Poster: 157 ePoster: 193

Evaluation of Changes in Serum FGF23 With Tenapanor Treatment in Patients With Chronic Kidney Disease on Dialysis

Pablo E. Pergola,[1] Steven Fishbane,[2] Suling Zhao,[3] Susan Edelstein,[3] David M. Spiegel,[3] David Rosenbaum[3]

[1]Renal Associates PA, San Antonio, TX, USA; [2]Zucker School of Medicine at Hofstra and Northwell Health, Great Neck, NY, USA; [3]Ardelyx, Inc., Waltham, MA, USA

Background

- Fibroblast growth factor 23 (FGF23) is a bone-derived hormone that regulates renal phosphate reabsorption and affects both parathyroid hormone (PTH) and calcitriol (active vitamin D) synthesis.[1,2]
- Elevated intact FGF23 (iFGF23) and hyperphosphatemia are independently associated with increased risk of cardiovascular disease and mortality in patients with chronic kidney disease (CKD).[3-6]
- Tenapanor is a novel, investigational phosphate absorption inhibitor (PAI) that targets the primary, paracellular pathway of phosphate absorption in the gastrointestinal tract through inhibition of sodium/hydrogen exchanger isoform 3 (NHE3).[7-9]
- FGF23 is upregulated by dietary phosphate intake or in response to decreasing kidney function as a physiological adaptation to maintain normal serum phosphate (sP) levels.[1,4]
- In the PHREEDOM (NCT03427125), NORMALIZE (NCT03988920; 18-month extension of PHREEDOM), and OPTIMIZE (NCT04549597) studies, tenapanor treatment significantly reduced sP levels in patients with CKD on dialysis.[10,12]
- In the PHREEDOM study, tenapanor treatment led to a median relative reduction of 23% for iFGF23 from the baseline to the end of the 26-week randomized treatment period.[10]
- Here we further investigate the effect of tenapanor on iFGF23 in patients with CKD from the NORMALIZE and OPTIMIZE studies.

Methods

- In NORMALIZE (**Figure 1A**), patients who completed the tenapanor arm in PHREEDOM continued tenapanor or added sevelamer to tenapanor if sP was >4.5 mg/dL (designated as group 1). Patients who completed the sevelamer arm in PHREEDOM added tenapanor (30 mg once a day if sP was >2.5 and ≤4.5 mg/dL, or 30 mg twice a day [bid] if sP was >4.5 mg/dL) at the initial visit and reduced sevelamer dosage per protocol-specified dose-titration schedule once sP was controlled at ≤4.5 mg/dL (designated as group 2). The tenapanor dose was reduced to a minimum of 10 mg once daily if sevelamer was stopped and sP was below 2.5 mg/dL.
- OPTIMIZE (**Figure 1B**) randomized patients with sP >5.5 but ≤10 mg/dL on phosphate binder therapy at screening to cohort 1 (discontinue phosphate binders and start tenapanor 30 mg bid) or cohort 2 (decrease phosphate binders by ≥50% and start tenapanor 30 mg bid). Phosphate binder–naïve patients with sP >4.5 but ≤10 mg/dL entered cohort 3 (start tenapanor 30 mg bid).
- For this analysis, changes in iFGF23 from baseline to the end of NORMALIZE (up to 18 months) and baseline to the end of OPTIMIZE part A (initial 10 weeks of treatment) were evaluated for the full analysis set (FAS) of each study.
 - In NORMALIZE, the FAS included all patients who met the study entry eligibility criteria, received at least 1 dose of study drug (tenapanor or sevelamer), and had at least 1 post-treatment sP measurement during the study.
 - In OPTIMIZE, the FAS included all randomized patients who met the study entry eligibility criteria, received at least 1 dose of study drug (tenapanor or phosphate binder), and had at least 1 postbaseline sP measurement during the study.

