Poster: FR-PO318

Tenapanor in Combination With Phosphate Binders Improves Short and Long-term Control of Serum Phosphate (sP) in Patients on Dialysis With Hyperphosphatemia

Daniel Weiner,[1] Robert Lynn,[2] David Spiegel,[3] Susan Edelstein,[3] Yang Yang,[3] David Rosenbaum[3]

[1]Tufts Medical Center, Boston, MA, USA; [2]Kidney Medical Associates, Bronx, NY, USA; [3]Ardelyx, Inc., Waltham, MA, USA

Background

- Tenapanor is a first-in-class, minimally absorbed phosphate inhibitor that reduces paracellular phosphate permeability, thereby blocking paracellular phosphate absorption, by local inhibition of the intestinal sodium/hydrogen exchanger isoform 3 (NHE3).[1-3]
 - In a previous 4-week study, tenapanor in combination with phosphate binders, including sevelamer carbonate (sevelamer), allowed more patients on maintenance dialysis to achieve serum phosphorus (sP) <5.5 mg/dL compared to binder treatment alone.[4]
- The randomized, phase 3 PHREEDOM trial (NCT03427125) compared tenapanor to sevelamer monotherapy,[5] demonstrating that tenapanor safely reduced serum phosphate in maintenance dialysis patients.
- NORMALIZE (NCT03988920), a post-registrational, open-label, extension study of the PHREEDOM study,[6] evaluated tenapanor alone or in combination with sevelamer.
 - The current report focuses on the subset of PHREEDOM patients randomized to sevelamer who had sP >4.5 mg/dL on completion of PHREEDOM and the change in sP experienced by this subset in response to tenapanor treatment in the NORMALIZE study.

Methods

- At study entry into NORMALIZE, patients from the sevelamer arm of PHREEDOM had tenapanor added to their sevelamer dose on the basis of their baseline sP level.
 - For those entering NORMALIZE with an sP >4.5 mg/dL, tenapanor was initiated at the dose of 30 mg twice a day (bid).
 - After starting tenapanor, the sevelamer dose was decreased as long as sP was controlled, with the intention of decreasing/eliminating sevelamer if possible.
 - Tenapanor could be up- or down-titrated in 10-mg increments, with a maximum dose of 30 mg bid, based on sP level and/or tolerability.
- Patients were monitored for safety and efficacy with in-office and telephone visits for up to 18 months.
- The efficacy outcome of NORMALIZE was achievement of sP ≤4.5 mg/dL.[6]

Results

Patients

- Of 61 patients from the sevelamer arm of PHREEDOM entering NORMALIZE, a subset of 43 (70.5%) patients had sP >4.5 mg/dL at NORMALIZE baseline, when tenapanor was added to the sevelamer dose at the initial dose of 30 mg bid.
- The efficacy analysis included 42 patients from this subset.
 - One patient from this subset did not have a post-initiation sP measurement and was not included in the efficacy analysis.
- Most patients in this subset were male, with a mean age of 57.4 years; the majority had sP ≥5.5 mg/dL and <7.5 mg/dL at baseline (**Table 1**).

Table 1. Baseline Demographics and Disease Characteristics

	Patients (N=43)
Mean age, years (SD)	57.4 (12.9)
Male, n (%)	32 (74.4)
Race, n (%)	
White	22 (51.2)
Black or African American	19 (44.2)
Asian	2 (4.7)
Mean BMI, kg/m² (SD)	29.7 (7.4)
Type of dialysis, n (%)	
Hemodialysis	38 (88.4)
Peritoneal dialysis	5 (11.6)
Mean duration of dialysis treatment, months (SD)	74.8 (50.2)
Mean sP, mg/dL (SD)	6.5 (1.3)
sP level, n (%)	
≥4.6 mg/dL and <5.5 mg/dL	9 (20.9)
≥5.5 mg/dL and <7.5 mg/dL	26 (60.5)
≥7.5 mg/dL	8 (18.6)

BMI, body mass index; sP, serum phosphorus.

sP Response

- With the addition of tenapanor, the percentage of patients achieving sP ≤4.5 mg/dL increased to 29% at Week 1 and ranged from 35% to 54% at subsequent study visits in this subset (**Figure 1**).

