Poster: TH-PO147

Patient Education Improves Adherence to Tenapanor Treatment in OPTIMIZE Study

Stephen Fadem,[1] David Tietjen,[2] David Spiegel,[3] Susan Edelstein,[3] Yang Yang,[3] and David Rosenbaum[3]

[1]Kidney Associates, Houston, TX, USA; [2]Nephrology Consultants, Huntsville, AL, USA; [3]Ardelyx, Inc., Waltham, MA, USA

Introduction

- Per the Dialysis Outcomes and Practice Patterns Study practice monitor, approximately 70% of patients with chronic kidney disease (CKD) on dialysis have serum phosphate (sP) levels >4.5 mg/dL in any given month despite treatment for hyperphosphatemia, suggesting that current treatment strategies are insufficient.[1,2]
- Tenapanor is a first-in-class, minimally absorbed phosphate absorption inhibitor that targets the paracellular pathway, the primary pathway of phosphate absorption in the gastrointestinal tract by selectively inhibiting sodium hydrogen exchanger isoform 3 (NHE3).[3-6]
- Tenapanor is approved for the control of sP in adult patients with CKD on maintenance dialysis.
- Tenapanor has demonstrated efficacy and acceptable safety in 2 pivotal monotherapy hyperphosphatemia clinical studies, BLOCK (NCT02675998)[7] and PHREEDOM (NCT03427125).[8]
 - Diarrhea was the most common adverse event, observed in >5% of patients.
- The OPTIMIZE study (NCT04549597) was designed to evaluate how to best manage the treatment of hyperphosphatemia in patients with CKD on dialysis using tenapanor.[9]
- Here, the benefit of patient education on adherence to prescribed tenapanor treatment during the OPTIMIZE study was evaluated.

Methods

- Methods for the trials have been previously described.[7-9] In each trial, patients randomized to the tenapanor dose-titration group were started on tenapanor 30 mg twice daily (bid).
- Data from the tenapanor dose-titration group was compared in the analysis period of BLOCK (8-week treatment period [TP]), PHREEDOM (first 8 weeks of TP), and OPTIMIZE (10-week TP).
- In OPTIMIZE, in addition to standard-of-care health education including dietary instruction, patients were educated at the start of the study about tenapanor by the study coordinator, who reviewed a patient brochure (Box) with the patient; instructions included how to take tenapanor, what they may experience while taking tenapanor, medications to be discontinued before starting tenapanor, and how best to mitigate the potential onset of loose stools or diarrhea.

Box: Patient Brochure

- The study coordinator reviewed the Patient Brochure with the patient before starting tenapanor, and the patient kept the brochure for future reference.
- The Patient Brochure presented the following questions about the treatment with answers:
 - What is tenapanor?
 - How do I take tenapanor?
 - How is tenapanor different from phosphate binders?
 - If I'm on a phosphate binder, do I continue taking the phosphate binder?
 - Does tenapanor block all the phosphorus I eat?
 - What may I experience with tenapanor?
 - When may a change in my bowel movements happen?
 - Do patients typically find the possible change in bowel movements bothersome?
 - What should I do to manage the possible increase in bowel movements or loose stools with tenapanor?