Figure 1. Study Diagram of NORMALIZE (A) and OPTIMIZE (B) Studies

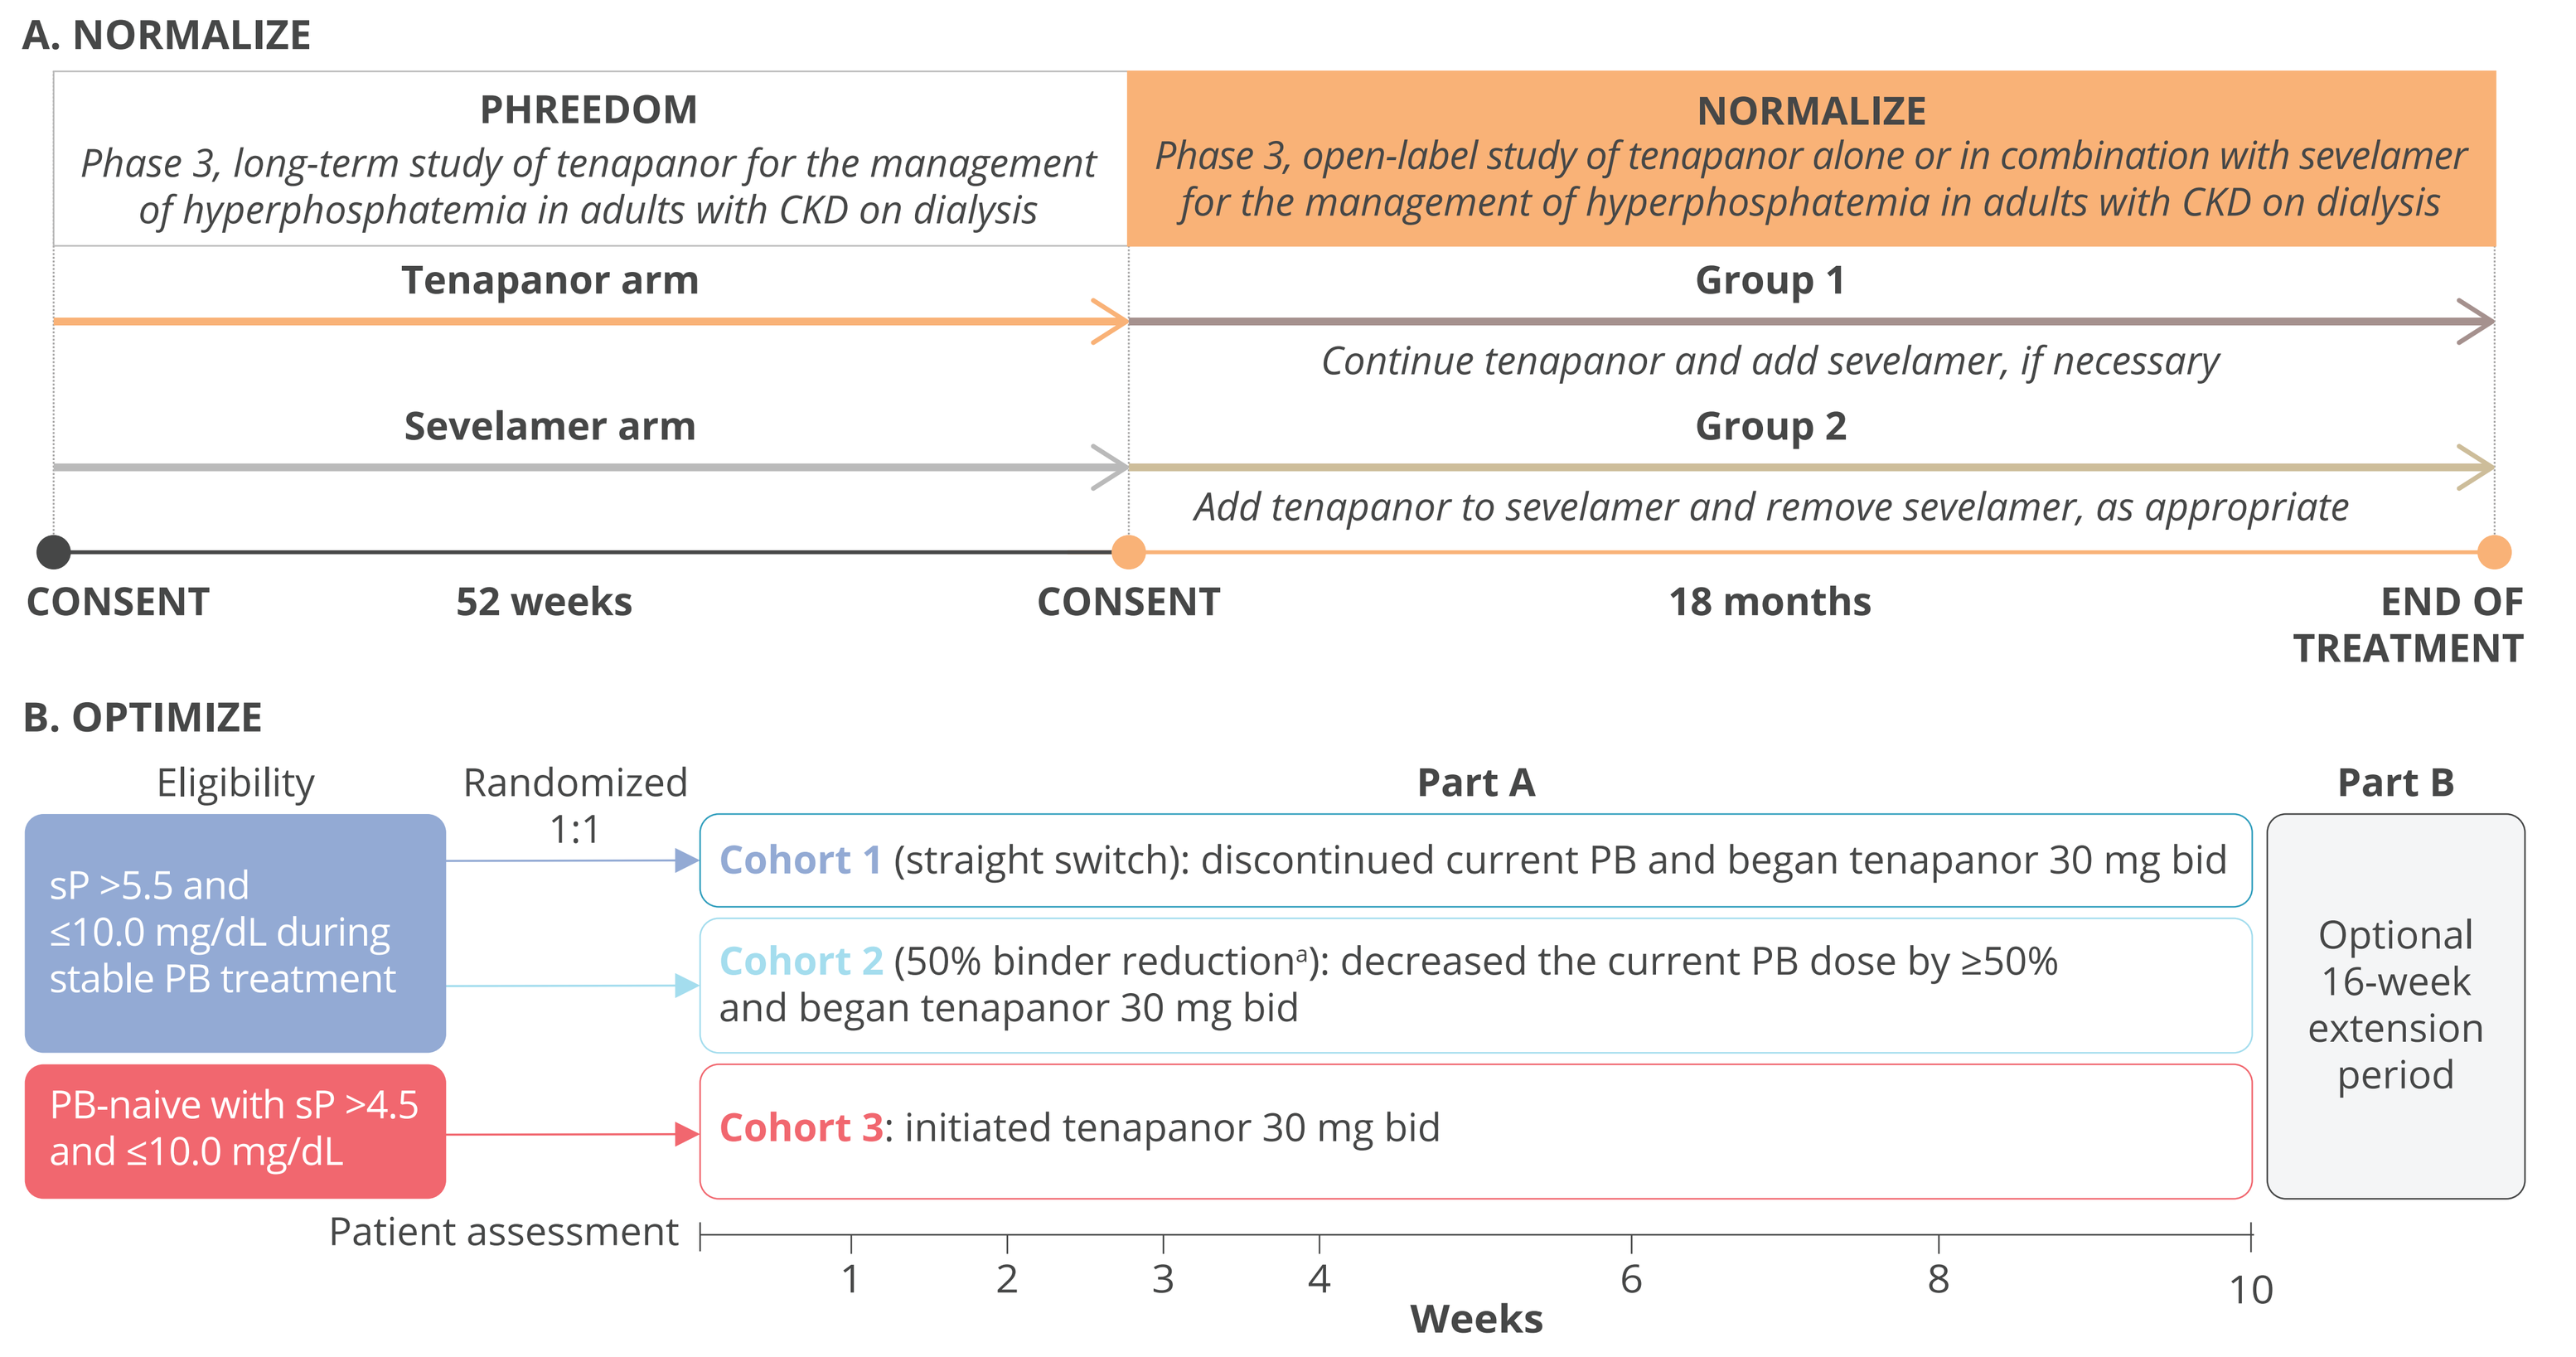
A. NORMALIZE
PHREEDOM
Phase 3, long-term study of tenapanor for the management of hyperphosphatemia in adults with CKD on dialysis
NORMALIZE
Phase 3, open-label study of tenapanor alone or in combination with sevelamer for the management of hyperphosphatemia in adults with CKD on dialysis
Tenapanor arm
Group 1
Continue tenapanor and add sevelamer, if necessary
Sevelamer arm
Group 2
Add tenapanor to sevelamer and remove sevelamer, as appropriate
CONSENT
52 weeks
CONSENT
18 months
END OF TREATMENT
B. OPTIMIZE
Eligibility
Randomized 1:1
Part A
Part B
sP >5.5 and ≤10.0 mg/dL during stable PB treatment
PB-naive with sP >4.5 and ≤10.0 mg/dL
Cohort 1 (straight switch): discontinued current PB and began tenapanor 30 mg bid
Cohort 2 (50% binder reduction[a]): decreased the current PB dose by ≥50% and began tenapanor 30 mg bid
Cohort 3: initiated tenapanor 30 mg bid
Optional 16-week extension period
Patient assessment
1
2
3
4
6
8
10
Weeks

[a]PB reduction was >50% if patient was taking an odd number of pills; eg, a patient taking 3 PB pills/day would reduce their dose to 1 PB pill/day, a patient taking 5 PB pills/day would reduce their dose to 2 PB pills/day. bid, twice a day; CKD, chronic kidney disease; PB, phosphate binder; sP, serum phosphate.

