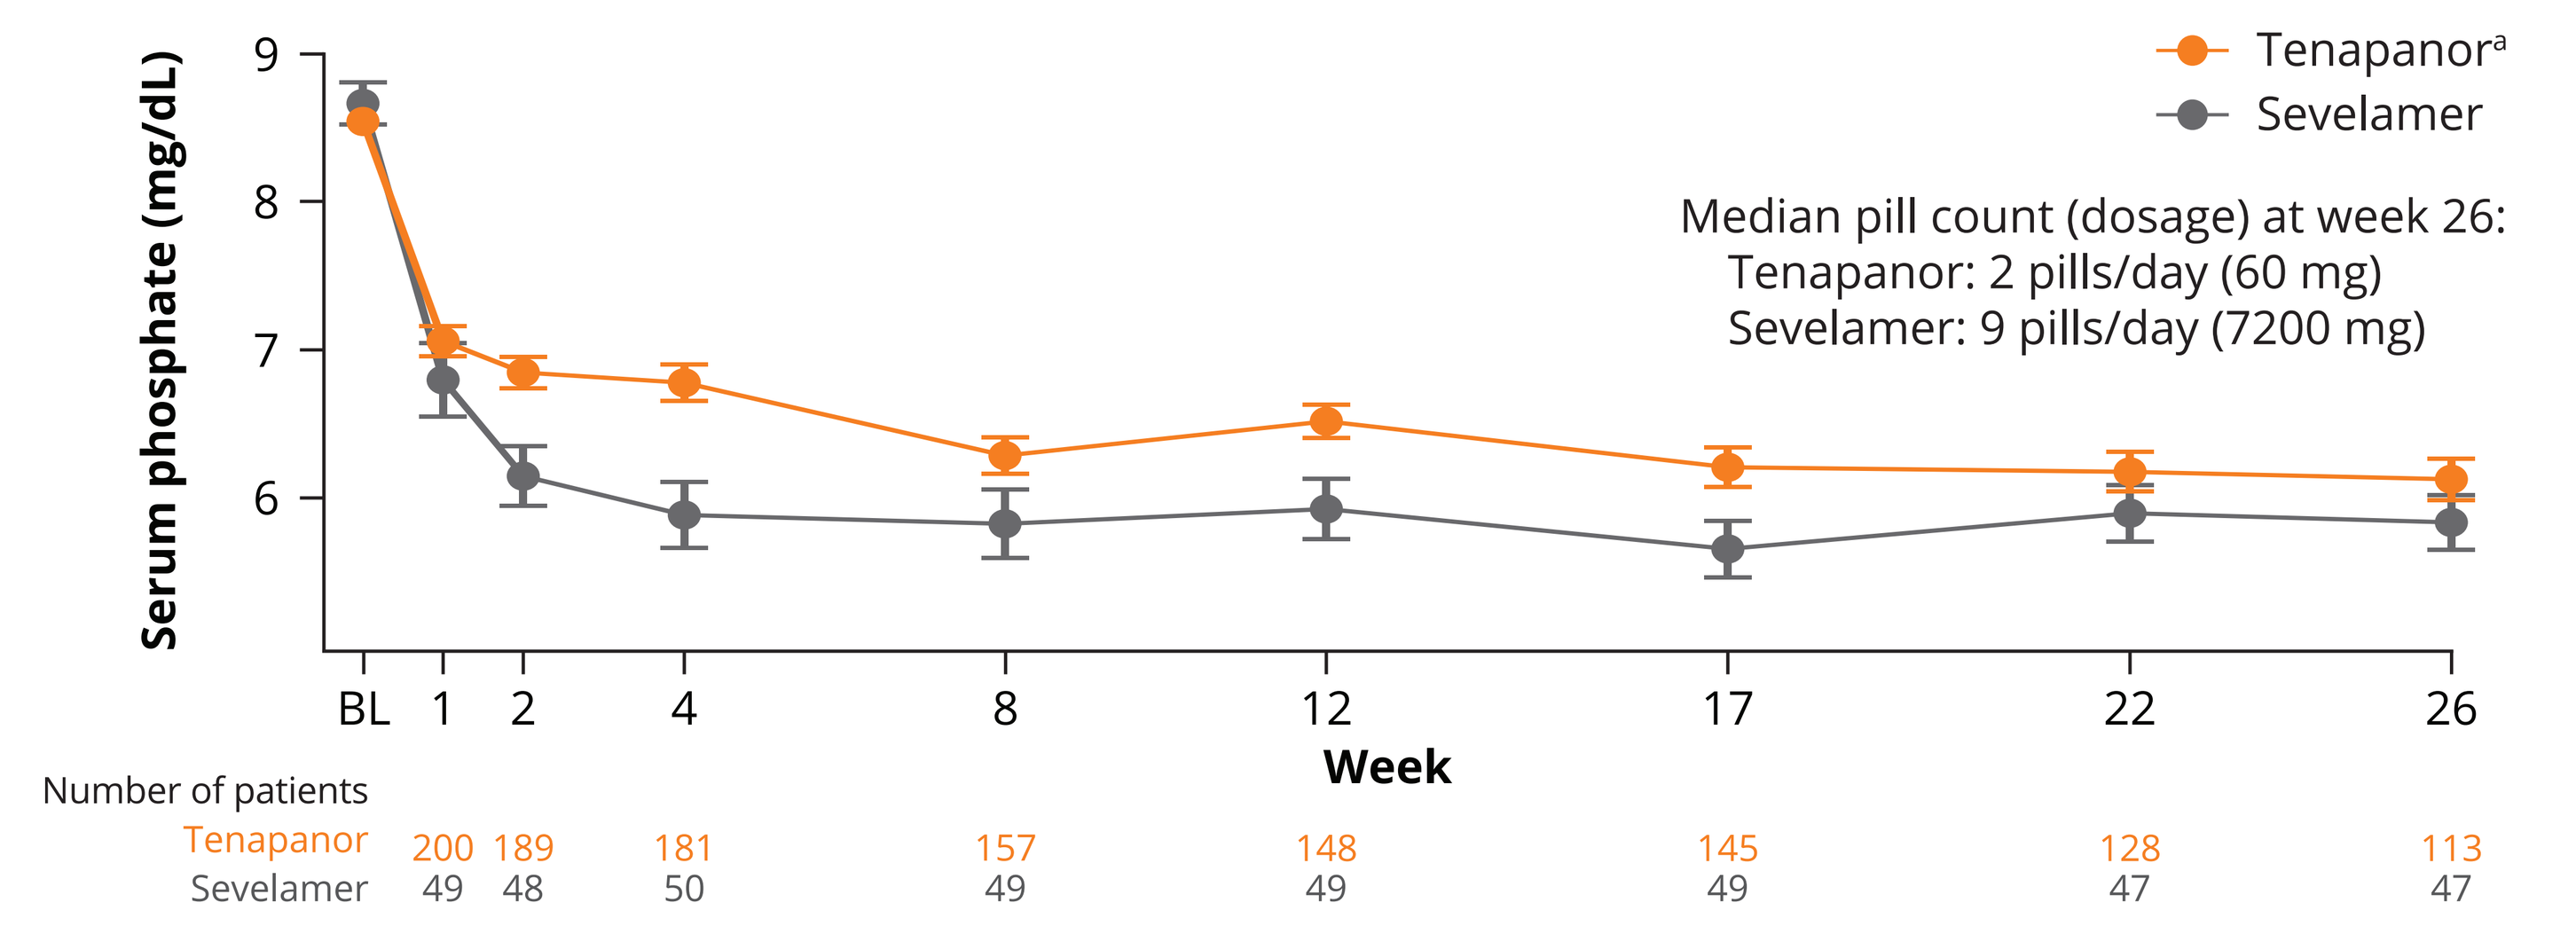
Serum phosphate (mg/dL)
9
8
7
6
Tenapanor[a]
Sevelamer
Median pill count (dosage) at week 26:
Tenapanor: 2 pills/day (60 mg)
Sevelamer: 9 pills/day (7200 mg)
BL 1 2 4 8 12 17 22 26
Week
Number of patients
Tenapanor 200 189 181 157 148 145 128 113
Sevelamer 49 48 50 49 49 49 47 47