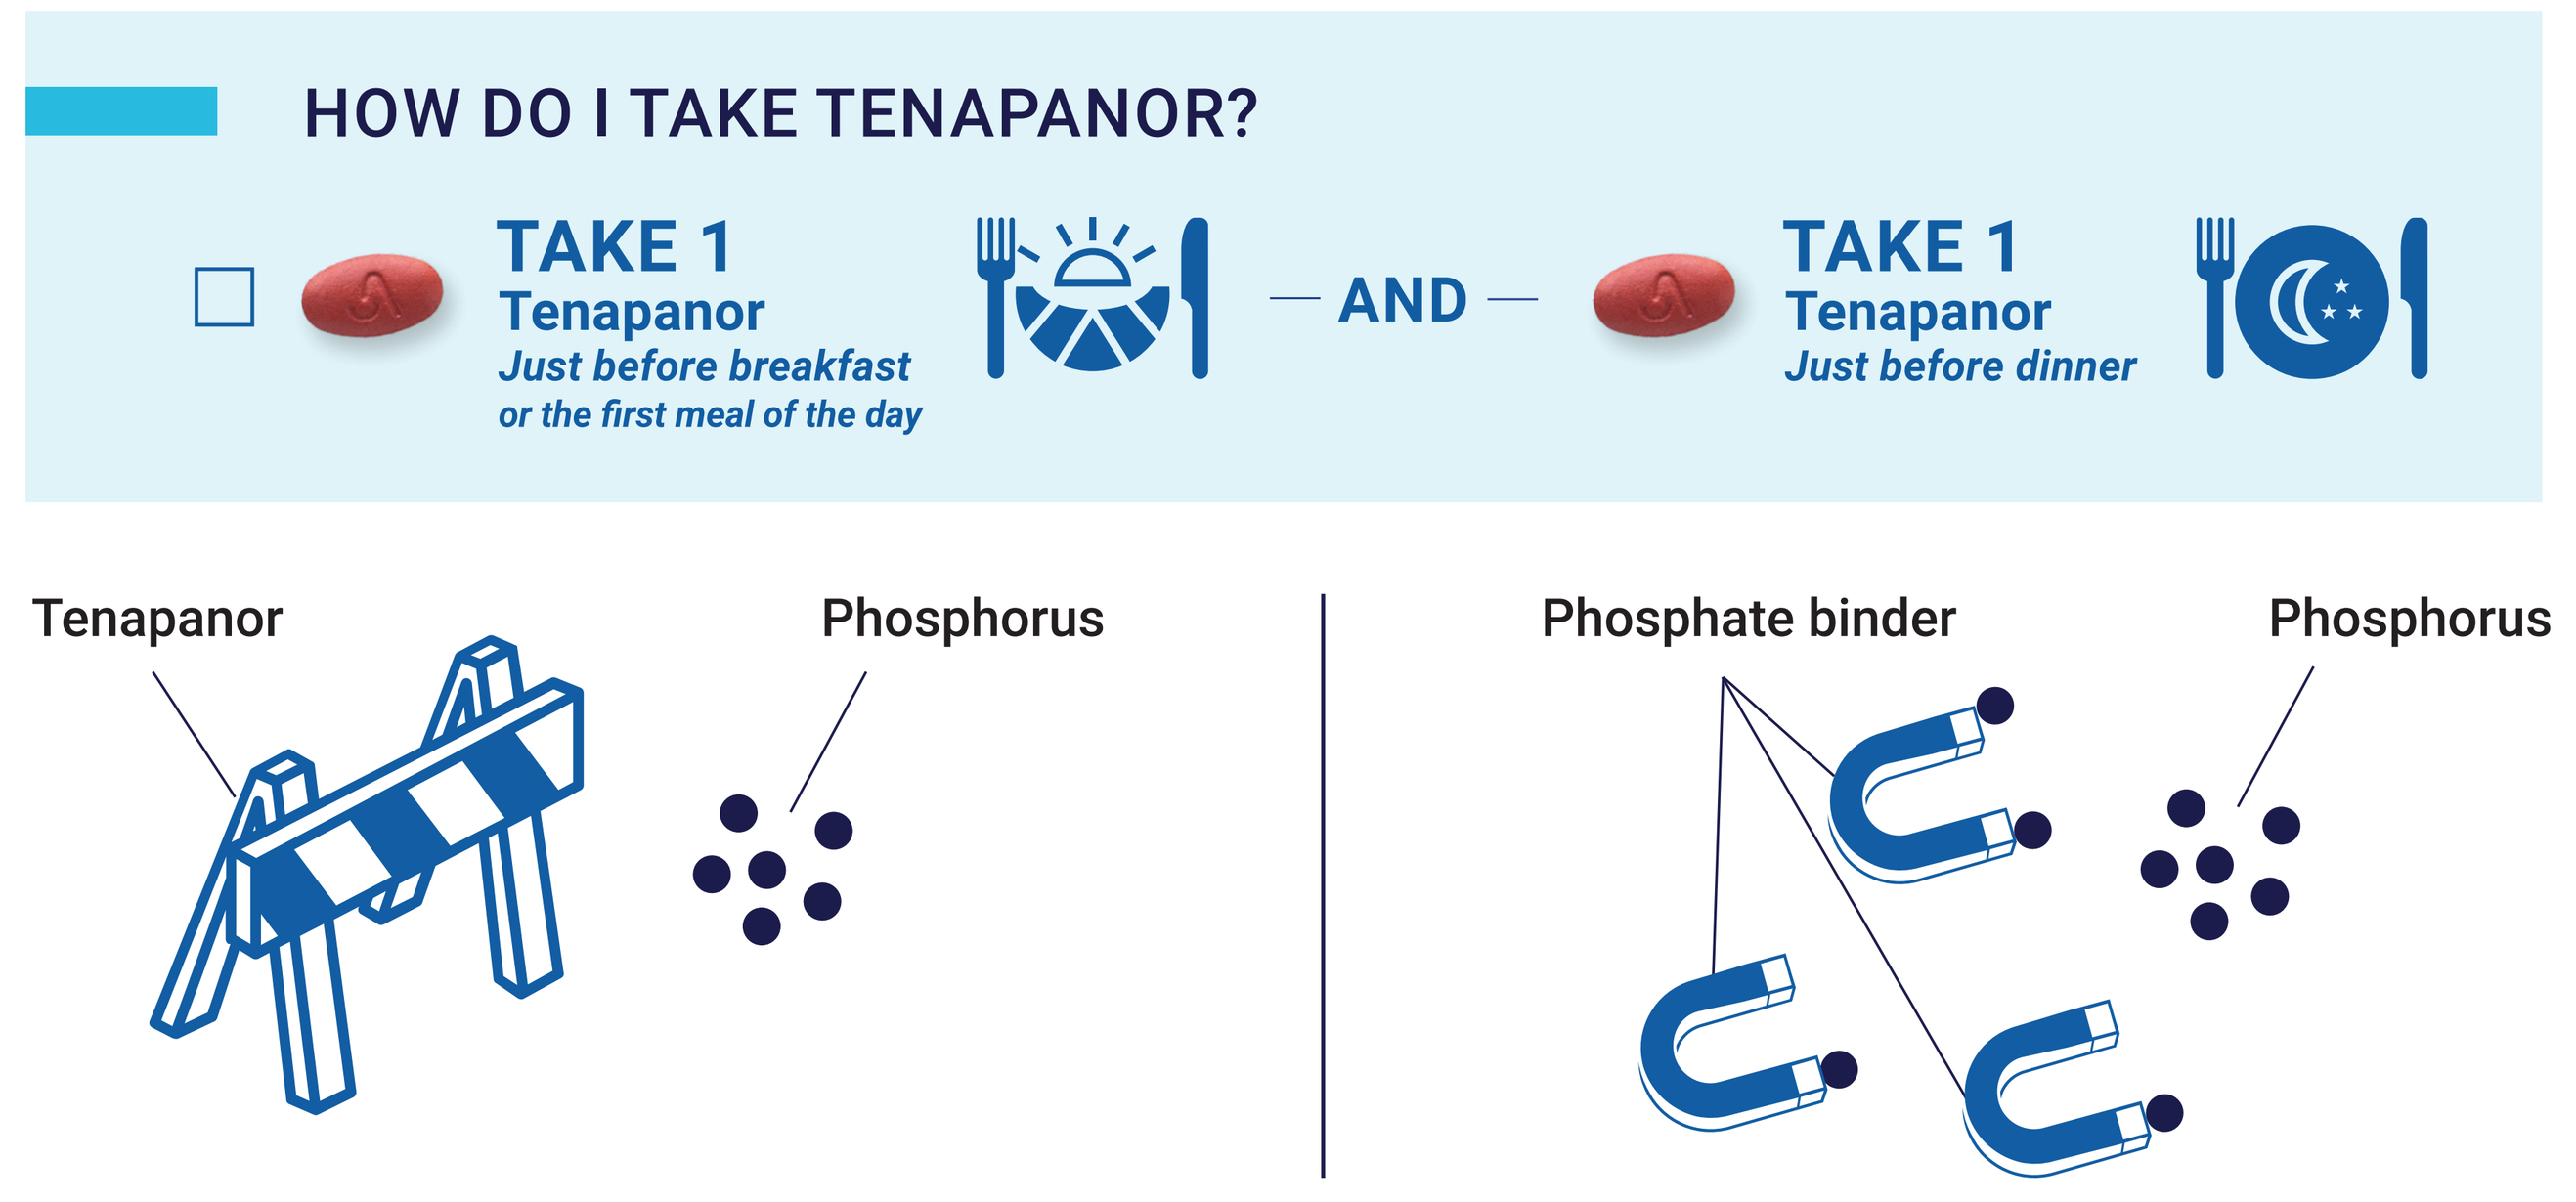
HOW DO I TAKE TENAPANOR?
TAKE 1 Tenapanor Just before breakfast or the first meal of the day
— AND —
TAKE 1 Tenapanor Just before dinner
Tenapanor
Phosphorus
Phosphate binder
Phosphorus