Results

Patients

- Patient demographics and baseline characteristics are presented for the NORMALIZE and OPTIMIZE studies in **Table 1**.
 - Baseline sP and iFGF23 levels were overall lower among patients in NORMALIZE compared with those of patients in cohort 1 and cohort 2 of OPTIMIZE due to patients in NORMALIZE having received prior treatment for hyperphosphatemia in the PHREEDOM study.

Table 1. Demographics and Baseline Characteristics (Full Analysis Set)

	NORMALIZE		OPTIMIZE		
Characteristic category/statistic	**Group 1 (N=111)**	**Group 2 (N=60)**	**Cohort 1 (N=151)**	**Cohort 2 (N=152)**	**Cohort 3 (N=30)**
Age at screening, years	56.0 (12.1)	58.3 (14.1)	52.4 (11.1)	53.2 (12.1)	55.4 (16.5)
Male, n (%)	65 (58.6)	43 (71.7)	107 (70.9)	102 (67.1)	18 (60.0)
Race, n (%)					
Black or African American	59 (53.2)	26 (43.3)	66 (43.7)	71 (46.7)	9 (30.0)
White	44 (39.6)	31 (51.7)	64 (42.4)	62 (40.8)	20 (66.7)
Asian	1 (0.9)	3 (5.0)	13 (8.6)	3 (2.0)	0
American Indian or Alaska Native	7 (6.3)	0	3 (2.0)	6 (3.9)	1 (3.3)
Native Hawaiian or Other Pacific Islander	0	0	2 (1.3)	4 (2.6)	0
Other/Unknown	0	0	3 (2.0)	6 (3.9)	0
Ethnicity, n (%)					
Not Hispanic or Latino	80 (72.1)	40 (66.7)	109 (72.2)	114 (75.0)	17 (56.7)
Hispanic or Latino	31 (27.9)	20 (33.3)	42 (27.8)	38 (25.0)	13 (43.3)
Duration since first dialysis at baseline, months[a]	59.2 (40.0)	74.3 (56.3)	57.4 (52.4)	57.7 (49.5)	19.3 (20.7)
Baseline sP, mg/dL[b]	5.8 (1.5)	5.8 (1.6)	7.1 (1.0)	7.2 (1.1)	6.1 (0.8)
Baseline iFGF23, pg/mL[b]	8800.6 (10133.2)	10830.5 (15198.4)	12391.8 (14832.1)	17631.2 (17706.4)	4340.5 (6146.4)
Baseline iPTH, pg/mL[b]	424.2 (321.7)	347.1 (211.9)	428.3 (242.9)	391.6 (243.3)	320.8 (132.1)
Most recent Kt/V at screening	1.7 (0.59)	1.9 (1.11)	1.7 (0.32)	1.6 (0.34)	1.7 (0.55)

Unless otherwise indicated, data are mean (SD).
[a]Baseline was defined as the date of informed consent collected in NORMALIZE and randomization/enrollment date in OPTIMIZE.
[b]Baseline was defined as the last observed measurement collected prior to the first treatment dose in the NORMALIZE and OPTIMIZE studies, respectively.
iFGF23, intact fibroblast growth factor 23; iPTH, intact parathyroid hormone; Kt/V, a measure of dialysis dose; sP, serum phosphate.

iFGF23 Responders

- At the end of the NORMALIZE trial (up to 18 months), 99 of 163 patients (60.7%) in the FAS had a reduction in iFGF23 (**Figure 2A**).
- At the end of part A in the OPTIMIZE trial, 211 of 317 patients (66.6%) in the FAS had a reduction in iFGF23 (**Figure 2B**).
- In general, patients who had a reduction in sP also experienced a reduction in iFGF23; however, there was no clear trend in the degree of sP and iFGF23 reduction (**Figure 2**).

Figure 2. Change in iFGF23 and sP (Full Analysis Set)