Error bars show SE.
[a]Tenapanor dose in PHREEDOM started at 30 mg bid and could be down-titrated.
bid, twice a day; BL, baseline; RTP, randomized treatment period; sP, serum phosphate.

Figure 3. LS Mean Change From Baseline in sP Over Time During the RTP of the PHREEDOM Trial in Patients with Severe Hyperphosphatemia (sP ≥7.5 mg/dL)

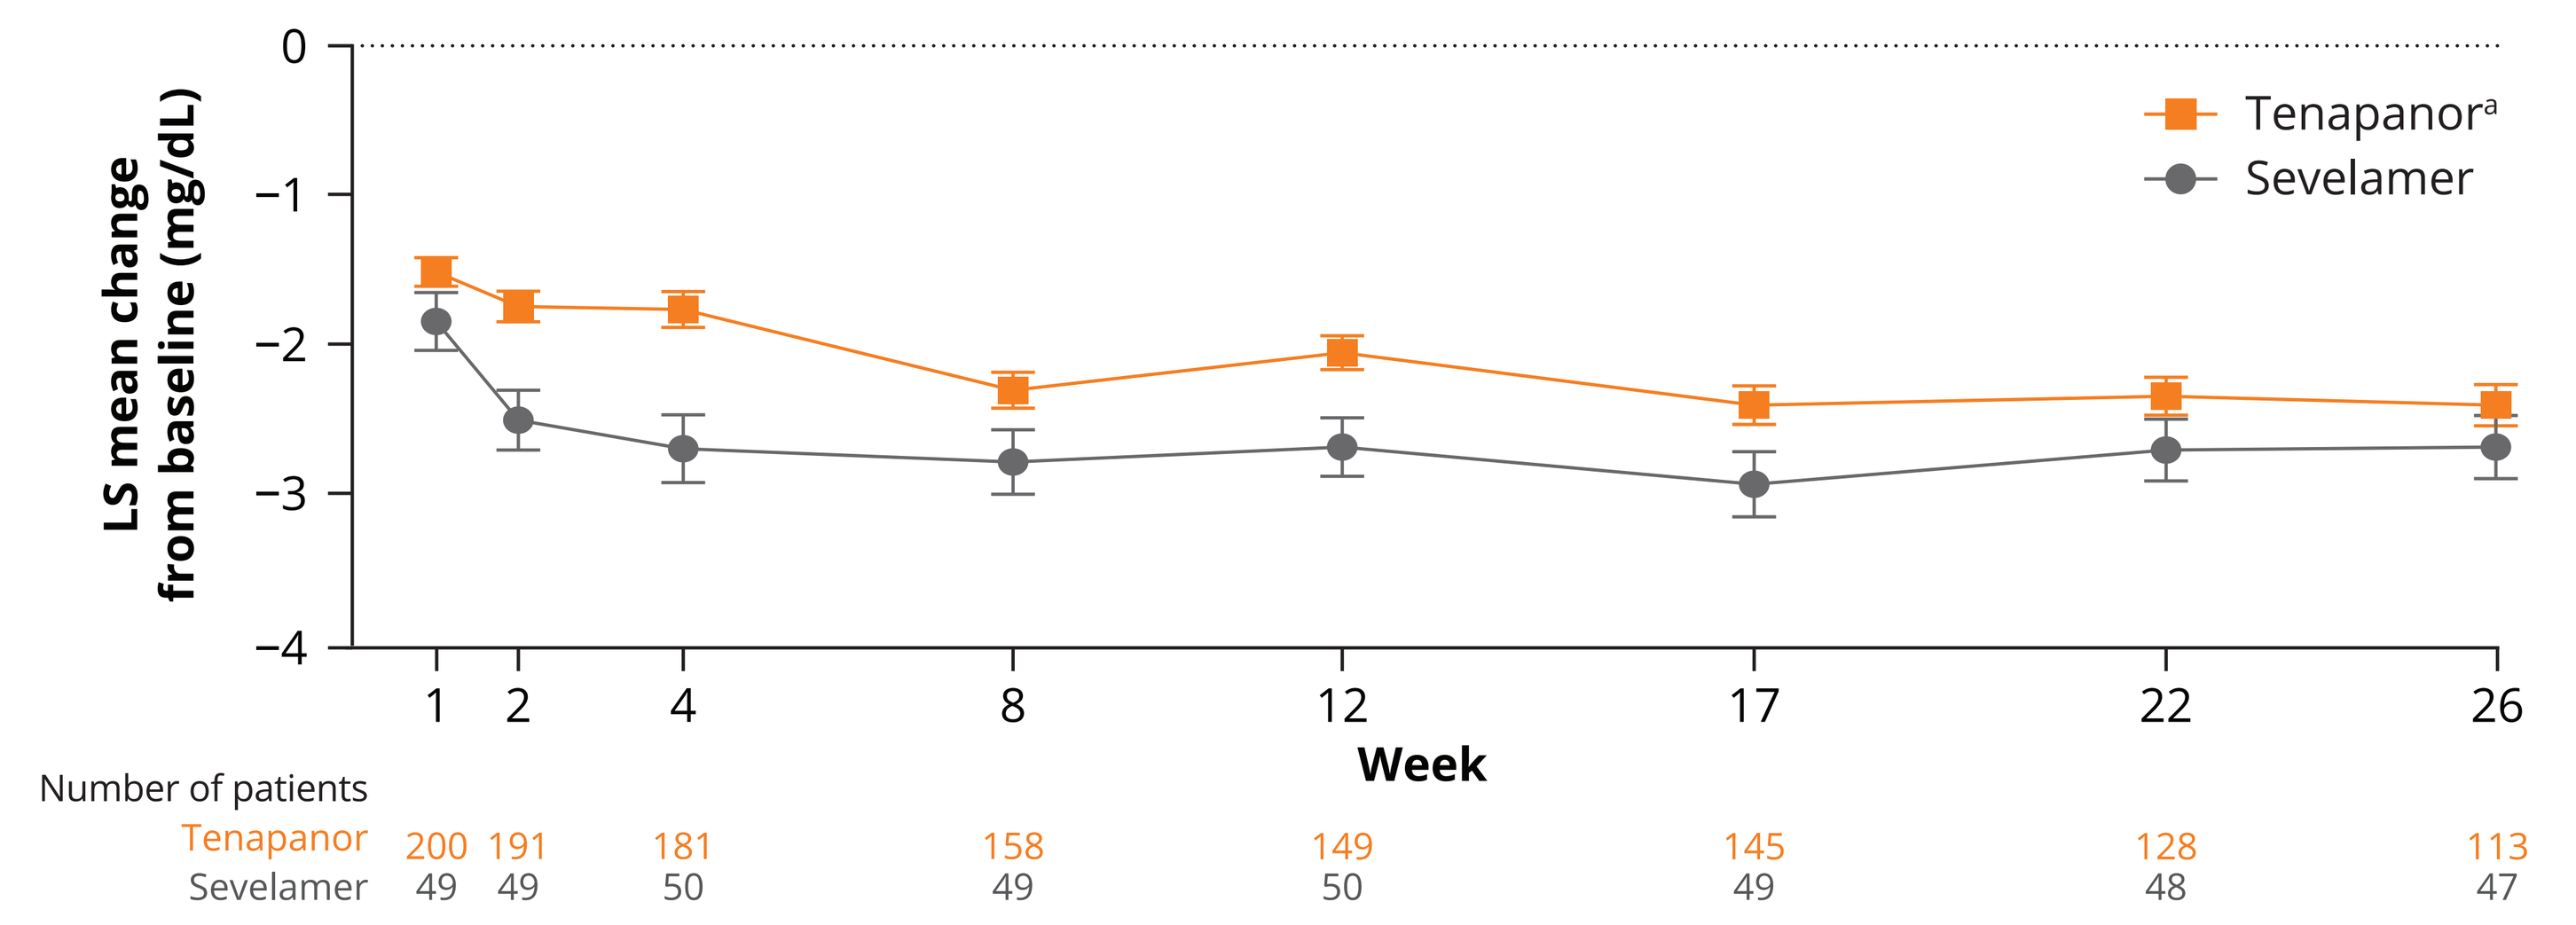
LS mean change from baseline (mg/dL)
0
−1
−2
−3
−4
Tenapanor[a]
Sevelamer
1 2 4 8 12 17 22 26
Week
Number of patients
Tenapanor 200 191 181 158 149 145 128 113
Sevelamer 49 49 50 49 50 49 48 47

Error bars show SE. LS means, SEs, and *P* values come from an ANCOVA model with treatment and pooled site as factors and period-level baseline value as a covariate. Only observed data were included in the analysis.
[a]Tenapanor dose in PHREEDOM started at 30 mg bid and could be down-titrated. **P*<0.05; ***P*<0.01; ****P*<0.001.
bid, twice a day; ANCOVA, analysis of covariance; LS, least squares; RTP, randomized treatment period.