Results

- Demographics of patients in the tenapanor dose-titration group were generally similar across the 3 studies, BLOCK, PHREEDOM, and OPTIMIZE (**Table 1**).

Table 1. Demographics and Baseline Characteristics of Tenapanor Dose Titration Groups of the BLOCK, PHREEDOM, and OPTIMIZE Studies

	Study		
Demographic/Characteristic	**BLOCK (N=71)**	**PHREEDOM (N=419)**	**OPTIMIZE (N=333)**
Age, years	54.2 (10.9)	57.7 (12.6)	53.0 (12.1)
Male sex, n (%)	48 (67.6)	265 (63.2)	227 (68.2)
Race, n (%)			
White	30 (42.3)	189 (45.1)	146 (43.8)
Black or African American	40 (56.3)	195 (46.5)	146 (43.8)
Other[a]	1 (1.4)	35 (8.4)	41 (12.3)
Ethnicity, n (%)			
Hispanic or Latino	18 (25.4)	115 (27.4)	93 (27.9)
Not Hispanic or Latino	53 (74.6)	302 (72.1)	240 (72.1)
BMI, kg/m²	33.4 (8.1)	31.3 (7.5)	32.2 (8.3)
Hemodialysis, n (%)	71 (100)	376 (89.7)	268 (80.5)
Duration of dialysis treatment, months	57.1 (57.1)	54.2 (51.0)	54.1 (50.1)
Baseline sP, (mg/dL)	7.6 (1.4)	7.4 (1.4)	7.1 (1.1)

Unless otherwise indicated, data are mean (SD).
BMI, body mass index; sP, serum phosphate.
[a]Includes Asian, Native American, or Alaskan, Native Hawaiian or Pacific Islander, other and unknown.

MED-US--2400029

- The comparison of diarrhea incidence among the tenapanor dose-titration groups of BLOCK, PHREEDOM, and OPTIMIZE shows that:
 - As expected, given the mechanism of action and data from past trials,[10-12] most diarrhea events during the 8- or 10-week analysis period (75.7%-79.6%) occurred within the first 2 weeks of treatment in all 3 studies, and most cases were mild to moderate.
 - Overall, the diarrhea incidence was lower during the analysis period in OPTIMIZE (39.3%) than in BLOCK (47.9%) and PHREEDOM (47.7%; **Figure 1**).
 - Similarly, during the first 2 weeks of study treatment, the diarrhea incidence was also lower in OPTIMIZE (33.6%) vs BLOCK (38.0%) and PHREEDOM (39.4%; **Figure 1**).
 - The incidence of diarrhea leading to tenapanor discontinuation was lower during the 10-week analysis period of OPTIMIZE (6.0%) than the 8-week analysis periods of BLOCK (7.0%) or PHREEDOM (13.4%; **Figure 2**).

Figure 1. Incidence of Diarrhea in Tenapanor Dose-Titration Groups of the BLOCK, PHREEDOM, and OPTIMIZE Studies