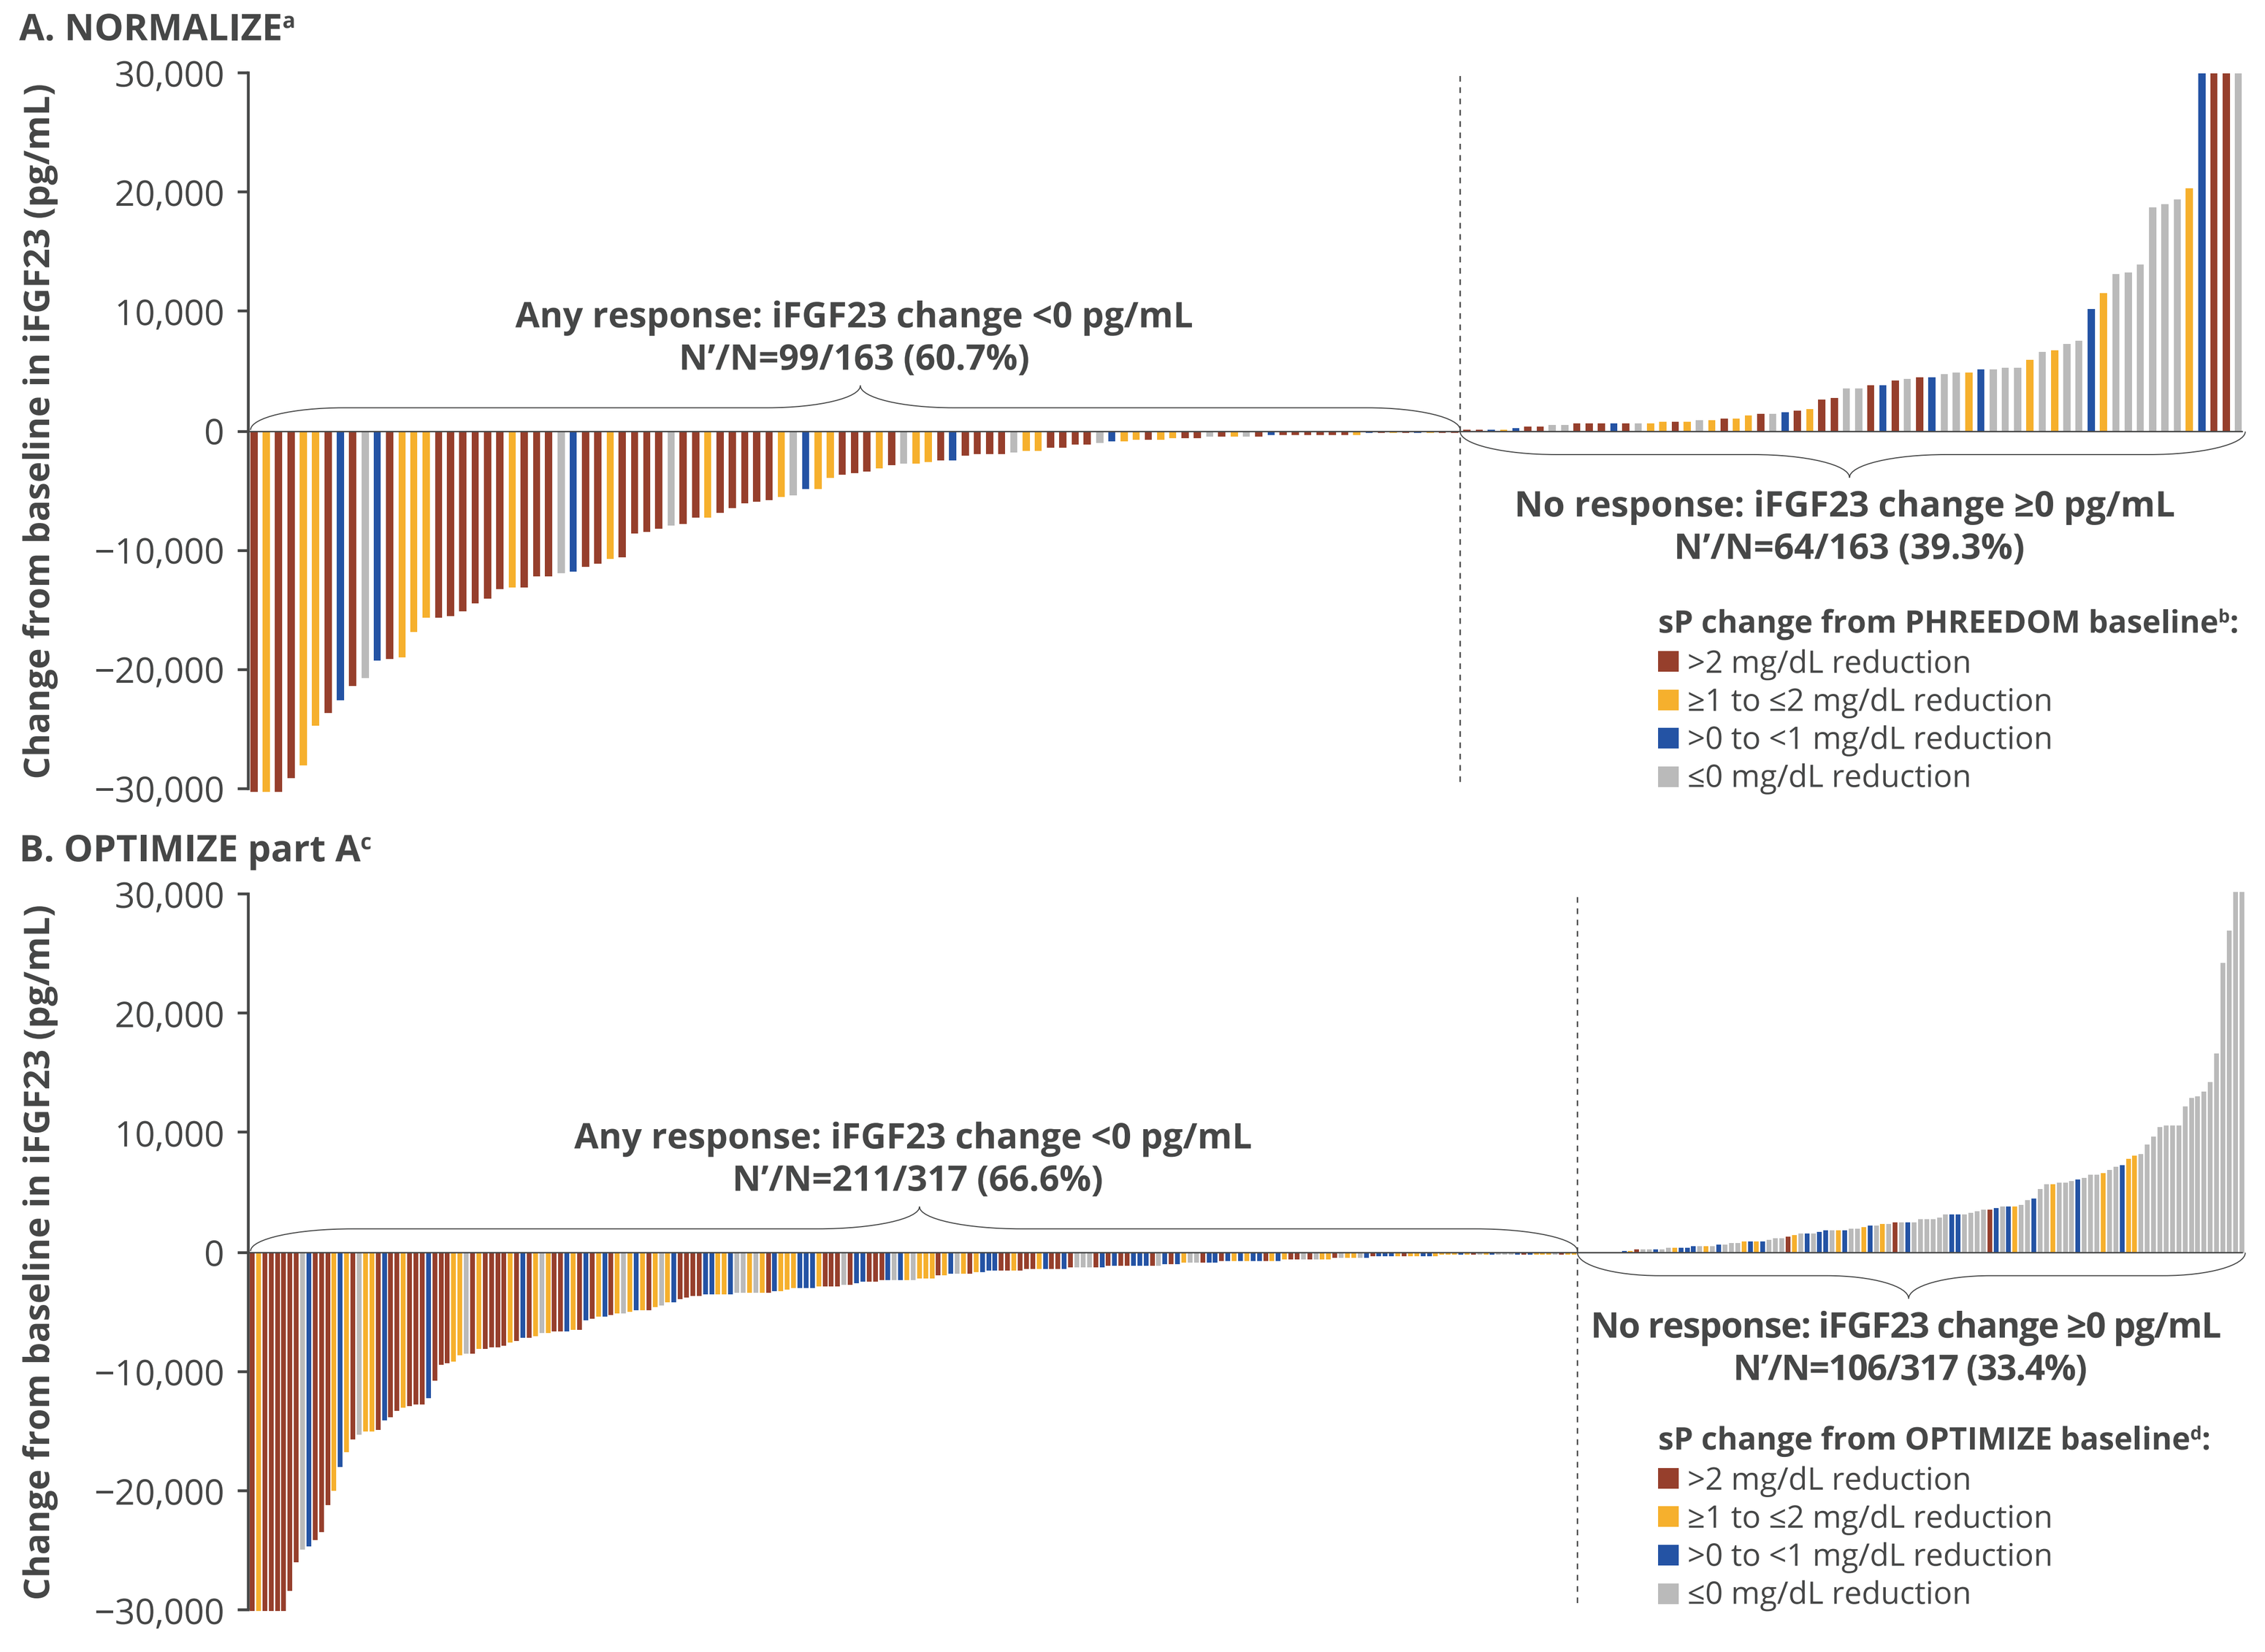
A. NORMALIZE[a]
30,000
20,000
10,000
0
−10,000
−20,000
−30,000
Change from baseline in iFGF23 (pg/mL)
Any response: iFGF23 change <0 pg/mL
N′/N=99/163 (60.7%)
No response: iFGF23 change ≥0 pg/mL
N′/N=64/163 (39.3%)
sP change from PHREEDOM baseline[b]:
>2 mg/dL reduction
≥1 to ≤2 mg/dL reduction
>0 to <1 mg/dL reduction
≤0 mg/dL reduction
B. OPTIMIZE part A[c]
30,000
20,000
10,000
0
−10,000
−20,000
−30,000
Change from baseline in iFGF23 (pg/mL)
Any response: iFGF23 change <0 pg/mL
N′/N=211/317 (66.6%)
No response: iFGF23 change ≥0 pg/mL
N′/N=106/317 (33.4%)
sP change from OPTIMIZE baseline[d]:
>2 mg/dL reduction
≥1 to ≤2 mg/dL reduction
>0 to <1 mg/dL reduction
≤0 mg/dL reduction

