Treatment Response to Tenapanor Categorized by Age and Comorbidities: A Post Hoc Analysis of the PHREEDOM Study

Daniel E. Weiner,[1] David M. Spiegel,[2] Yang Yang,[2] David P. Rosenbaum,[2] and Susan Edelstein[2]

[1]Tufts Medical Center, Boston, MA, USA; [2]Ardelyx, Inc., Waltham, MA, USA

Background

- Management of serum phosphate (P) in patients with chronic kidney disease (CKD) receiving maintenance dialysis is challenging.
 - Current approaches include restriction of dietary phosphorus and phosphate binder therapy.[1,2]
- Tenapanor is a first-in-class, minimally absorbed phosphate absorption inhibitor that targets the primary pathway of phosphate absorption in the gastrointestinal tract, the paracellular pathway, by selectively inhibiting sodium/hydrogen exchanger isoform 3 (NHE3).[3-5]
 - Tenapanor is approved as add-on therapy to reduce serum P in patients with CKD on dialysis who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.[6]
- The randomized, phase 3 PHREEDOM trial (NCT03427125) demonstrated that tenapanor monotherapy safely reduced serum P in patients on maintenance dialysis. Sevelamer monotherapy was employed as an active safety control arm.[7]
- Diabetes, hypertension, and cardiovascular complications are common comorbidities in patients on dialysis and can often coexist in the same patient.[8-10]
 - There are complex interactions that can occur with the treatment of these comorbidities and elevated serum P.[8]
- Here, we evaluated trends in serum P in adult patients on dialysis treated with tenapanor over the 26-week treatment period of the PHREEDOM trial (NTC03427125), categorized by baseline comorbidities and age.

Methods

- PHREEDOM enrolled patients on maintenance dialysis with serum P ≥6.0 and ≤10.0 mg/dL and a ≥1.5-mg/dL increase in serum P after phosphate binder washout.
- Patients were randomized 3:1 to receive tenapanor 30 mg twice a day (bid) during a 26-week randomized treatment period (RTP) or sevelamer (active safety control) per standard of care for 52 weeks (**Figure 1**).[7]