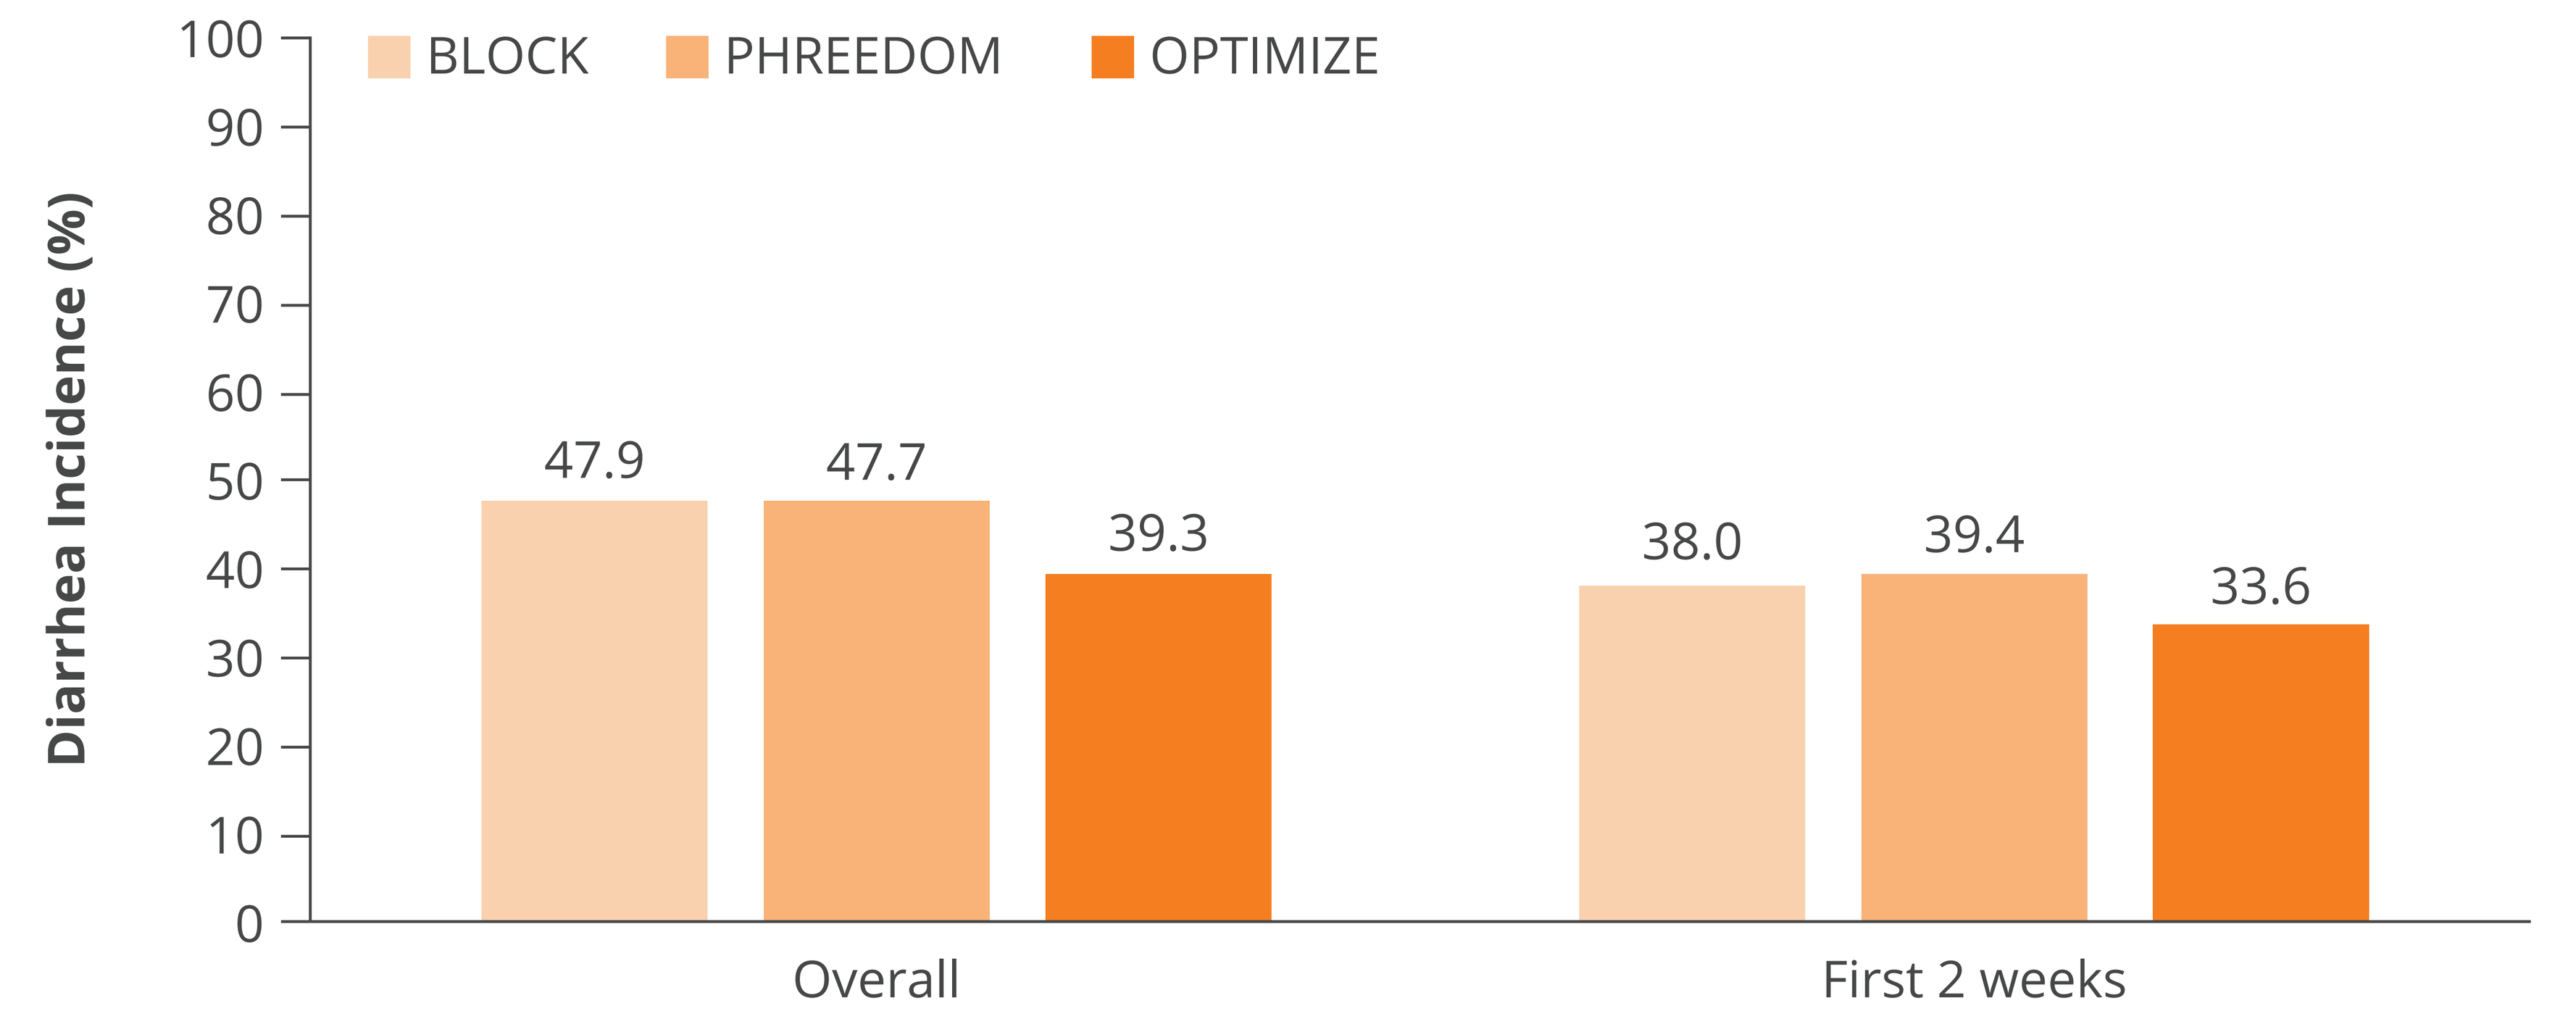
BLOCK
PHREEDOM
OPTIMIZE
Diarrhea Incidence (%)
100
90
80
70
60
50
40
30
20
10
0
47.9
47.7
39.3
38.0
39.4
33.6
Overall
First 2 weeks