[a]Change in iFGF23 was calculated from last observed measurement collected prior to the first treatment dose in the NORMALIZE study; eight patients in NORMALIZE did not have iFGF23 data available at the endpoint visit (the visit with the last observed sP assessment). The graph only shows data between −30000 and 30000; the 3 leftmost columns had iFGF23 changes of −57486.4, −31791.4, and −30328.5 pg/mL and the 4 rightmost columns had changes of 46734.0, 58160.7, 61773.2, and 64022.1 pg/mL. [b]Change in sP was calculated from last measurement collected prior to the first dose of study drug in the PHREEDOM study. [c]Change in iFGF23 was calculated from last observed measurement collected collected prior to the first treatment dose in the OPTIMIZE study; sixteen patients in OPTIMIZE did not have iFGF23 data available at the part A endpoint visit (the visit with the last observed measurement during the 10-week treatment period). The graph only shows data between −30000 and 30000; the 6 leftmost columns had iFGF23 changes of −59956.9, −57272.8, −41375.0, −40071.0, −31518.0, and −31441.0 pg/mL and the 2 rightmost columns had changes of 33041.0 and 43690.1 pg/mL. [d]Change in sP was calculated from last measurement collected prior to the first dose of study drug in the OPTIMIZE study. iFGF23, intact fibroblast growth factor 23; sP, serum phosphate.

iFGF23 Reduction

- Among all patients in NORMALIZE from the FAS, median iFGF23 reduction (percent reduction) was 384.0 pg/mL (42.2%) from the NORMALIZE baseline to the end of the study (**Figure 3A**).
 - The median relative reduction from baseline to the end of the study in iFGF23 levels was 44.1% in group 1 and 35.9% in group 2.
 - The estimated least squares (LS) mean relative reduction from baseline iFGF23 was 41% in group 1 and 29% in group 2.
- In OPTIMIZE, at the end of part A, the median iFGF23 reduction (percent reduction) was 739.2 pg/mL (20%) from baseline (**Figure 3B**) in the FAS.
 - The median relative reduction from baseline to the end of part A in iFGF23 levels was 12.2% in cohort 1, 22.0% in cohort 2, and 38.7% in cohort 3.
 - The estimated LS mean relative reduction from baseline iFGF23 was 15% in cohort 1 and 30% in cohort 2.
 - The LS mean relative reduction was not estimated for phosphate binder–naïve patients in cohort 3 as they were from a different population than cohort 1 and cohort 2.