Figure 1: PHREEDOM Study Design

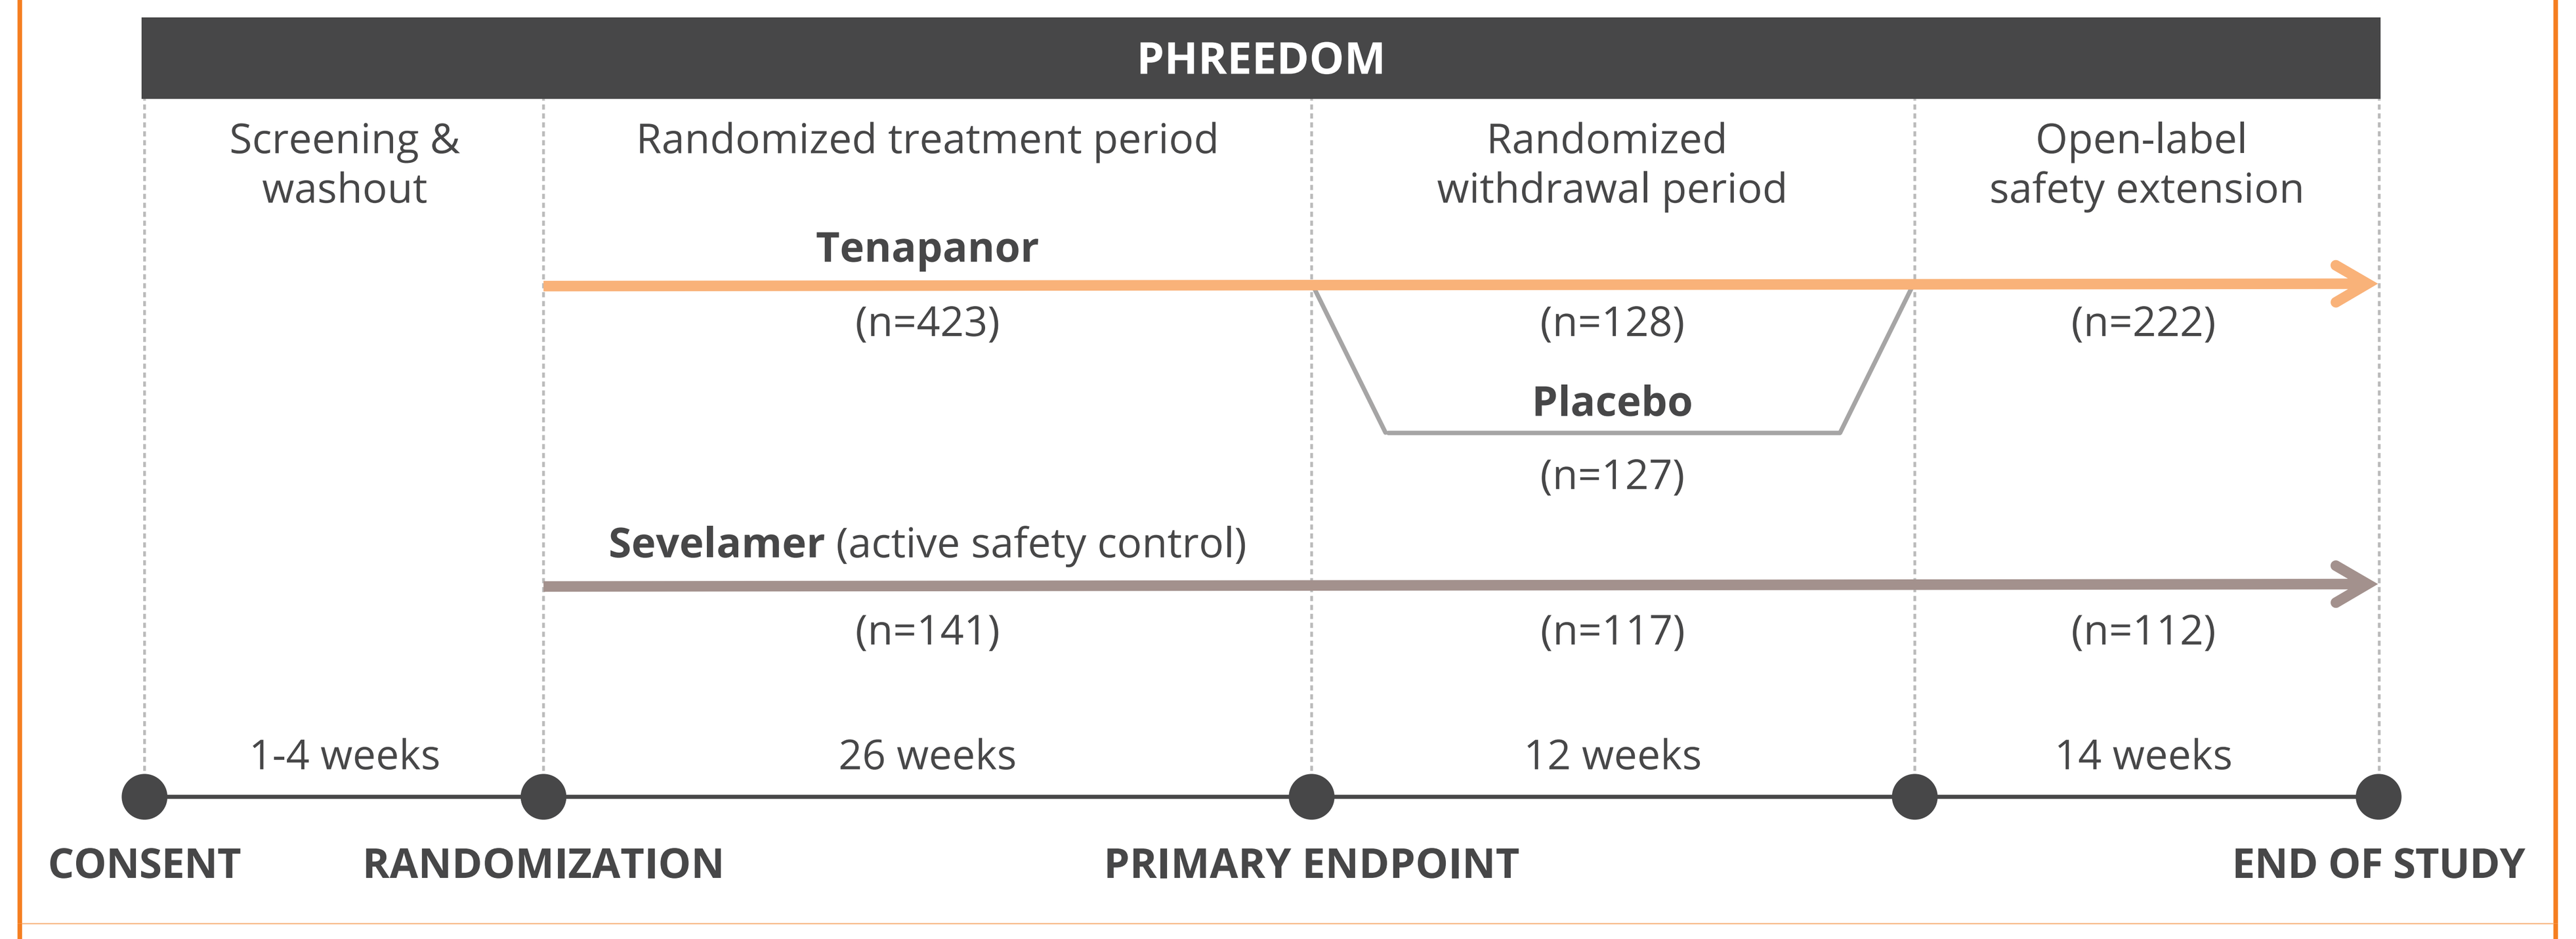
PHREEDOM
Screening & washout
Randomized treatment period
Randomized withdrawal period
Open-label safety extension
Tenapanor
(n=423)
(n=128)
Placebo
(n=127)
(n=222)
Sevelamer (active safety control)
(n=141)
(n=117)
(n=112)
1-4 weeks
26 weeks
12 weeks
14 weeks
CONSENT
RANDOMIZATION
PRIMARY ENDPOINT
END OF STUDY

The number of patients who received ≥1 dose of study drug (safety analysis set) during each study period is shown.

- Investigators could titrate tenapanor dosage in 10-mg increments to a minimum of 10 mg bid or a maximum of 30 mg bid, based on serum P concentration and gastrointestinal tolerability.
- Outcomes were evaluated in subgroups of patients with baseline diabetes, hypertension, heart failure or cardiac disease, and age subgroups (aged <65 or ≥65 years) receiving tenapanor during the 26-week RTP.

Results

Patients

- Among 423 patients randomized to receive tenapanor during the RTP, 419 were included in the safety analysis set (**Table 1**).
- Of the 419 tenapanor-treated patients in the safety analysis set, about two-thirds of patients were <65 years of age, most had hypertension, and baseline mean serum P was 7.4 mg/dL.