Figure 4. Additional Measures of Treatment Effect in Patients With Severe Hyperphosphatemia Who Completed 26 or 52 Weeks of the PHREEDOM Trial

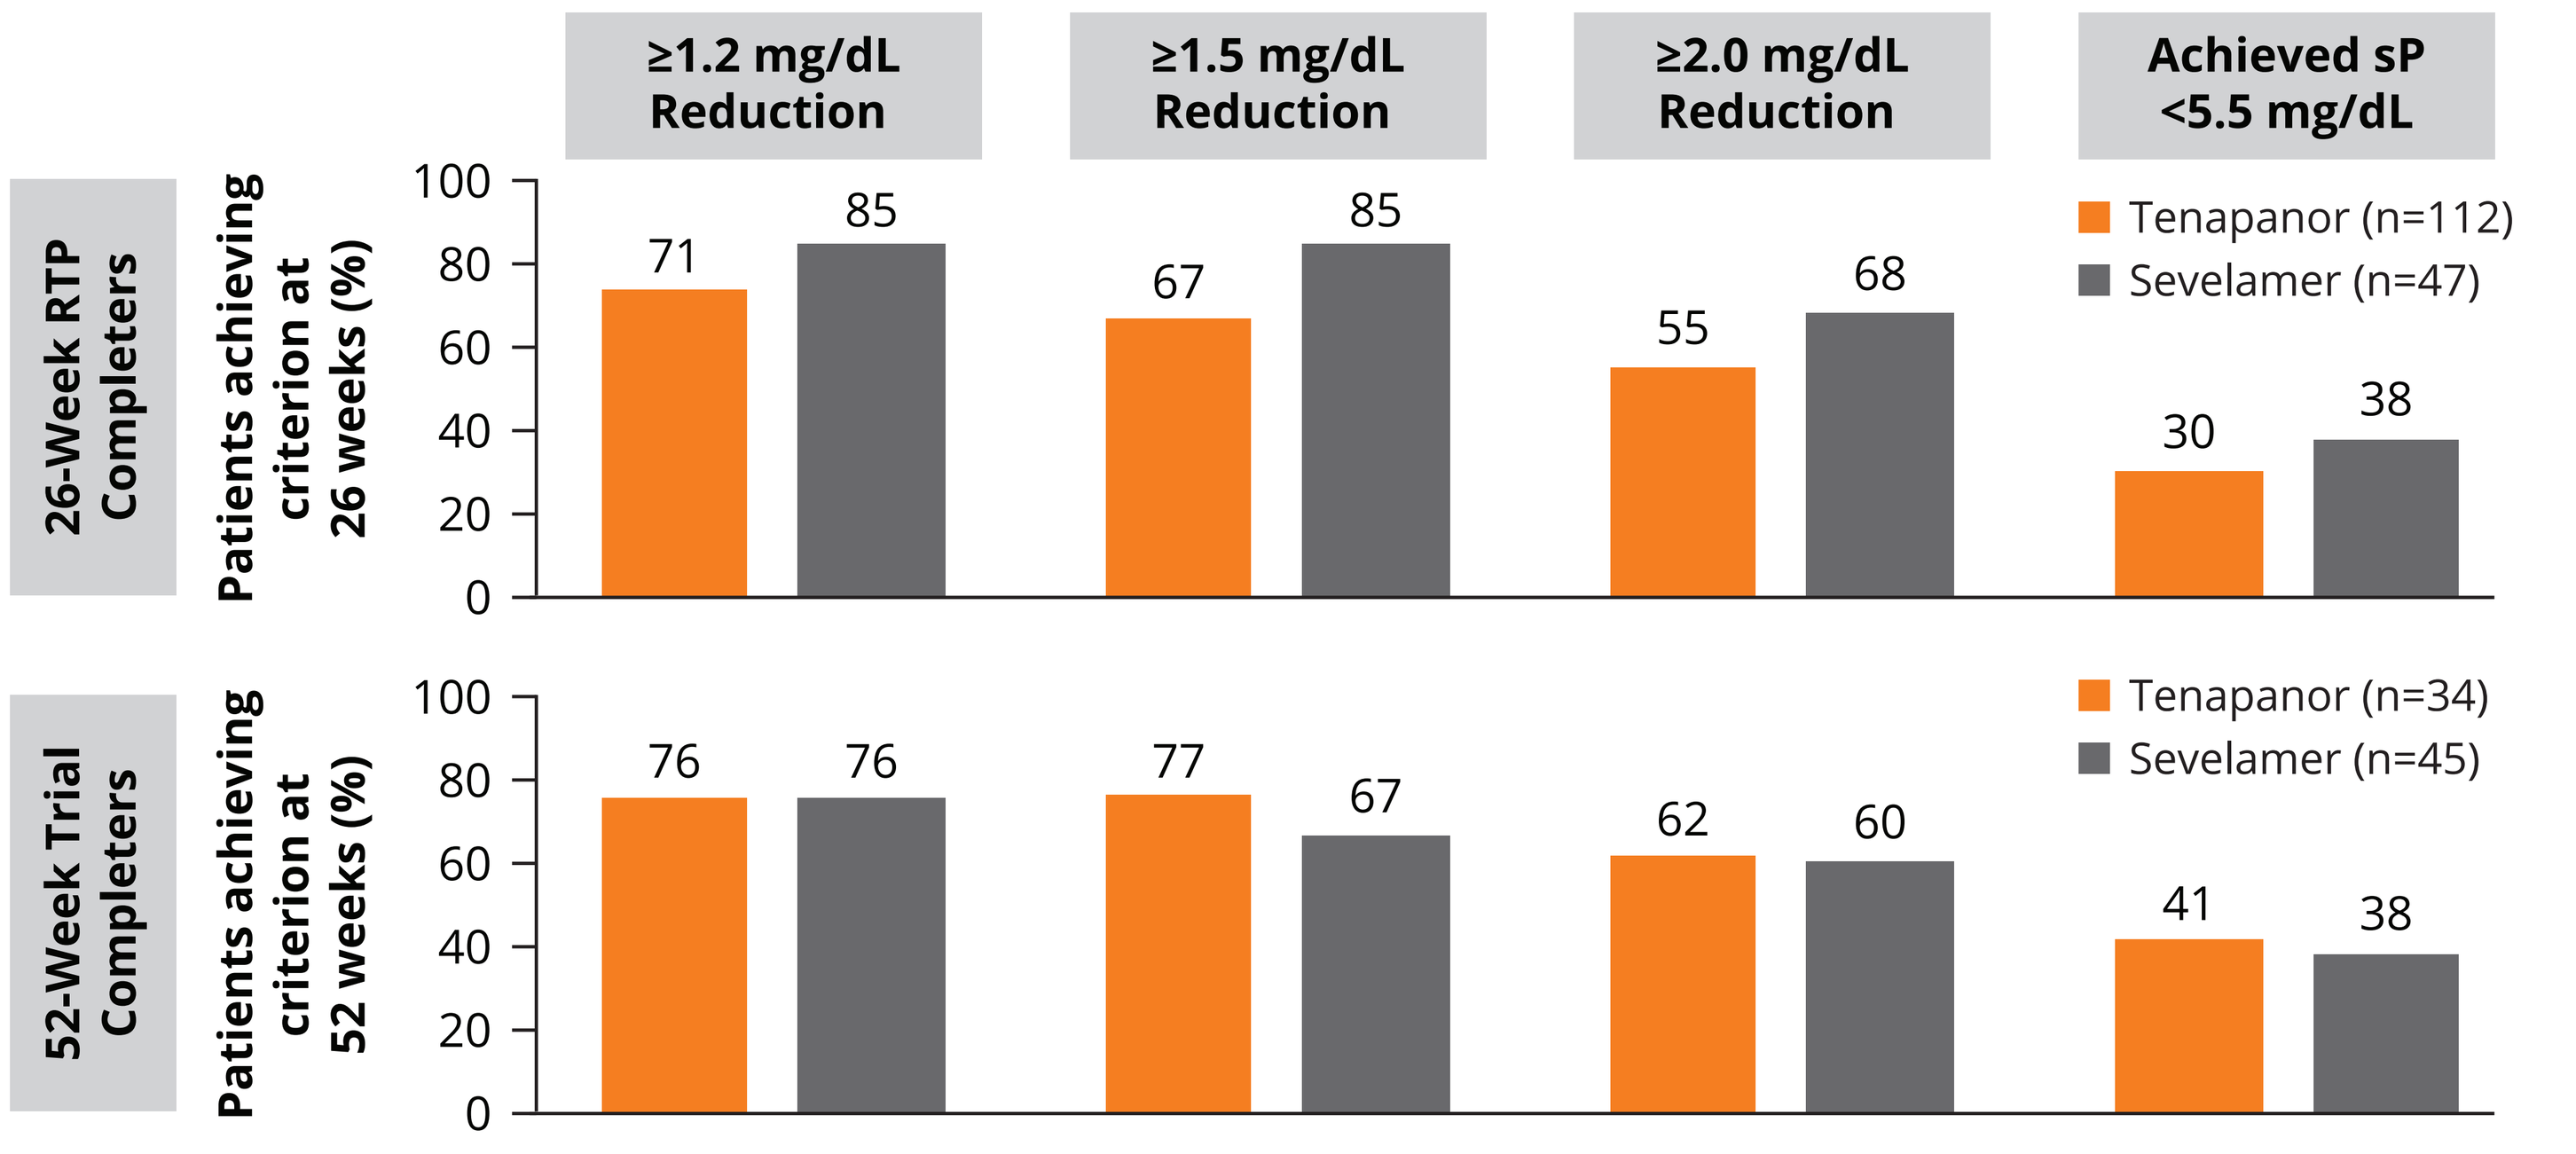
≥1.2 mg/dL Reduction
≥1.5 mg/dL Reduction
≥2.0 mg/dL Reduction
Achieved sP <5.5 mg/dL
26-Week RTP Completers
Patients achieving criterion at 26 weeks (%)
71 85
67 85
55 68
30 38
Tenapanor (n=112)
Sevelamer (n=47)
52-Week Trial Completers
Patients achieving criterion at 52 weeks (%)
76 76
77 67
62 60
41 38
Tenapanor (n=34)
Sevelamer (n=45)
100
80
60
40
20
0