Figure 1. Percentage of Patients With sP ≤4.5 mg/dL by Analysis Visit

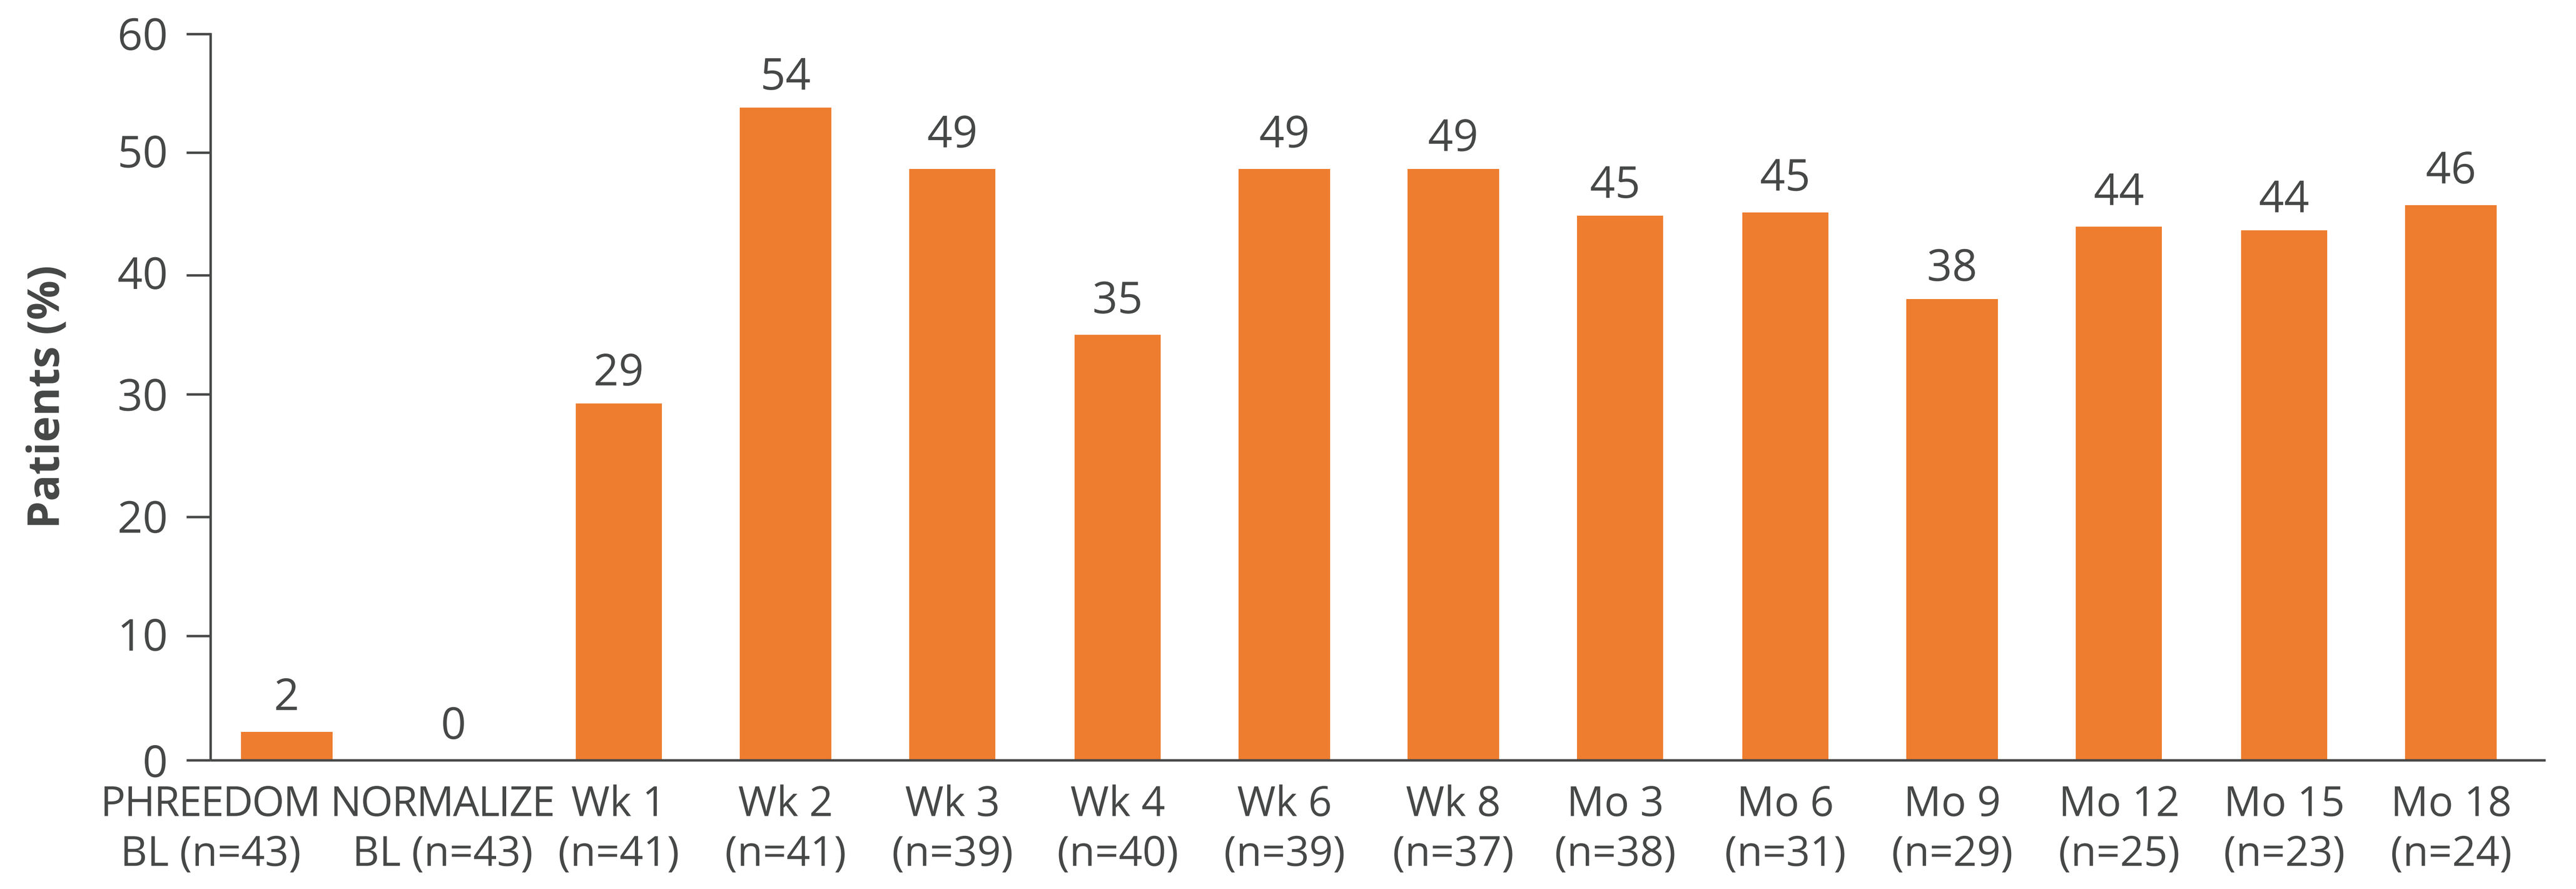

Patients (%)
60
50
40
30
20
10
0
2
0
29
54
49
35
49
49
45
45
38
44
44
46
PHREEDOM BL (n=43)
NORMALIZE BL (n=43)
Wk 1 (n=41)
Wk 2 (n=41)
Wk 3 (n=39)
Wk 4 (n=40)
Wk 6 (n=39)
Wk 8 (n=37)
Mo 3 (n=38)
Mo 6 (n=31)
Mo 9 (n=29)
Mo 12 (n=25)
Mo 15 (n=23)
Mo 18 (n=24)

BL, baseline.

Safety

- Adverse events (AE) reported for this subset were similar to those seen in other tenapanor studies; no new safety issues were identified (**Table 2**). Diarrhea was the most common AE in this subset.

Table 2. Adverse Events

	Patients (N=43)
Any TEAE	35 (81.4)
Any SAE	15 (34.9)
TEAE leading to tenapanor discontinuation	8 (18.6)
TEAE leading to sevelamer discontinuation	7 (16.3)
TEAEs with incidence >3% by PT	
Diarrhea	18 (41.9)
Fall	5 (11.6)
Hyperkalemia	4 (9.3)
COVID-19 infection	3 (7.0)
Hypertension	3 (7.0)
Troponin increased	3 (7.0)
Hypotension	2 (4.7)
Pain in extremity	2 (4.7)
Vomiting	2 (4.7)
Sepsis	2 (4.7)
Constipation	2 (4.7)

Data provided as n (%).
COVID-19, coronavirus disease 2019; PT, preferred term; SAE, serious adverse event; TEAE, treatment-emergent adverse event.

Conclusions

The NORMALIZE study confirmed the findings from a 4-week study, AMPLIFY, that tenapanor in combination with sevelamer can enable a greater percentage of patients to achieve sP target—in this study, ≤4.5 mg/dL—with no new safety issues.