Table 1: Baseline Demographics and Characteristics by Baseline Comorbidity and Age (Tenapanor-Treated Patients in Safety Analysis Set)

	Baseline comorbidity				Baseline age	
Baseline demographic or characteristic	**Diabetes (N=264)**	**Hypertension (N=400)**	**Heart failure (N=83)**	**Any cardiac disorder (N=231)**	**<65 Years (N=287)**	**≥65 Years (N=132)**
Age (years)	60.2 (10.5)	57.9 (12.5)	59.4 (11.6)	59.4 (12.2)	51.4 (9.6)	71.5 (5.5)
<65 years, n (%)	170 (64.4)	274 (68.5)	52 (62.7)	148 (64.1)	287 (100)	0
≥65 years, n (%)	94 (35.6)	126 (31.5)	31 (37.3)	83 (35.9)	0	132 (100)
Male, n (%)	170 (64.4)	254 (63.5)	55 (66.3)	144 (62.3)	183 (63.8)	82 (62.1)
Race, n (%)						
White	121 (45.8)	180 (45.0)	34 (41.0)	106 (45.9)	125 (43.6)	64 (48.5)
Black or African American	121 (45.8)	188 (47.0)	44 (53.0)	109 (47.2)	137 (47.7)	58 (43.9)
Other[a]	22 (8.3)	32 (8.0)	5 (6.0)	16 (6.9)	25 (8.7)	10 (7.6)
Ethnicity, n (%)						
Non-Hispanic/Latino	185 (70.1)	288 (72.0)	69 (83.1)	175 (75.8)	215 (74.9)	87 (65.9)
Hispanic/Latino	77 (29.2)	110 (27.5)	14 (16.9)	55 (23.8)	72 (25.1)	43 (32.6)
Not reported/unknown	2 (0.8)	2 (0.5)	0	1 (0.4)	0	2 (1.5)
BMI (kg/m²)	32.4 (7.8)	31.4 (7.5)	32.9 (7.9)	31.9 (7.5)	32.3 (7.9)	29.1 (6.1)
Duration since ESKD diagnosis (years)	4.3 (3.7)	4.7 (4.4)	4.4 (3.6)	4.9 (4.2)	4.8 (4.6)	4.8 (4.1)
Baseline serum P (mg/dL)	7.3 (1.4)	7.4 (1.4)	7.4 (1.3)	7.3 (1.3)	7.6 (1.5)	7.0 (1.2)
iFGF23, median (IQR) (pg/mL)	5784.2 (1707.8, 14579.5)	6642.9 (2188.2, 17385)	6358.3 (2097.7, 16604)	5299.2 (1748, 13696)	8307.6 (2213.2, 20735)	4469.7 (1640.6, 12937)
PTH (pg/mL)	395.0 (217.1)	421.6 (250.3)	390.5 (224.6)	436.2 (257.1)	430.8 (253.6)	400.8 (248.9)
Ca (mg/dL)	8.3 (0.8)	8.4 (0.8)	8.4 (0.7)	8.4 (0.7)	8.4 (0.8)	8.4 (0.8)
Kt/V	1.6 (0.3)	1.6 (0.3)	1.6 (0.3)	1.6 (0.3)	1.6 (0.3)	1.7 (0.4)

Unless otherwise indicated, data are mean (SD). Baseline was defined as the last measurement collected before the first dose of tenapanor in the study. Percentage values may not total 100 due to rounding.
[a]Includes Native American or Alaskan, Asian, and Native Hawaiian or other Pacific Islander.
BMI, body mass index; Ca, calcium; ESKD, end-stage kidney disease; iFGF23, intact fibroblast growth factor 23; IQR, interquartile range; Kt/V, a measure of dialysis dose; P, phosphate; PTH, parathyroid hormone.

Treatment Response by Comorbidity

- Regardless of baseline comorbidity, tenapanor-treated patients exhibited similar mean baseline serum P and serum P reductions during PHREEDOM (**Figure 2**).
- 49% of patients with diabetes, 56% of patients with hypertension, 54% of patients with heart failure, and 55% of patients with any cardiac disorder had a serum P reduction of ≥1.2 mg/dL from baseline at week 26 (**Figure 3**).

Figure 2: (A) Mean Serum P Over Time by Comorbidity Subgroup and (B) Mean Change From Baseline in Serum P by Comorbidity Subgroup (ITT Analysis Set)