Data are shown for patients with severe hyperphosphatemia who completed 26 weeks of the PHREEDOM RTP or 52 weeks of the PHREEDOM trial.
RTP, randomized treatment period.

Safety

- Treatment-emergent adverse events and serious adverse events during the RTPs of both studies for patients with severe hyperphosphatemia in the safety analysis sets are shown in **Table 2**.
- Diarrhea was the most commonly reported adverse event in patients with severe hyperphosphatemia (NCT02675998 trial, 43.1%; PHREEDOM tenapanor, 51.2%; **Table 2**), and diarrhea led to study drug discontinuation in 6.4% and 17.7% of patients, respectively.
- In PHREEDOM, serious adverse events occurred more frequently in the sevelamer group.

Table 2. Summary of Adverse Events in Patients With Severe Hyperphosphatemia (sP ≥7.5 mg/dL)

	NCT02675998 trial		PHREEDOM	
	All doses[a] (n=109)	30 mg dose (n=38)	Tenapanor (n=209)[b]	Sevelamer (n=52)
Any TEAE, n (%)	75 (68.8)	28 (73.7)	166 (79.4)	36 (69.2)
TEAEs in >5% of patients, n (%)[c]				
Diarrhea	47 (43.1)	20 (52.6)	107 (51.2)	6 (11.5)
Hyperphosphatemia	10 (9.2)	3 (7.9)	20 (9.6)	1 (1.9)
Hyperkalemia	2 (1.8)	1 (2.6)	11 (5.3)	4 (7.7)
Hypocalcemia	3 (2.8)	2 (5.3)	3 (1.4)	1 (1.9)
Upper respiratory tract infection	2 (1.8)	2 (5.3)	1 (0.5)	2 (3.8)
AV fistula site complication	3 (2.8)	2 (5.3)	4 (1.9)	3 (5.8)
Any serious TEAEs, n (%)	8 (7.3)	1 (2.6)	40 (19.1)	16 (30.8)
Serious TEAEs in >1% of patients, n (%)[c]				
Pneumonia	2 (1.8)	0	2 (1.0)	2 (3.8)
Chronic kidney disease	2 (1.8)	0	0	0
Pulmonary embolism	1 (0.9)	1 (2.6)	0	0
Fluid overload	0	0	4 (1.9)	2 (3.8)
Hyperkalemia	1 (0.9)	0	4 (1.9)	2 (3.8)
Acute respiratory failure	0	0	4 (1.9)	0
Hypertensive crisis	0	0	3 (1.4)	0
Acute myocardial infarction	0	0	3 (1.4)	1 (1.9)

Data are reported for patients with severe hyperphosphatemia from the safety analysis set.
[a]Patients in the NCT02675998 trial received tenapanor at a dose of 3 mg (fixed), 10 mg (fixed), or 30 mg (with dose titration) bid. [b]Tenapanor dose in PHREEDOM started at 30 mg bid and could be down-titrated. [c]TEAEs or serious AEs listed occurred in >5% or >1%, respectively, of tenapanor-treated patients with severe hyperphosphatemia from NCT02675998 trial (all doses or 30 mg dose) or the PHREEDOM tenapanor group.
AE, adverse event; AV, arteriovenous; bid, twice a day; TEAE, treatment emergent adverse event.

Conclusions

- During the 8-week RTP of NCT02675998 and the 26-week RTP of PHREEDOM, the mean sP decreased over time in patients with severe hyperphosphatemia (baseline sP ≥7.5 mg/dL) who were treated with tenapanor.
- In PHREEDOM, the mean sP reduction in patients with severe hyperphosphatemia was similar between patients treated with tenapanor and sevelamer; however, this treatment effect seen with tenapanor was achieved with a lower pill burden, 1 pill twice a day.
- Overall, the safety of tenapanor in the subgroup of patients with severe hyperphosphatemia was similar to that observed in the overall study populations during the RTP of the NCT02675998 and PHREEDOM trials.[9,10]
 - Diarrhea was the most commonly reported adverse event in patients with severe hyperphosphatemia who received tenapanor treatment, and rates of diarrhea were similar to those seen in the overall study populations during the RTP of the NCT02675998 and PHREEDOM trials.[9,10]
- These results suggest that tenapanor, a novel investigational PAI dosed as a single twice-daily pill, may provide a clinically meaningful reduction in sP with an acceptable safety profile for patients with severe hyperphosphatemia on maintenance dialysis who are at high risk of cardiovascular morbidity and mortality.

References

1. King AJ et al. *Sci Transl Med*. 2018;10(456):eaam6474.
2. Spencer AG et al. *Sci Transl Med*. 2014;6(227):227ra36.
3. Block GA et al. *J Am Soc Nephrol*. 2004;15(8):2208-18.
4. Kestenbaum B et al. *J Am Soc Nephrol*. 2005;16(2):520-8.
5. Palmer SC et al. *JAMA*. 2011;305(11):1119-27.
6. Block GA et al. *Clin J Am Soc Nephrol*. 2013;8(12):2132-40.
7. Shang D et al. *BMC Nephrol*. 2015;16:107.
8. Major RW et al. *PLoS One*. 2018;13(3):e0192895.
9. Block GA et al. *J Am Soc Nephrol*. 2019;30(4):641-52.
10. Block GA et al. *Kidney360*. 2021;2(10):1600-10.
11. Pergola PE et al. *J Am Soc Nephrol*. 2021;32(6):1465-73.

Disclosures

Stuart M. Sprague receives research grants from and is a consultant for Ardelyx, Inc.
Yang Yang, David M. Spiegel, and David P. Rosenbaum are employees of Ardelyx, Inc.

Acknowledgements

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Contact Information

The presenting author, Dr. Stuart Sprague, can be reached at ssprague@northshore.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

ASN Kidney Week, November 3-6, 2022; Orlando, FL, USA and Virtual Meeting