Figure 3. Reduction in iFGF23 From Baseline in the NORMALIZE and OPTIMIZE Trials

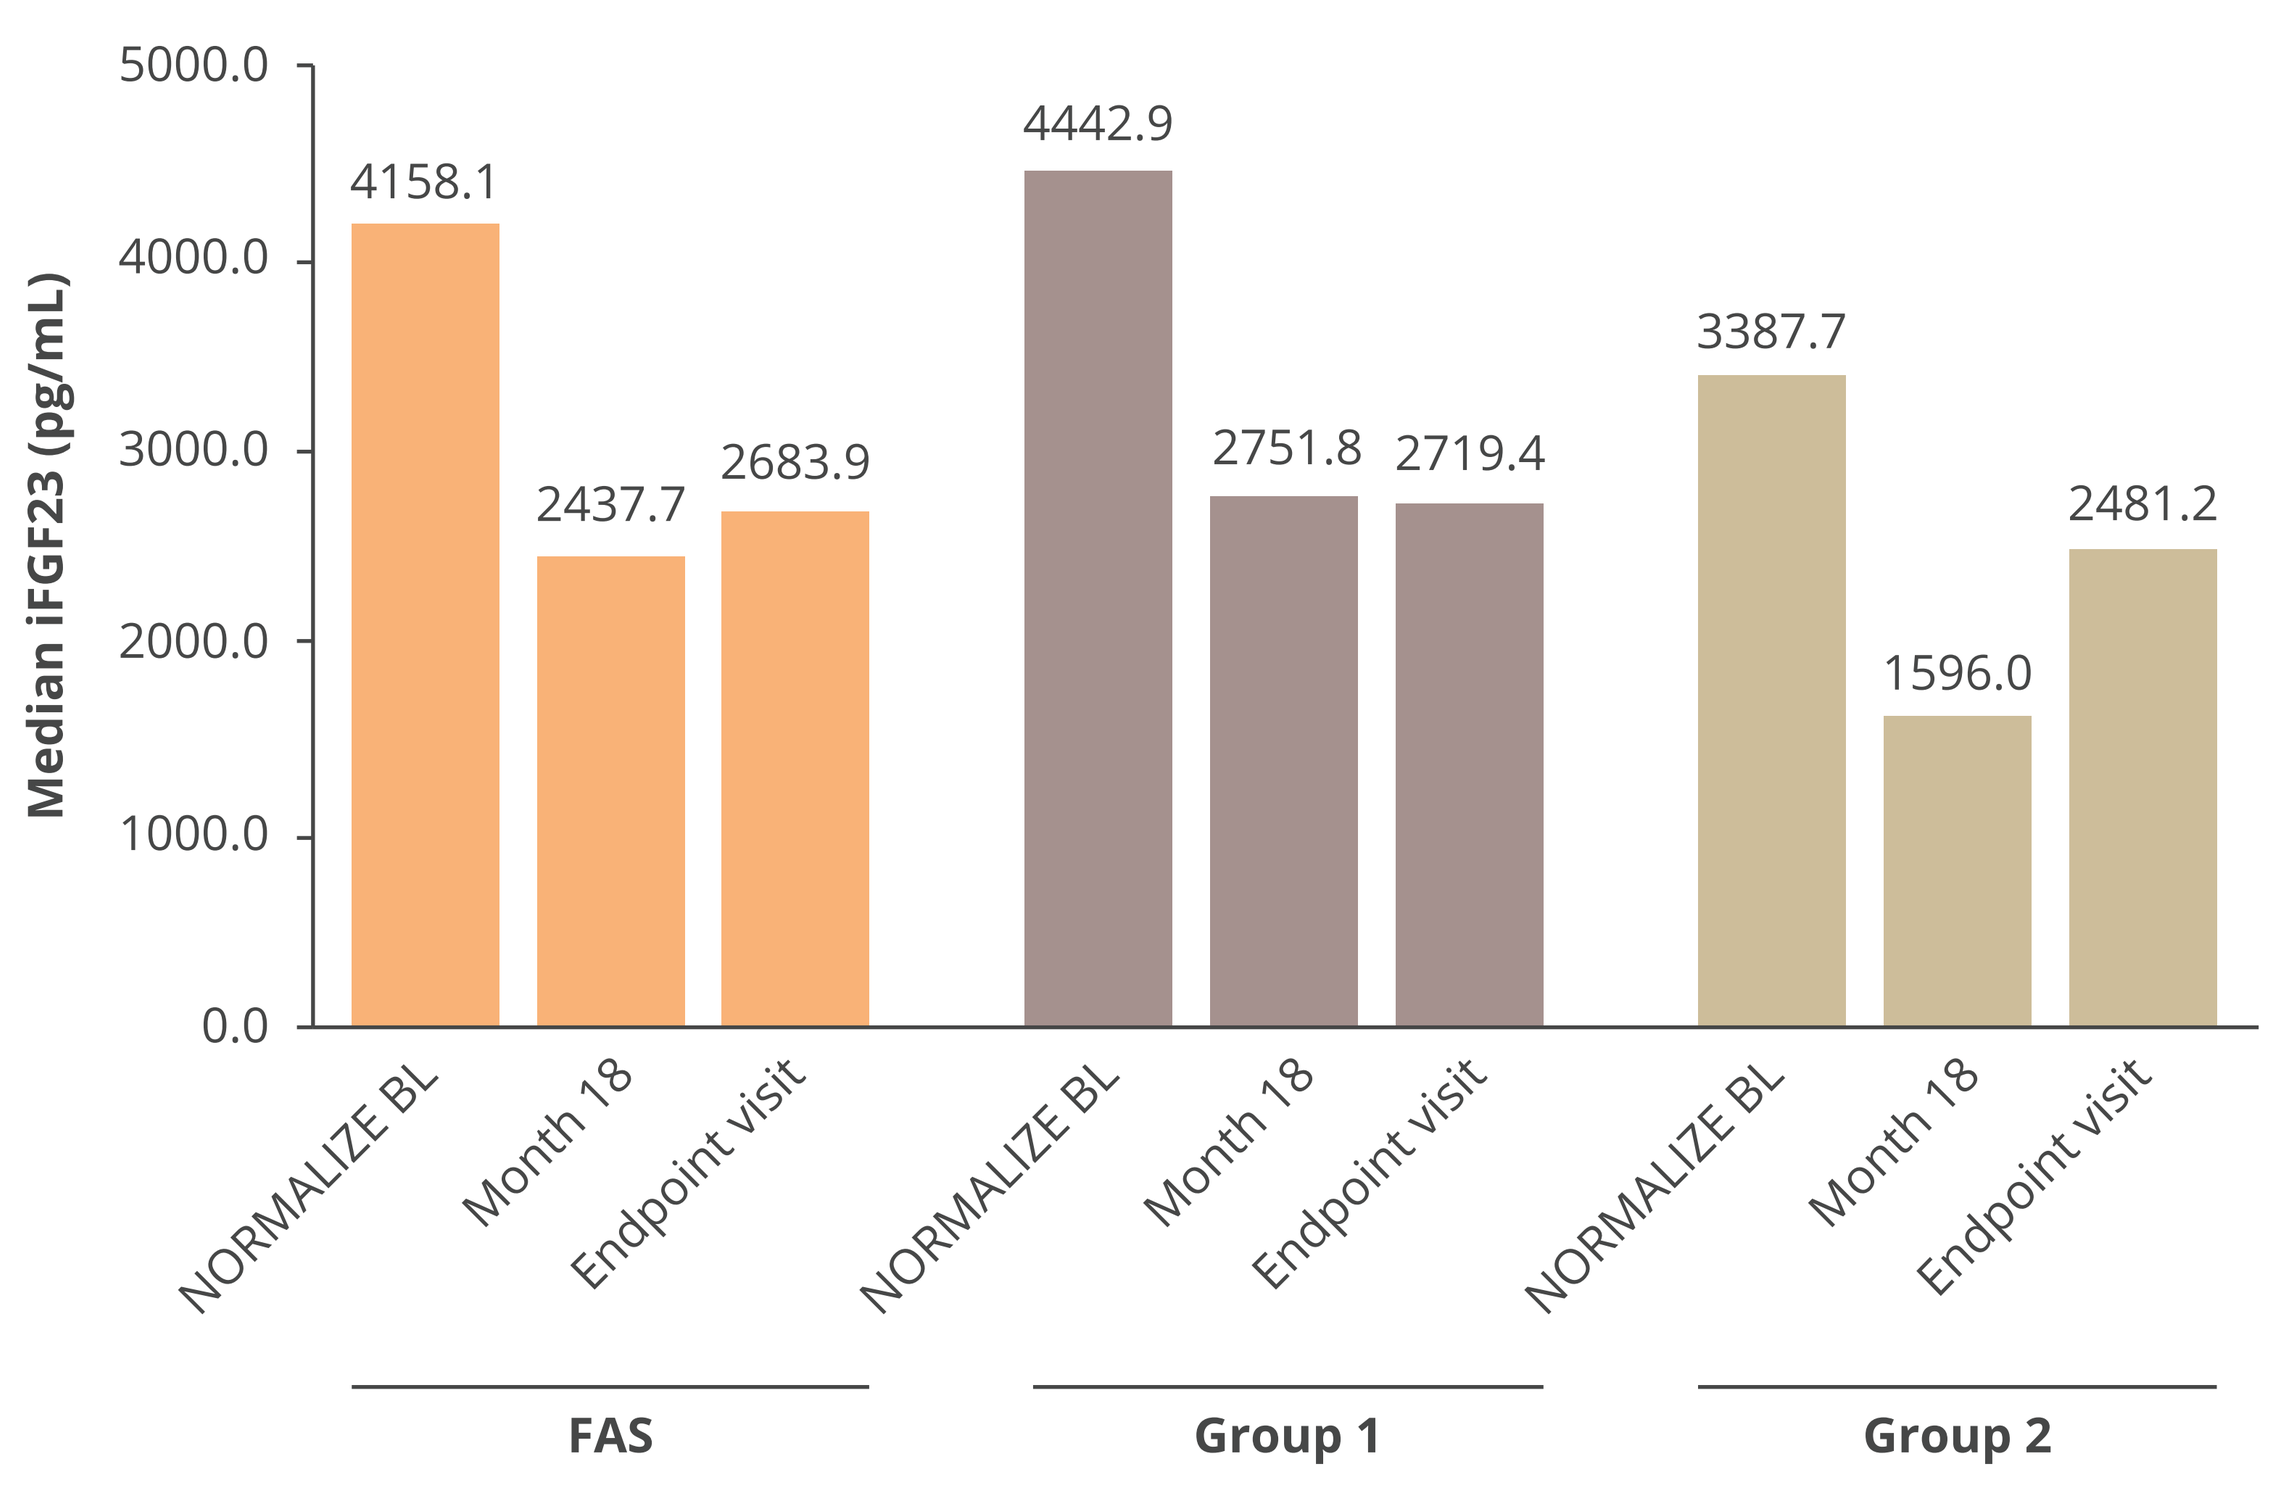
A. NORMALIZE
5000.0
4000.0
3000.0
2000.0
1000.0
0.0
Median iFGF23 (pg/mL)
4158.1
2437.7
2683.9
4442.9
2751.8
2719.4
3387.7
1596.0
2481.2
NORMALIZE BL
Month 18
Endpoint visit
NORMALIZE BL
Month 18
Endpoint visit
NORMALIZE BL
Month 18
Endpoint visit
FAS
Group 1
Group 2

Median iFGF23 reduction from BL to endpoint visit[a]

	Reduction, pg/mL	Relative reduction, %
FAS	384.0	42.2
Group 1	534.3	44.1
Group 2	215.7	35.9