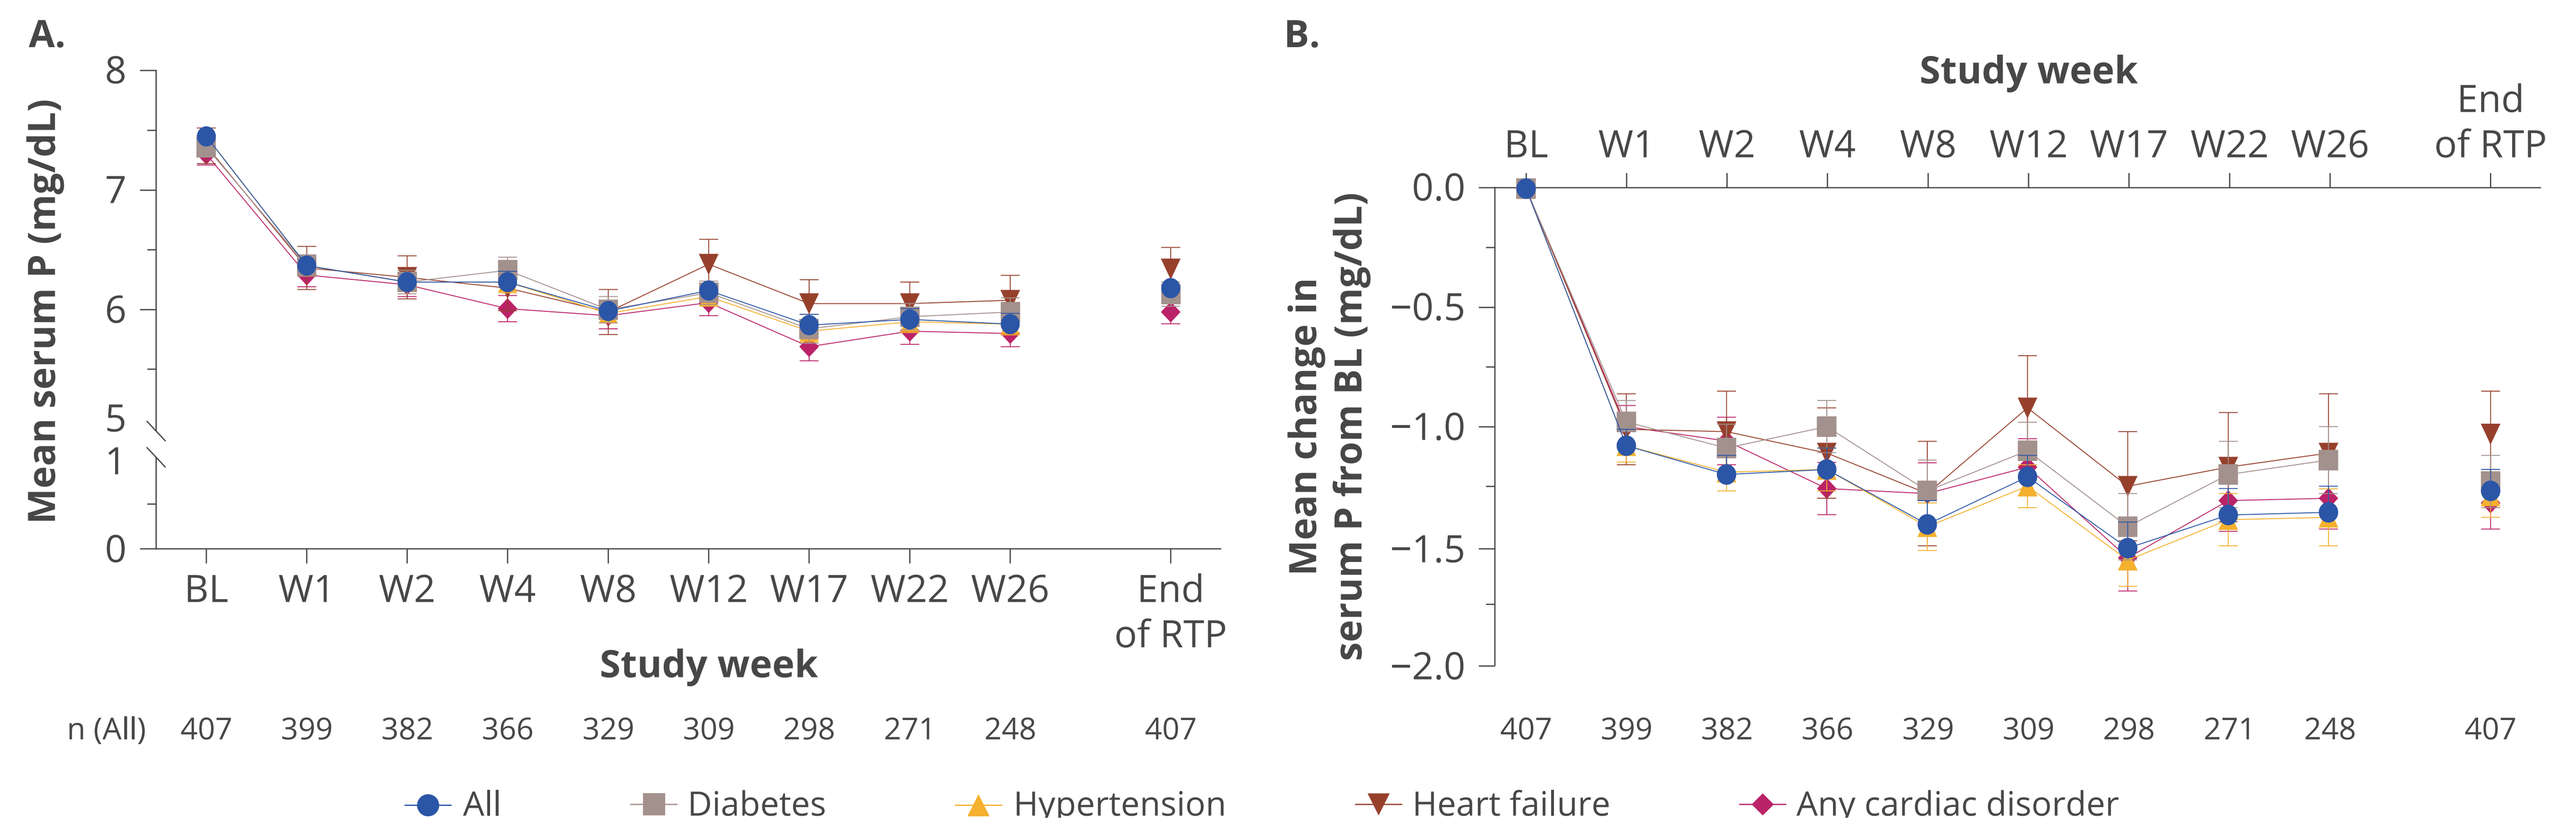
A.
Mean serum P (mg/dL)
8
7
6
5
1
0
BL W1 W2 W4 W8 W12 W17 W22 W26 End of RTP
Study week
n (All) 407 399 382 366 329 309 298 271 248 407
B.
Study week
BL W1 W2 W4 W8 W12 W17 W22 W26 End of RTP
Mean change in serum P from BL (mg/dL)
0.0
−0.5
−1.0
−1.5
−2.0
407 399 382 366 329 309 298 271 248 407
All
Diabetes
Hypertension
Heart failure
Any cardiac disorder

Error bars depict ± SE. End of RTP is the last observed measurement for each patient during the 26-week period.
BL, baseline; P, phosphate; ITT, intent-to-treat; RTP, randomized treatment period; W, week.