These data also demonstrate that tenapanor in combination with sevelamer can achieve long-term sP control in patients when sevelamer monotherapy fails to normalize sP.

Disclosures

Daniel Weiner is the Medical Director of Clinical Research for Dialysis Clinic, Inc. (DCI) with institutional support from DCI paid to Tufts Medical Center. Dr. Weiner was a site principal investigator for several tenapanor clinical trials from Ardelyx, Inc. (not compensated). Robert Lynn is a principal investigator for Ardelyx, Inc. David Spiegel, Susan Edelstein, Yang Yang, and David Rosenbaum are employees of Ardelyx, Inc.

Dr. Weiner can be contacted for further information on this study at dweiner@tuftsmedicalcenter.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

References

1. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
2. Spencer AG et al. *Sci Transl Med*. 2014;6:227ra36.
3. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
4. Pergola PE et al. *J Am Soc Nephrol*. 2021;32:1465-73.
5. Rosenbaum DP et al. Poster presented at: National Kidney Foundation Spring Clinical Meetings; April 6-10, 2022; Boston, MA.
6. Block GA et al. *Kidney 360*. 2021;2:1600-10.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

American Society of Nephrology Kidney Week (ASN 2023); November 2–5, 2023; Philadelphia, PA, USA

MED-US--2400031