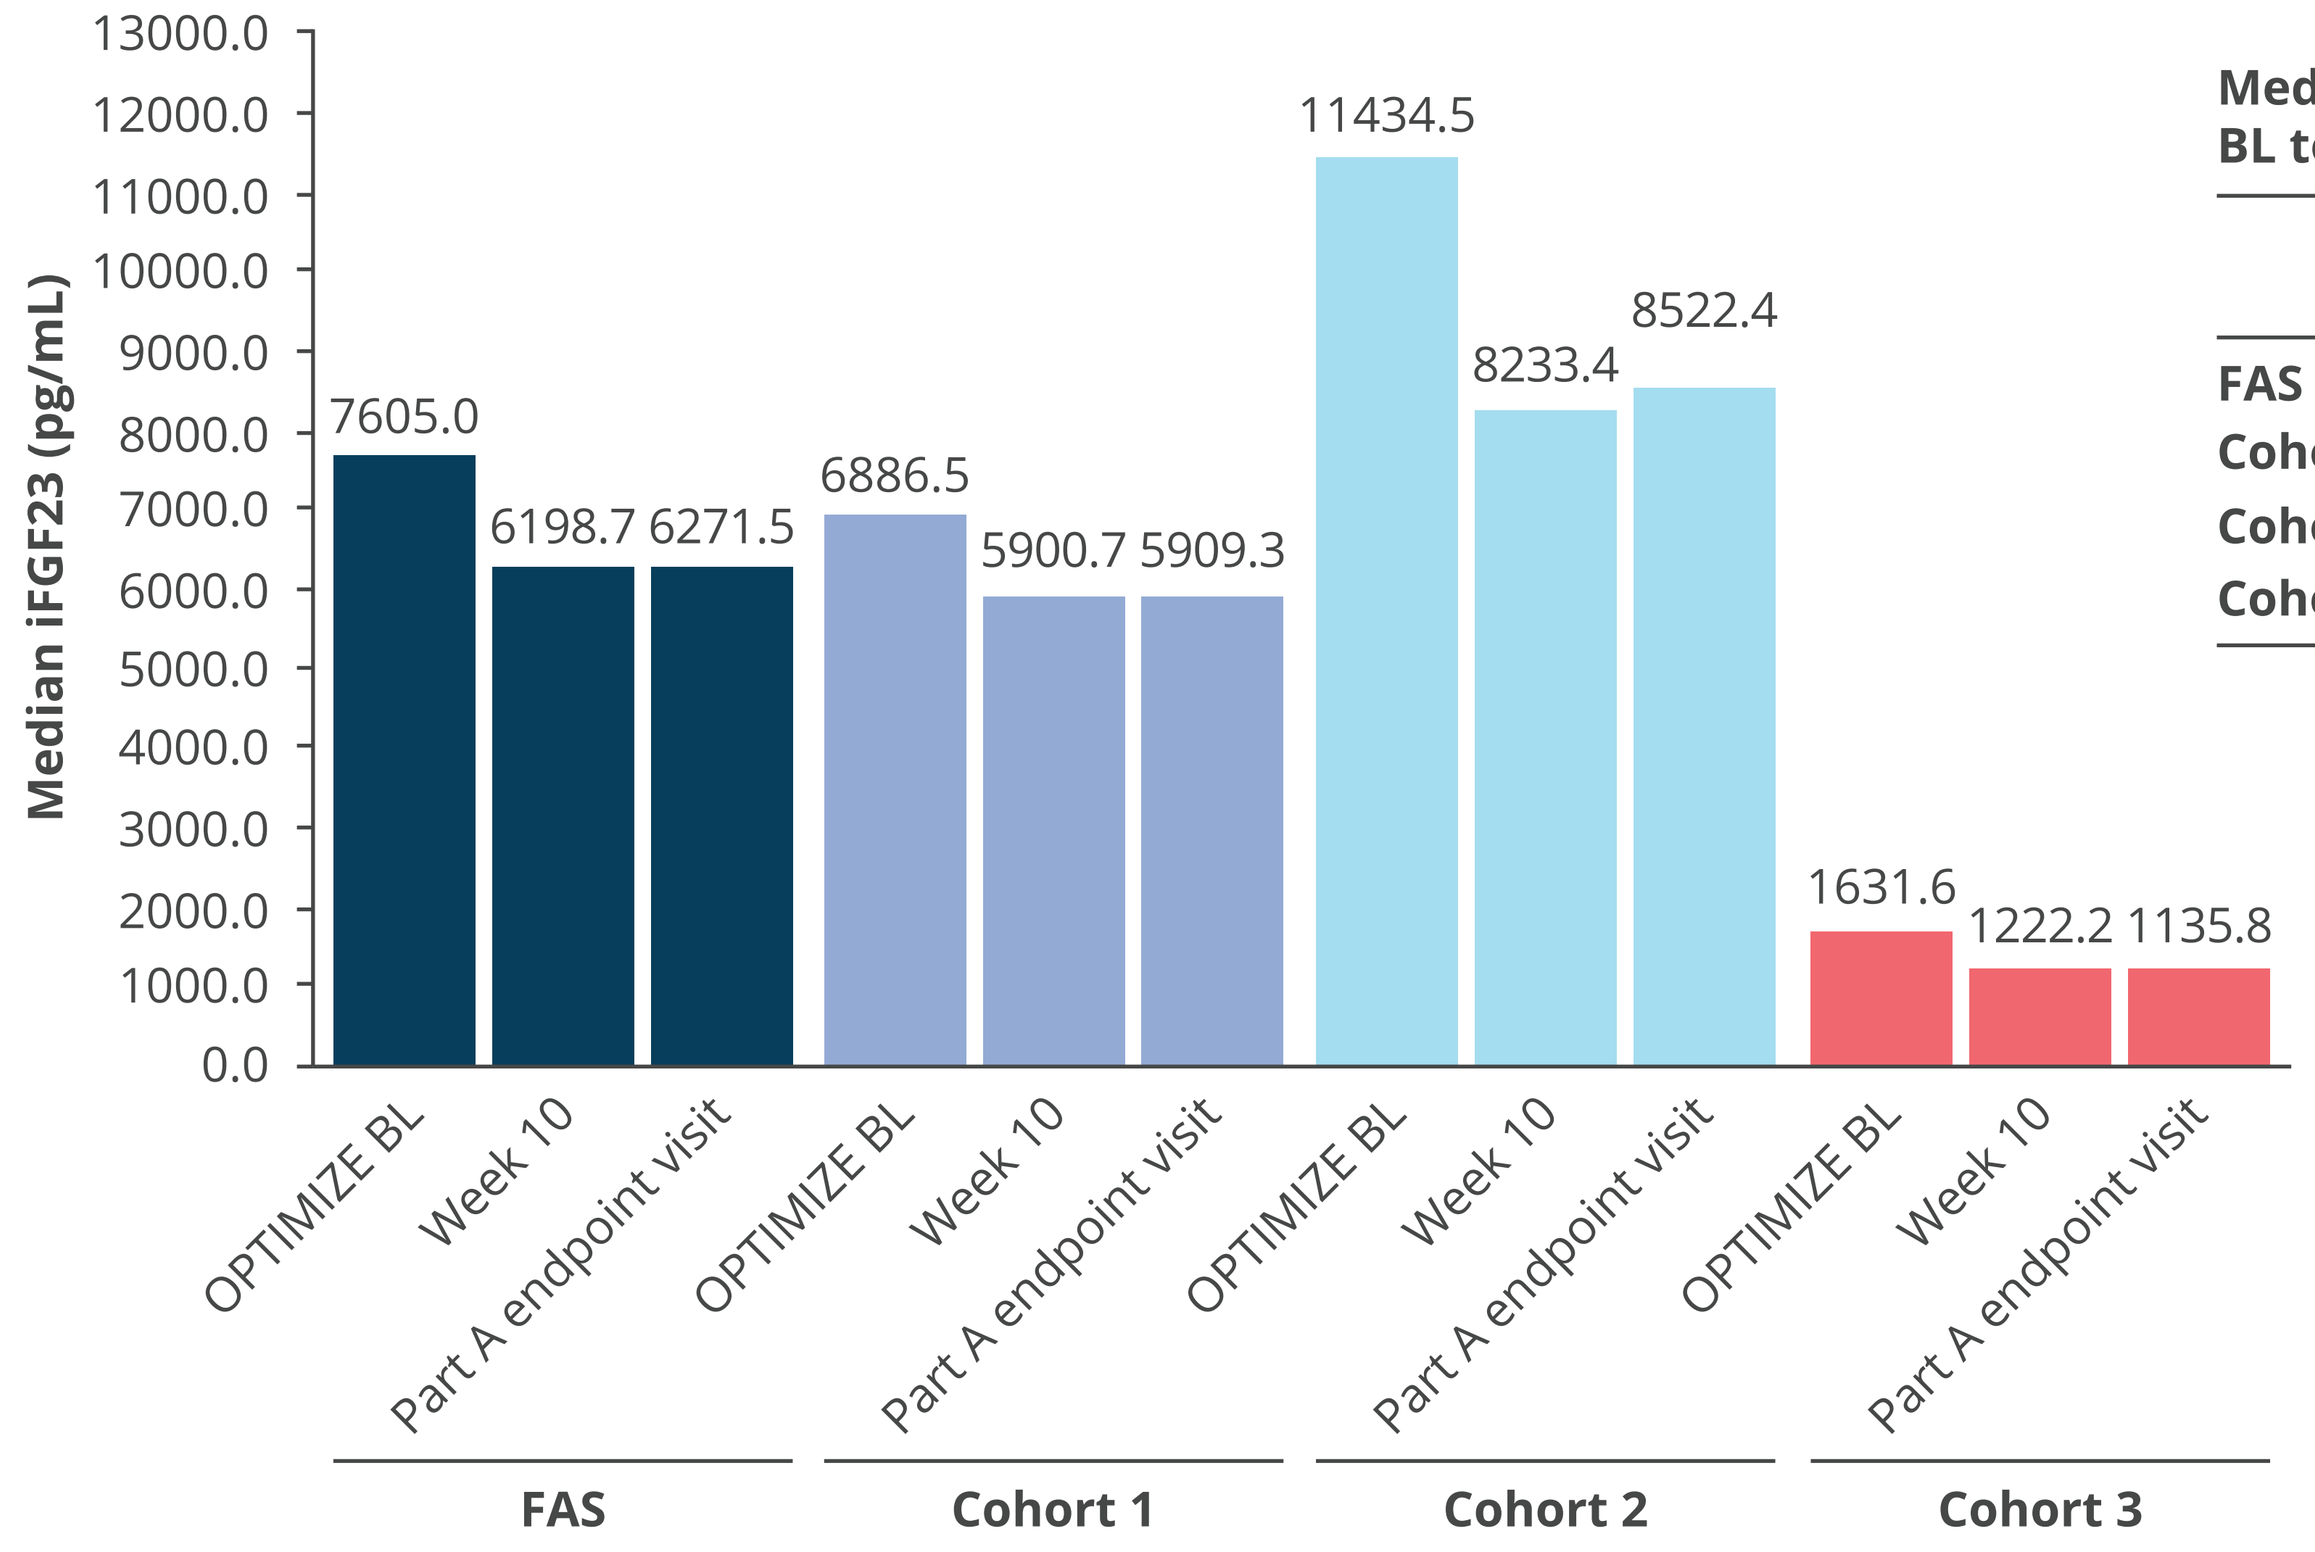
B. OPTIMIZE part A
13000.0
12000.0
11000.0
10000.0
9000.0
8000.0
7000.0
6000.0
5000.0
4000.0
3000.0
2000.0
1000.0
0.0
Median iFGF23 (pg/mL)
7605.0
6198.7
6271.5
6886.5
5900.7
5909.3
11434.5
8233.4
8522.4
1631.6
1222.2
1135.8
OPTIMIZE BL
Week 10
Part A endpoint visit
OPTIMIZE BL
Week 10
Part A endpoint visit
OPTIMIZE BL
Week 10
Part A endpoint visit
OPTIMIZE BL
Week 10
Part A endpoint visit
FAS
Cohort 1
Cohort 2
Cohort 3