Figure 3: Proportion of Patients Achieving ≥1.2-mg/dL Reduction in Serum P at Week 26 Based on Observed Data by Comorbidity Subgroup (ITT Analysis Set)

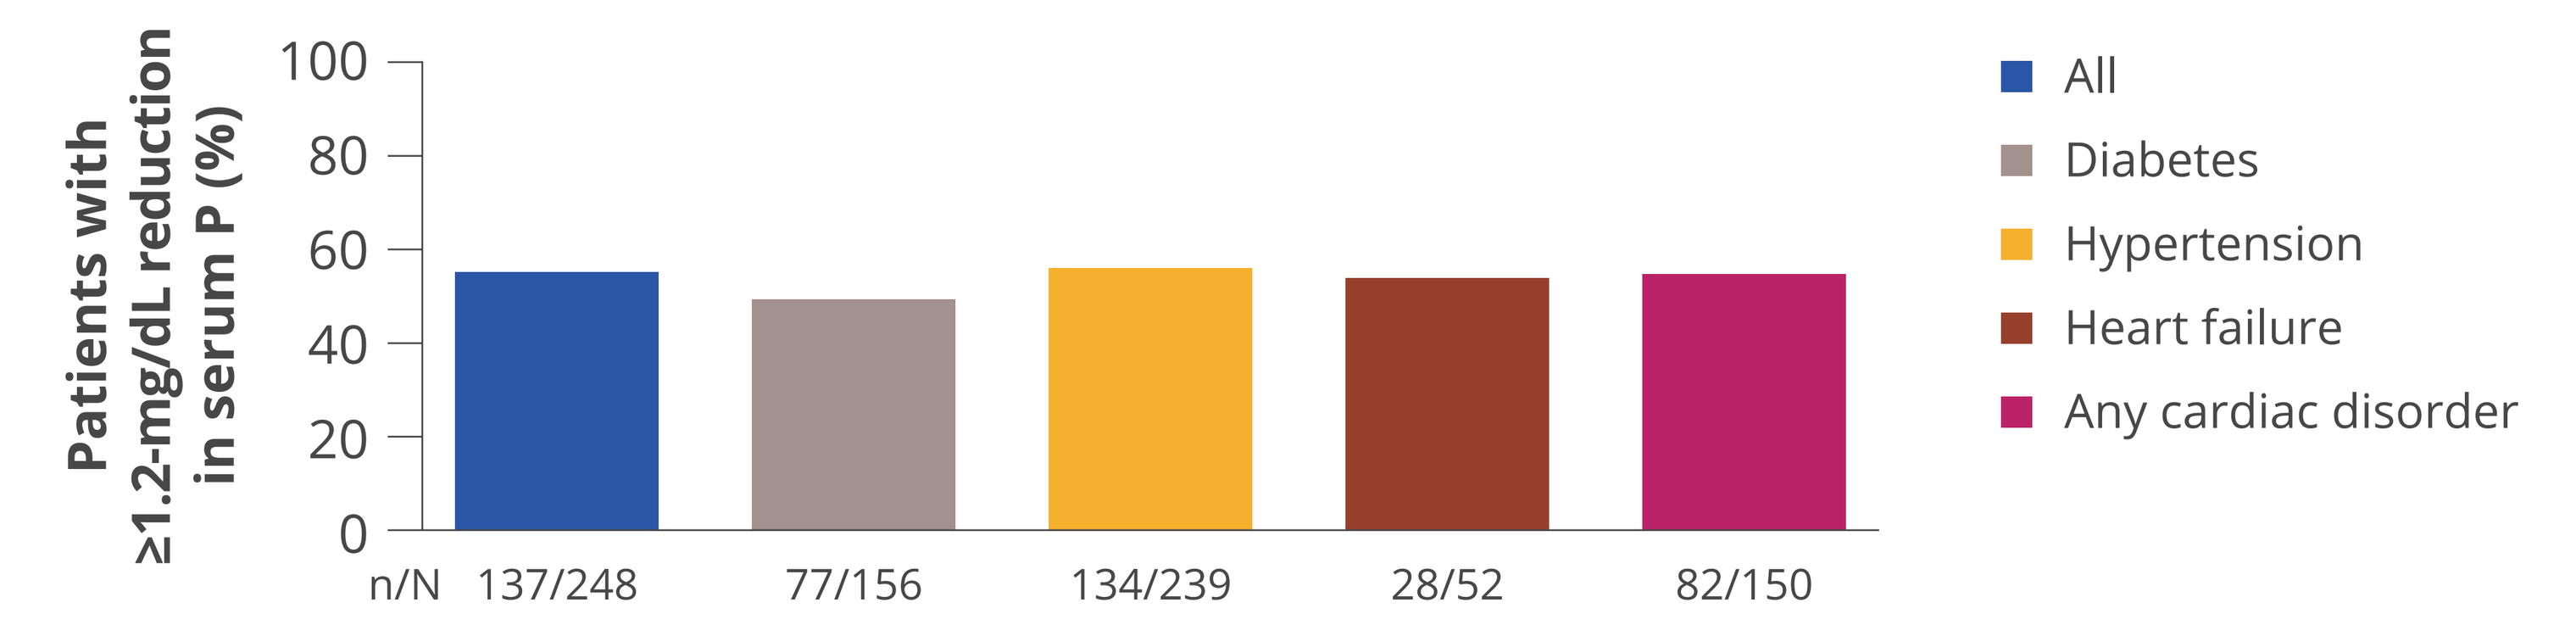
Patients with ≥1.2-mg/dL reduction in serum P (%)
100
80
60
40
20
0
n/N 137/248 77/156 134/239 28/52 82/150
All
Diabetes
Hypertension
Heart failure
Any cardiac disorder

ITT, intent-to-treat; P, phosphate.