Figure 2. Incidence of Diarrhea Leading to Tenapanor Discontinuation in Tenapanor Dose-Titration Groups of the BLOCK, PHREEDOM, and OPTIMIZE Studies

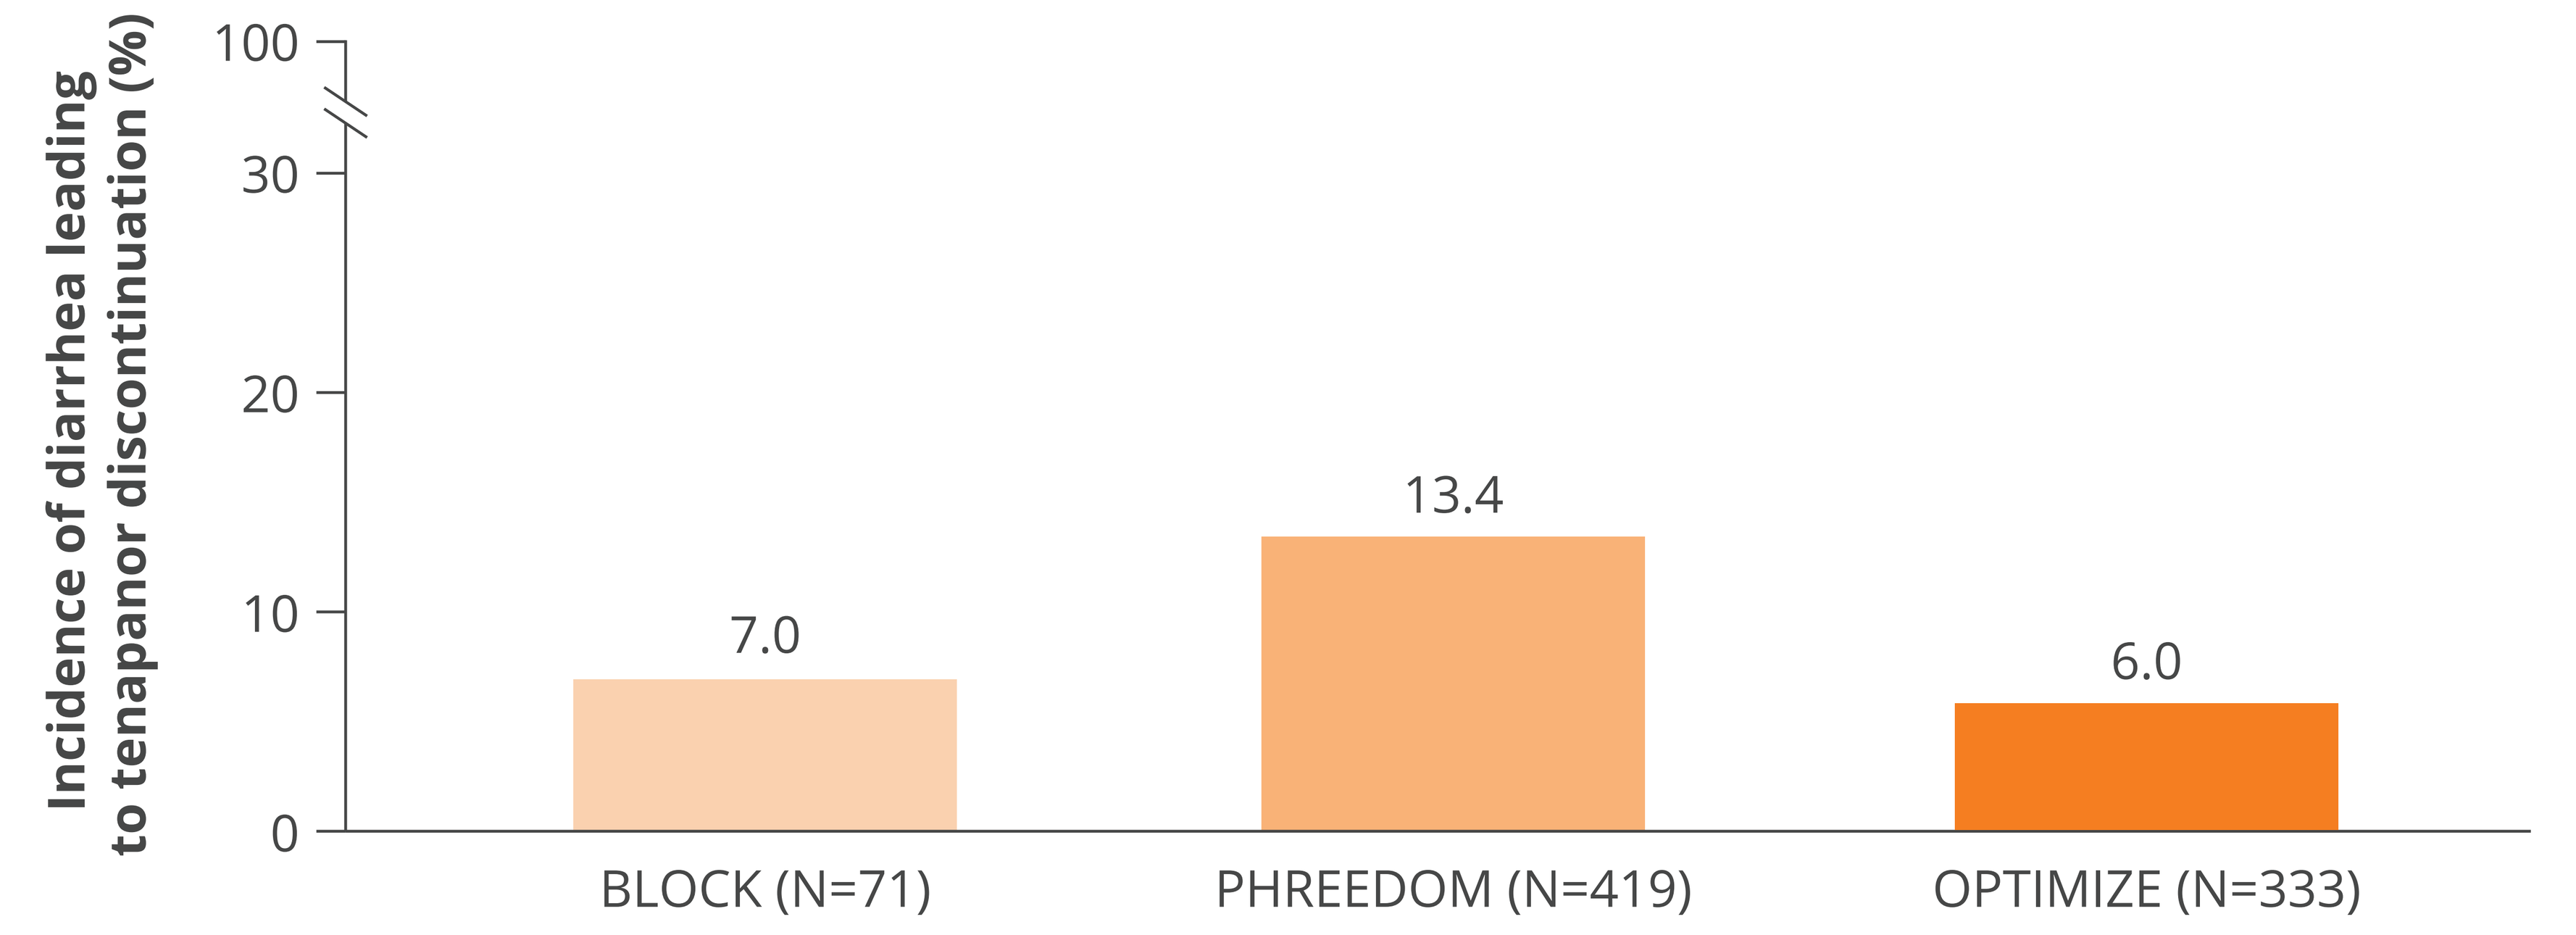
Incidence of diarrhea leading to tenapanor discontinuation (%)
100
30
20
10
0
7.0
13.4
6.0
BLOCK (N=71)
PHREEDOM (N=419)
OPTIMIZE (N=333)