Median iFGF23 reduction from BL to part A endpoint visit[b]

	Reduction, pg/mL	Relative reduction, %
FAS	739.2	20.0
Cohort 1	573.2	12.2
Cohort 2	1180.8	22.0
Cohort 3	424.5	38.7

[a]The endpoint visit of the NORMALIZE study was the visit with the last observed sP assessment. [b]The part A endpoint visit of the OPTIMIZE study was the visit with the last observed measurement during the 10-week treatment period.
BL, baseline; FAS, full analysis set; iFGF23, intact fibroblast growth factor 23; sP, serum phosphate.

Safety

- In the safety analysis set of NORMALIZE (n=172) and OPTIMIZE (n=333), diarrhea was the most common adverse event (AE), occurring in 38 (22.1%) and 133 (39.9%) patients, respectively.
- Previous reports showed that diarrhea was mostly mild to moderate. However, it was the most common AE that led to tenapanor discontinuation in both NORMALIZE (4 of 172 patients [2.3%]) and OPTIMIZE (22 of 333 patients [6.6%]).[11,12]
- There were 14 deaths due to AEs (9 treatment-emergent) in the NORMALIZE trial[11] and 7 deaths due to AEs (2 treatment-emergent) in the OPTIMIZE trial. None were considered to be related to tenapanor treatment.

Conclusions

While there was variability in the degree of reduction, many patients who responded to tenapanor as core therapy with a reduction in sP levels also experienced reduction in iFGF23 levels.

The NORMALIZE and OPTIMIZE trials demonstrate that tenapanor is effective at reducing iFGF23 levels in patients with CKD on maintenance dialysis, which is consistent with findings from the PHREEDOM study.[10]

Results from both NORMALIZE and OPTIMIZE trials support the long-term safety of tenapanor; AEs were similar to those of previous studies, with diarrhea being the most common AE.[10,13]

Disclosures

Pablo E. Pergola serves as a scientific advisor for Ardelyx, Inc., and was an OPTIMIZE study investigator. Steven Fishbane served as a scientific advisor for Ardelyx, Inc. Suling Zhao, Susan Edelstein, David Rosenbaum, and David Spiegel are full-time employees of Ardelyx, Inc.

References

1. Rausch S et al. *Pflug Arch*. 2022;474:281-92.
2. Bergwitz C et al. *Annu Rev Med*. 2010;61:91-104.
3. Gutiérrez OM et al. *N Eng J Med*. 2008;359:584-92.
4. Isakova T et al. *JAMA*. 2011;305:2432-9.
5. Block GA et al. *J Am Soc Nephrol*. 2004;15:2208-18.
6. Zhou C et al. *Front Cell Dev Biol*. 2021;9:644363.
7. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
8. Spencer AG et al. *Sci Transl Med*. 2014;6:227ra36.
9. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
10. Block GA et al. *Kidney360*. 2021;2:1600-10.
11. Rosenbaum DP et al. Poster presented at: National Kidney Foundation Spring Clinical Meetings; April 6-10, 2022; Boston, MA, USA & virtual.
12. Edelstein S et al. Poster presented at: National Kidney Foundation Spring Clinical Meetings; April 6-10, 2022; Boston, MA, USA & virtual.
13. Pergola PE et al. *J Am Soc Nephrol*. 2021;32(6):1465-73.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr Pergola can be contacted for further information on this study at ppergola@raparesearch.com.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

National Kidney Foundation Spring Clinical Meetings (SCM23); April 11-15, 2023; Austin, TX, USA

MED-US--2400026