Treatment Response by Age

- There was no difference in the mean serum P reduction from baseline between patients aged <65 and ≥65 years (**Figure 4**).
- 55% of patients aged <65 years and 57% of patients aged ≥65 years had a serum P reduction of ≥1.2 mg/dL from baseline at week 26 (**Figure 5**).

Figure 4: (A) Mean Serum P Over Time by Age Subgroup and (B) Mean Change From Baseline in Serum P by Age Subgroup (ITT Analysis Set)

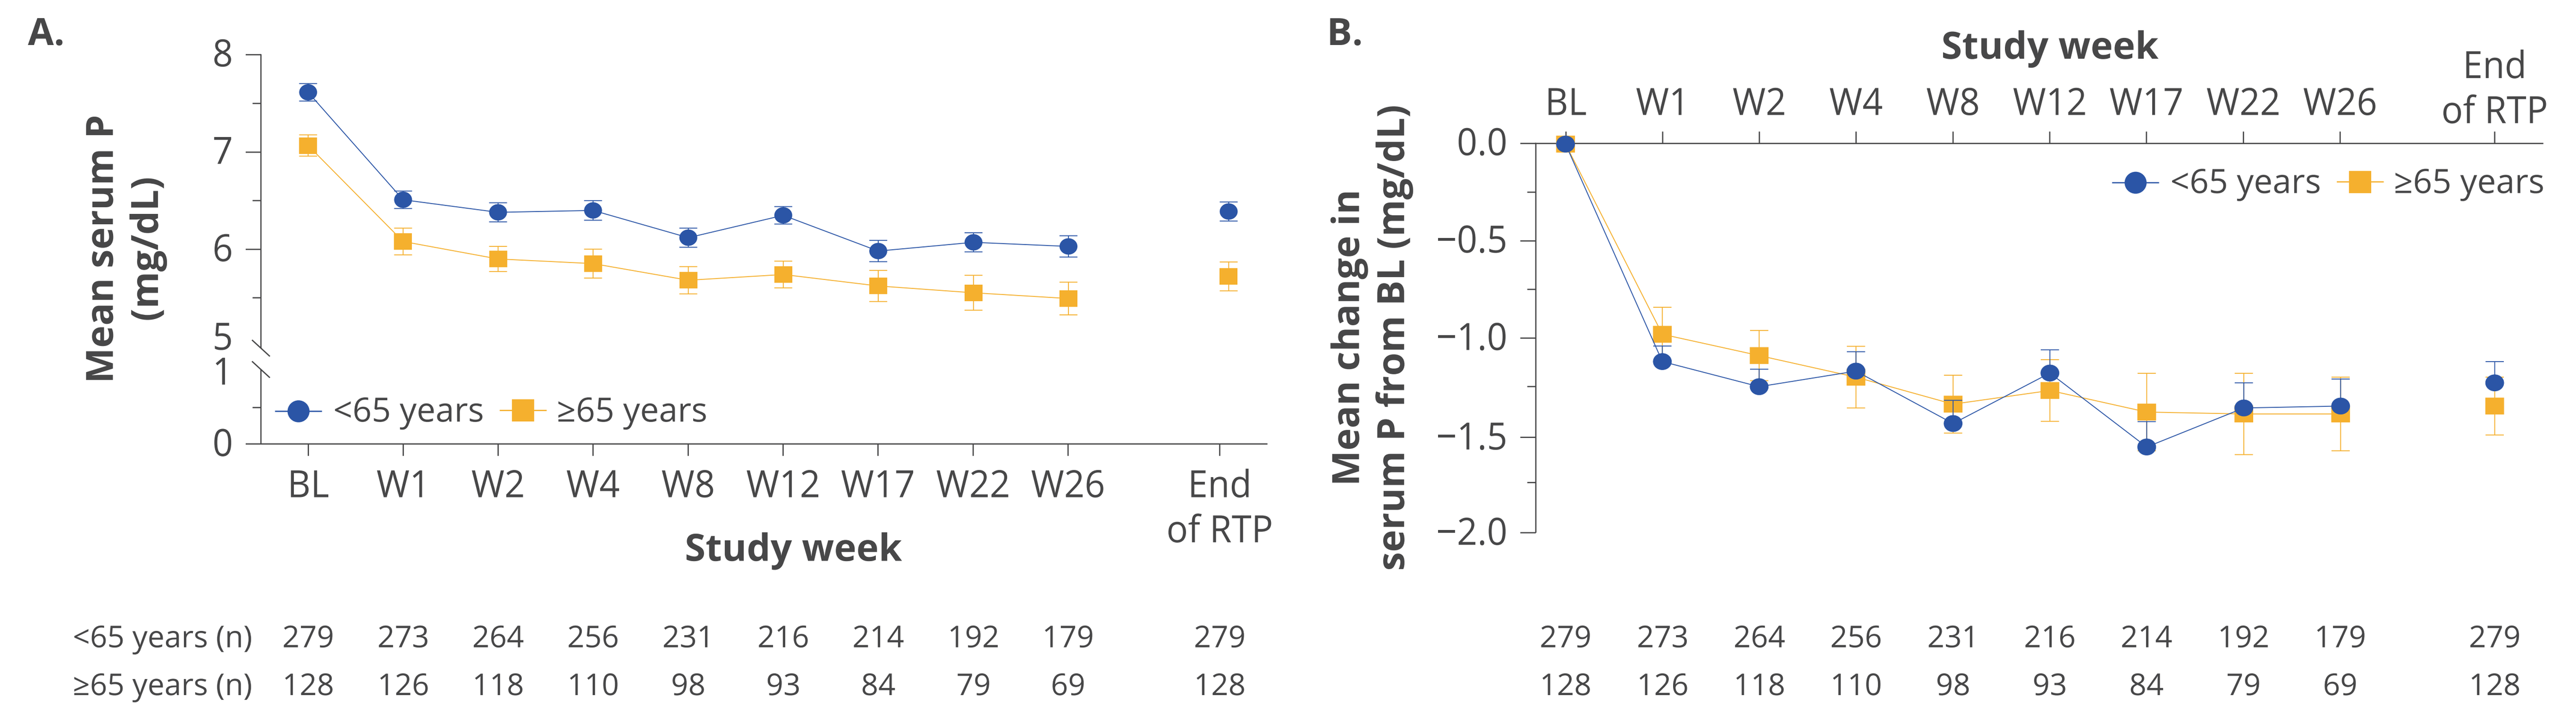
A.
Mean serum P (mg/dL)
8
7
6
5
1
0
<65 years
≥65 years
BL W1 W2 W4 W8 W12 W17 W22 W26 End of RTP
Study week
<65 years (n) 279 273 264 256 231 216 214 192 179 279
≥65 years (n) 128 126 118 110 98 93 84 79 69 128
B.
Study week
BL W1 W2 W4 W8 W12 W17 W22 W26 End of RTP
Mean change in serum P from BL (mg/dL)
0.0
−0.5
−1.0
−1.5
−2.0
<65 years
≥65 years
279 273 264 256 231 216 214 192 179 279
128 126 118 110 98 93 84 79 69 128