Conclusions

Patient education appears to improve tenapanor medication adherence and reduce diarrhea in dialysis patients.

Specifically, because many patients on dialysis take medications to alleviate constipation, advising patients to discontinue these medications before starting tenapanor may also help reduce the occurrence of diarrhea.

Disclosures

Stephen Z. Fadem is a principal investigator for Ardelyx, Inc. David Tietjen has nothing to disclose. David Spiegel, Susan Edelstein, Yang Yang, and David Rosenbaum are employees of Ardelyx, Inc.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr Fadem can be contacted for further information on this study at fadem@bcm.edu.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

References

1. Clegg DJ et al. *Nephrol Renal Diseases*. 2022;7:1-7.
2. Guedes M et al. *Kidney Med*. 2023;5:100584.
3. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
4. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
5. Spencer AG et al. *Sci Transl Med*. 2014;6:227ra236.
6. Labonté ED et al. *J Am Soc Nephrol*. 2015;26:1138-49.
7. Block GA et al. *J Am Soc Nephrol*. 2019;30:641-52.
8. Block GA et al. *Kidney360*. 2021;2:1600-10.
9. Edelstein S et al. Presented at National Kidney Foundation Spring Clinical Meetings; April 6-10, 2022; Boston, MA, USA.
10. Chey WD et al. *Am J Gastroenterol*. 2017;112:763-74.
11. Chey WD et al. *Am J Gastroenterol*. 2020;115:281-93.
12. Chey WD et al. *Am J Gastroenterol*. 2021;116:1294-1303.

American Society of Nephrology Kidney Week (ASN 2023); November 2–5, 2023; Philadelphia, PA, USA

MED-US--2400029