Error bars depict ± SE. End of RTP is the last observed measurement for each patient during the 26-week period.
BL, baseline; ITT, intent-to-treat; P, phosphate; RTP, randomized treatment period; W, week.

Figure 5: Proportion of Patients Achieving ≥1.2-mg/dL Reduction in Serum P at Week 26 Based on Observed Data by Age (ITT Analysis Set)

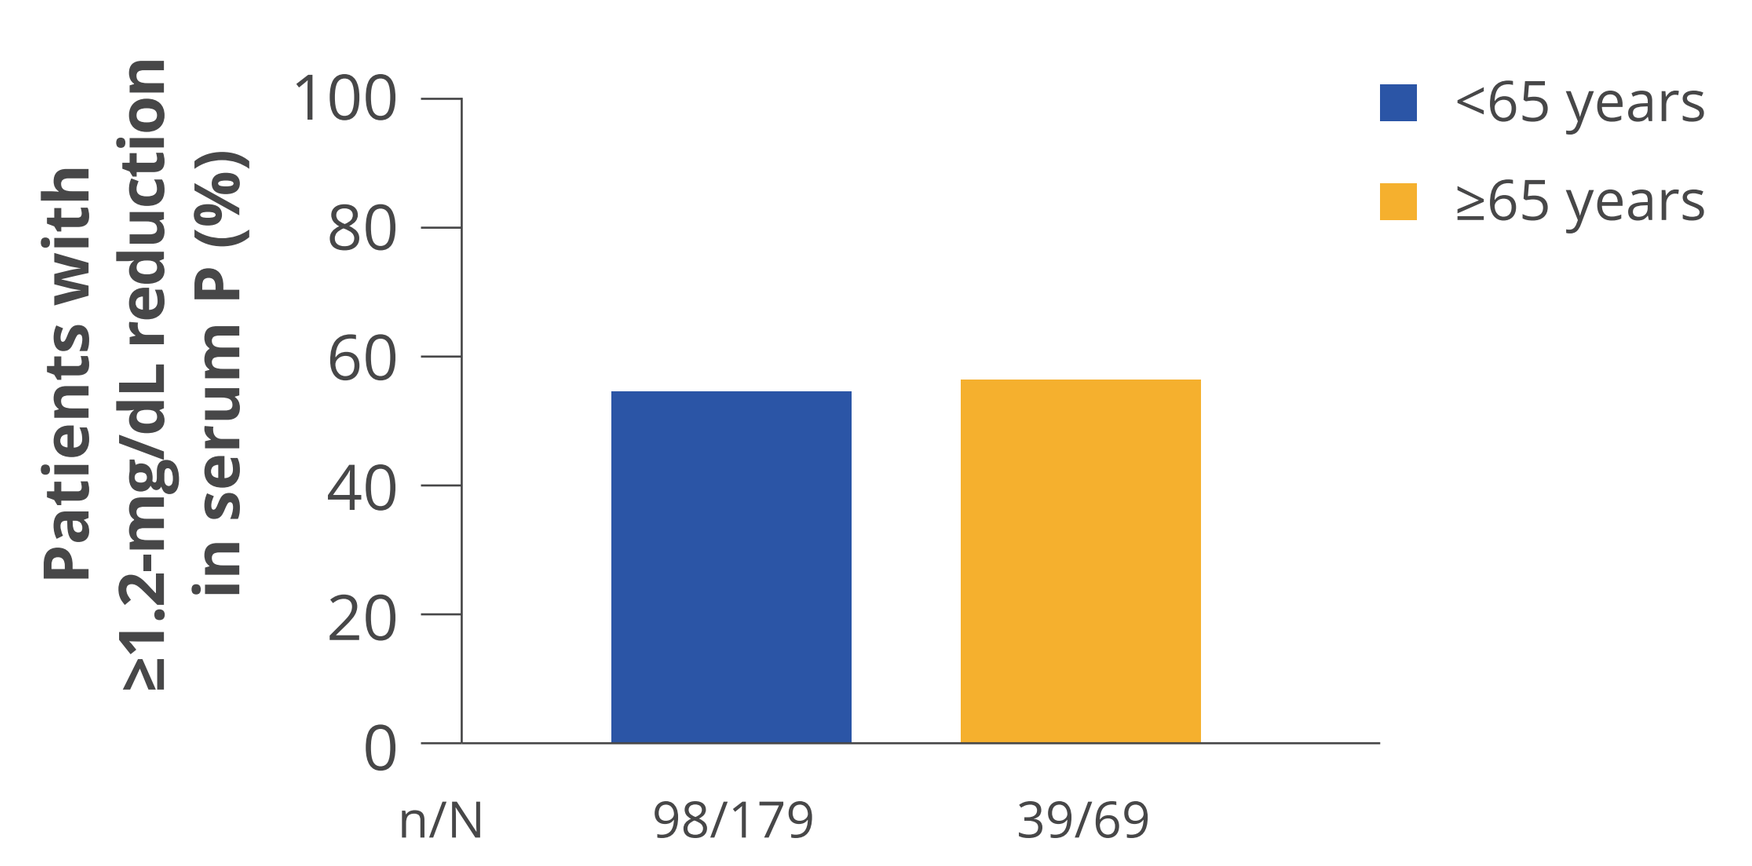
Patients with ≥1.2-mg/dL reduction in serum P (%)
100
80
60
40
20
0
n/N 98/179 39/69
<65 years
≥65 years

ITT, intent-to-treat; P, phosphate.

Safety

- During the 26-week RTP, similar TEAE profiles were observed among the comorbidity and age subgroups.
- Gastrointestinal disorders were the most common TEAEs, with diarrhea reported in approximately 50% of patients across all comorbidity and age subgroups.
 - Only 2 patients reported serious diarrhea.
- During the 26-week RTP, 17% of patients treated with tenapanor and 23% of patients treated with sevelamer experienced a serious adverse event (SAE), and all of these events led to hospitalization.
- Of the patients treated with tenapanor, 24% experienced a TEAE, mostly diarrhea, that led to tenapanor discontinuation.
 - Of the patients treated with sevelamer, 2% experienced a TEAE that led to sevelamer discontinuation.
 - 21% of patients receiving tenapanor experienced an SAE, mostly diarrhea, that led to tenapanor discontinuation. No patients discontinued sevelamer due to an SAE.

Conclusions

Tenapanor demonstrated similar efficacy and safety profiles in patients with CKD on dialysis independent of the presence of comorbidities (eg, diabetes, hypertension, heart failure, cardiovascular disease) and age.

References

1. Fouque D et al. *Drugs*. 2018;78:1171-86.
2. Barreto FC et al. *Kidney Int Rep*. 2019;4:1043-56.
3. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
4. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
5. Labonté ED et al. *J Am Soc Nephrol*. 2015;26:1138-49.
6. XPHOZAH. Prescribing information. Ardelyx Inc.; 2023.
7. Block GA et al. *Kidney360*. 2021;2:1600-10.
8. MacRae C et al. *Br J Gen Pract*. 2021;71:e243-9.
9. Marassi M et al. *Cardiovasc Diabetol*. 2023;22:195.
10. Prichard SS. *Kidney Int*. 2000;57(Suppl. 74):S100-4.

Disclosures

Daniel E. Weiner is the Medical Director of Clinical Research for Dialysis Clinic, Inc. (DCI), with institutional support from DCI paid to Tufts Medical Center. Dr. Weiner was a site Principal Investigator for several tenapanor clinical trials from Ardelyx, Inc. (not compensated). David M. Spiegel, Yang Yang, David Rosembaum, and Susan Edelstein are employees of Ardelyx, Inc.

Acknowledgements

Analysis of the safety data for the comorbidity and age subgroups was completed by Suling Zhao. Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr. Weiner, can be contacted for further information on this study at Daniel.Weiner@tuftsmedicine.org

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

National Kidney Foundation – Spring Clinical Meetings 2024 (SCM24); May 14-18, 2024; Long Beach, CA, USA

MED-US—2400174 07/2025

INDICATION

XPHOZAH (tenapanor) is indicated to reduce serum phosphorus in adults with chronic kidney disease (CKD) on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.

IMPORTANT SAFETY INFORMATION

CONTRAINDICATIONS

XPHOZAH is contraindicated in patients under 6 years of age.

XPHOZAH is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Diarrhea

Patients may experience severe diarrhea. Treatment with XPHOZAH should be discontinued in patients who develop severe diarrhea.

MOST COMMON ADVERSE REACTIONS

Diarrhea, which occurred in 43% to 53% of patients, was the only adverse reaction reported in at least 5% of XPHOZAH-treated patients with CKD on dialysis across trials. The majority of diarrhea events in the XPHOZAH-treated patients were reported to be mild to moderate in severity and resolved over time or with dose reduction. Diarrhea was typically reported soon after initiation but could occur at any time during treatment with XPHOZAH. Severe diarrhea was reported in 5% of XPHOZAH-treated patients in these trials.

For additional safety information, please see full Prescribing Information, available here.

CKD, chronic kidney disease.
XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only